Acute Care Events and Treatment Patterns 12 Months After Initiating Olanzapine/Samidorphan: Subgroup Analyses of Patients With Schizophrenia or Bipolar I Disorder

Hemangi R. Panchmatia, MSc,[1] Andrew J. Cutler, MD,[2,3] Noah S. Webb, PhD,[4] Alejandro G. Hughes, MPH,[4] Michael J. Doane, PhD,[1] Hara E. Oyedeji, DNP, PMHNP-BC,[5] Rakesh Jain, MD, MPH[6]

[1]Alkermes, Inc., Waltham, MA, USA; [2]Department of Psychiatry, SUNY Upstate Medical University, Syracuse, NY, USA; [3]Neuroscience Education Institute, Lakewood Ranch, FL, USA; [4]Optum, Inc., Eden Prairie, MN, USA; [5]Fortitude Behavioral Health, Baltimore, MD, USA; [6]Department of Psychiatry, Texas Tech University School of Medicine-Permian Basin, Midland, TX, USA

BACKGROUND

- The combination of olanzapine and samidorphan (OLZ/SAM) is approved for the treatment of schizophrenia and bipolar I disorder (BD-I)[1]
 – It provides the established antipsychotic efficacy of olanzapine while mitigating olanzapine-associated weight gain[2,3]
- In a prior real-world analysis of patients with schizophrenia or BD-I, OLZ/SAM treatment significantly reduced acute care events, including disease-related events used as a proxy for relapse,[4,5] during the 12 months after treatment initiation compared with the 12 months prior[6]
- This analysis assessed the impact of OLZ/SAM initiation on treatment patterns and acute care events across different subgroups defined by baseline patient characteristics

OBJECTIVE

- Examine 12-month treatment patterns and acute care events by subgroups of patients with schizophrenia or BD-I after initiating OLZ/SAM treatment

METHODS

- This claims analysis used data from the Komodo Healthcare Map, a fully deidentified database of ~150 million patients covered by a commercial (66%), Medicaid (27%), or Medicare Advantage (7%) plan
- Eligible patients were aged ≥18 years with ≥1 pharmacy/medical claim for OLZ/SAM and ≥12 months continuous enrollment in pharmacy/medical benefits before (baseline period) and after (follow-up period) the index date (**Figure 1**)
- Outcomes of interest included treatment patterns (adherence, persistence, and discontinuation), as well as acute care events (inpatient [IP] admissions and emergency department [ED] visits) categorized by all-cause, mental health–related, and disease-related events
- *P*≤0.05 indicates a statistically significant difference when comparing the periods before vs after starting OLZ/SAM

Figure 1. Study Design

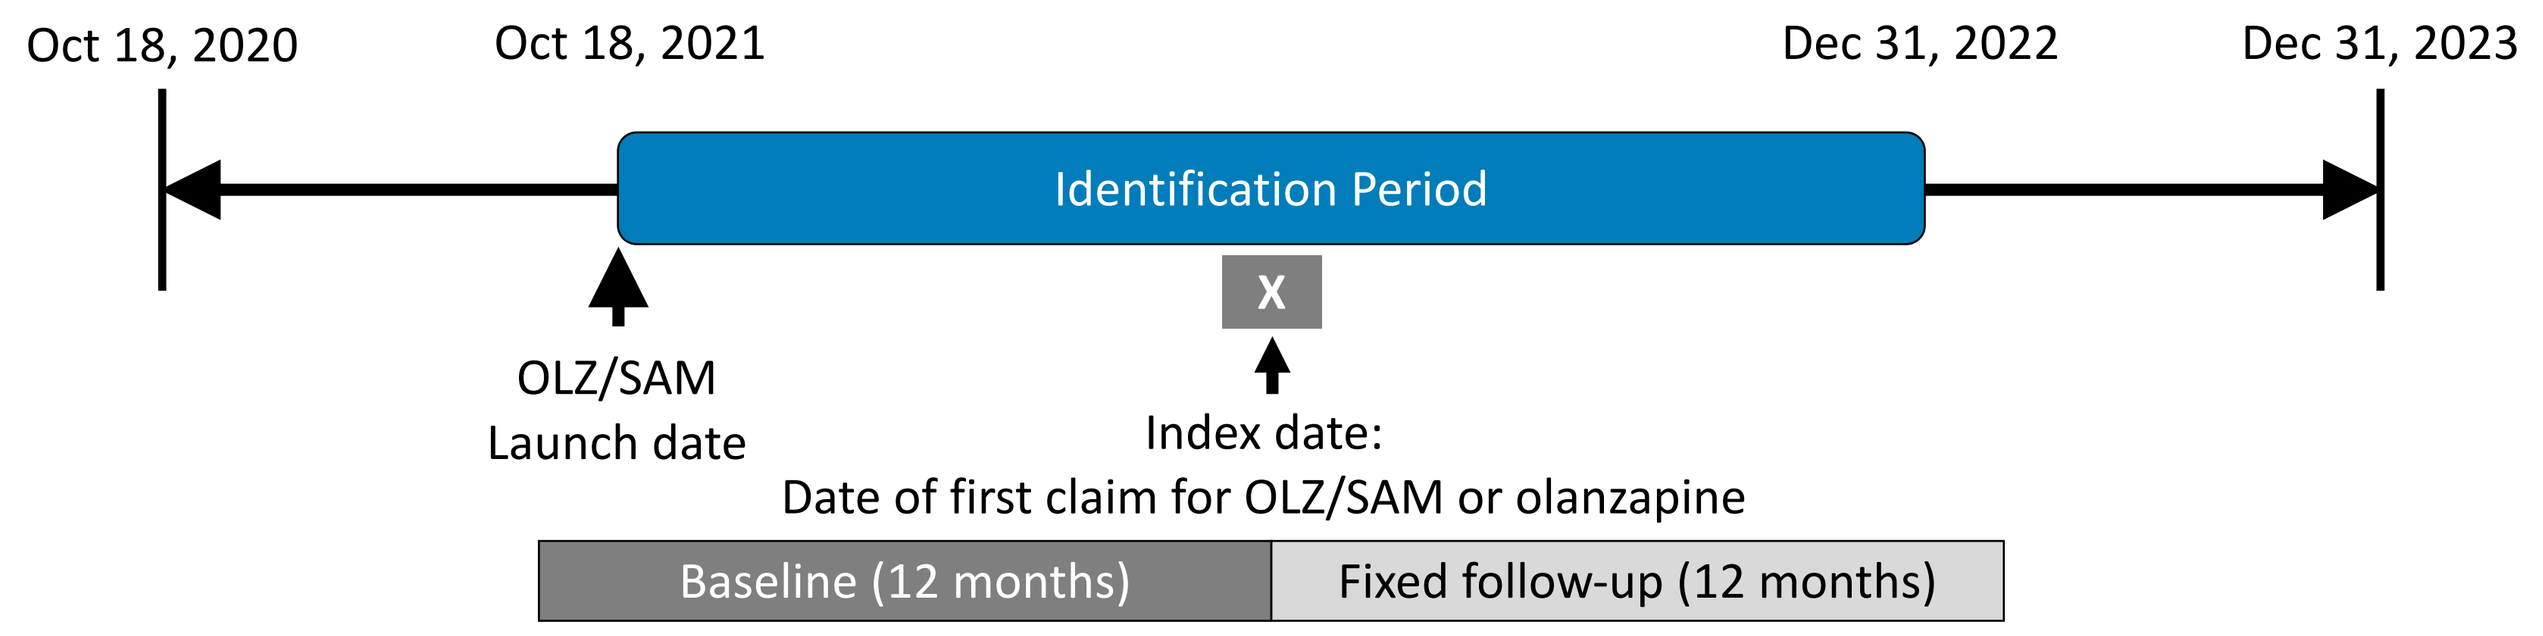
Oct 18, 2020
Oct 18, 2021
Dec 31, 2022
Dec 31, 2023
Identification Period
X
OLZ/SAM
Launch date
Index date:
Date of first claim for OLZ/SAM or olanzapine
Baseline (12 months)
Fixed follow-up (12 months)

OLZ/SAM, combined olanzapine and samidorphan.

RESULTS

- For both the schizophrenia and BD-I cohorts, baseline characteristics were generally similar across subgroups
- Measures of adherence, persistence, and discontinuation were generally similar across subgroups in both the schizophrenia (**Table 1**) and BD-I (**Table 2**) cohorts
- Across most subgroups in both cohorts (**Figure 2** and **Figure 3**), OLZ/SAM initiation was associated with lower proportions of patients experiencing ≥1 all-cause, mental health–related, or disease-related acute care event
 – The largest reductions in acute care events were observed in the commercially insured and 18- to 34-year-old subgroups of patients with schizophrenia and in the olanzapine as most recent prior antipsychotic and 18- to 34-year-old subgroups of those with BD-I; in both cases, these reductions were larger than those observed in the full study

Table 1. Treatment Patterns for Patients With Schizophrenia

Subgroups	N	MPR, mean	Persistence, mean, days	Discontinuation, %
All Patients	1287	0.87	196.6	62.6
Age, years				
18–34	564	0.87	197.7	62.6
35–54	532	0.88	194.5	63.4
≥55	191	0.87	199.0	60.7
Sex				
Female	590	0.87	188.7	65.8
Male	681	0.88	203.0	60.1
Most recent antipsychotic				
Olanzapine	673	0.89	205.0	59.1
Non-olanzapine	582	0.86	189.0	66.0
Insurance type				
Commercial	185	0.84	166.3	72.4
Medicaid	810	0.88	203.1	60.9
Medicare Advantage	289	0.87	196.3	61.6

MPR, medication possession ratio.

Acute Care Events in Patients With Schizophrenia

Figure 2. Absolute Changes in Proportions of Patients With Schizophrenia With ≥1 IP Admission or ED Visit After Initiating OLZ/SAM[a]

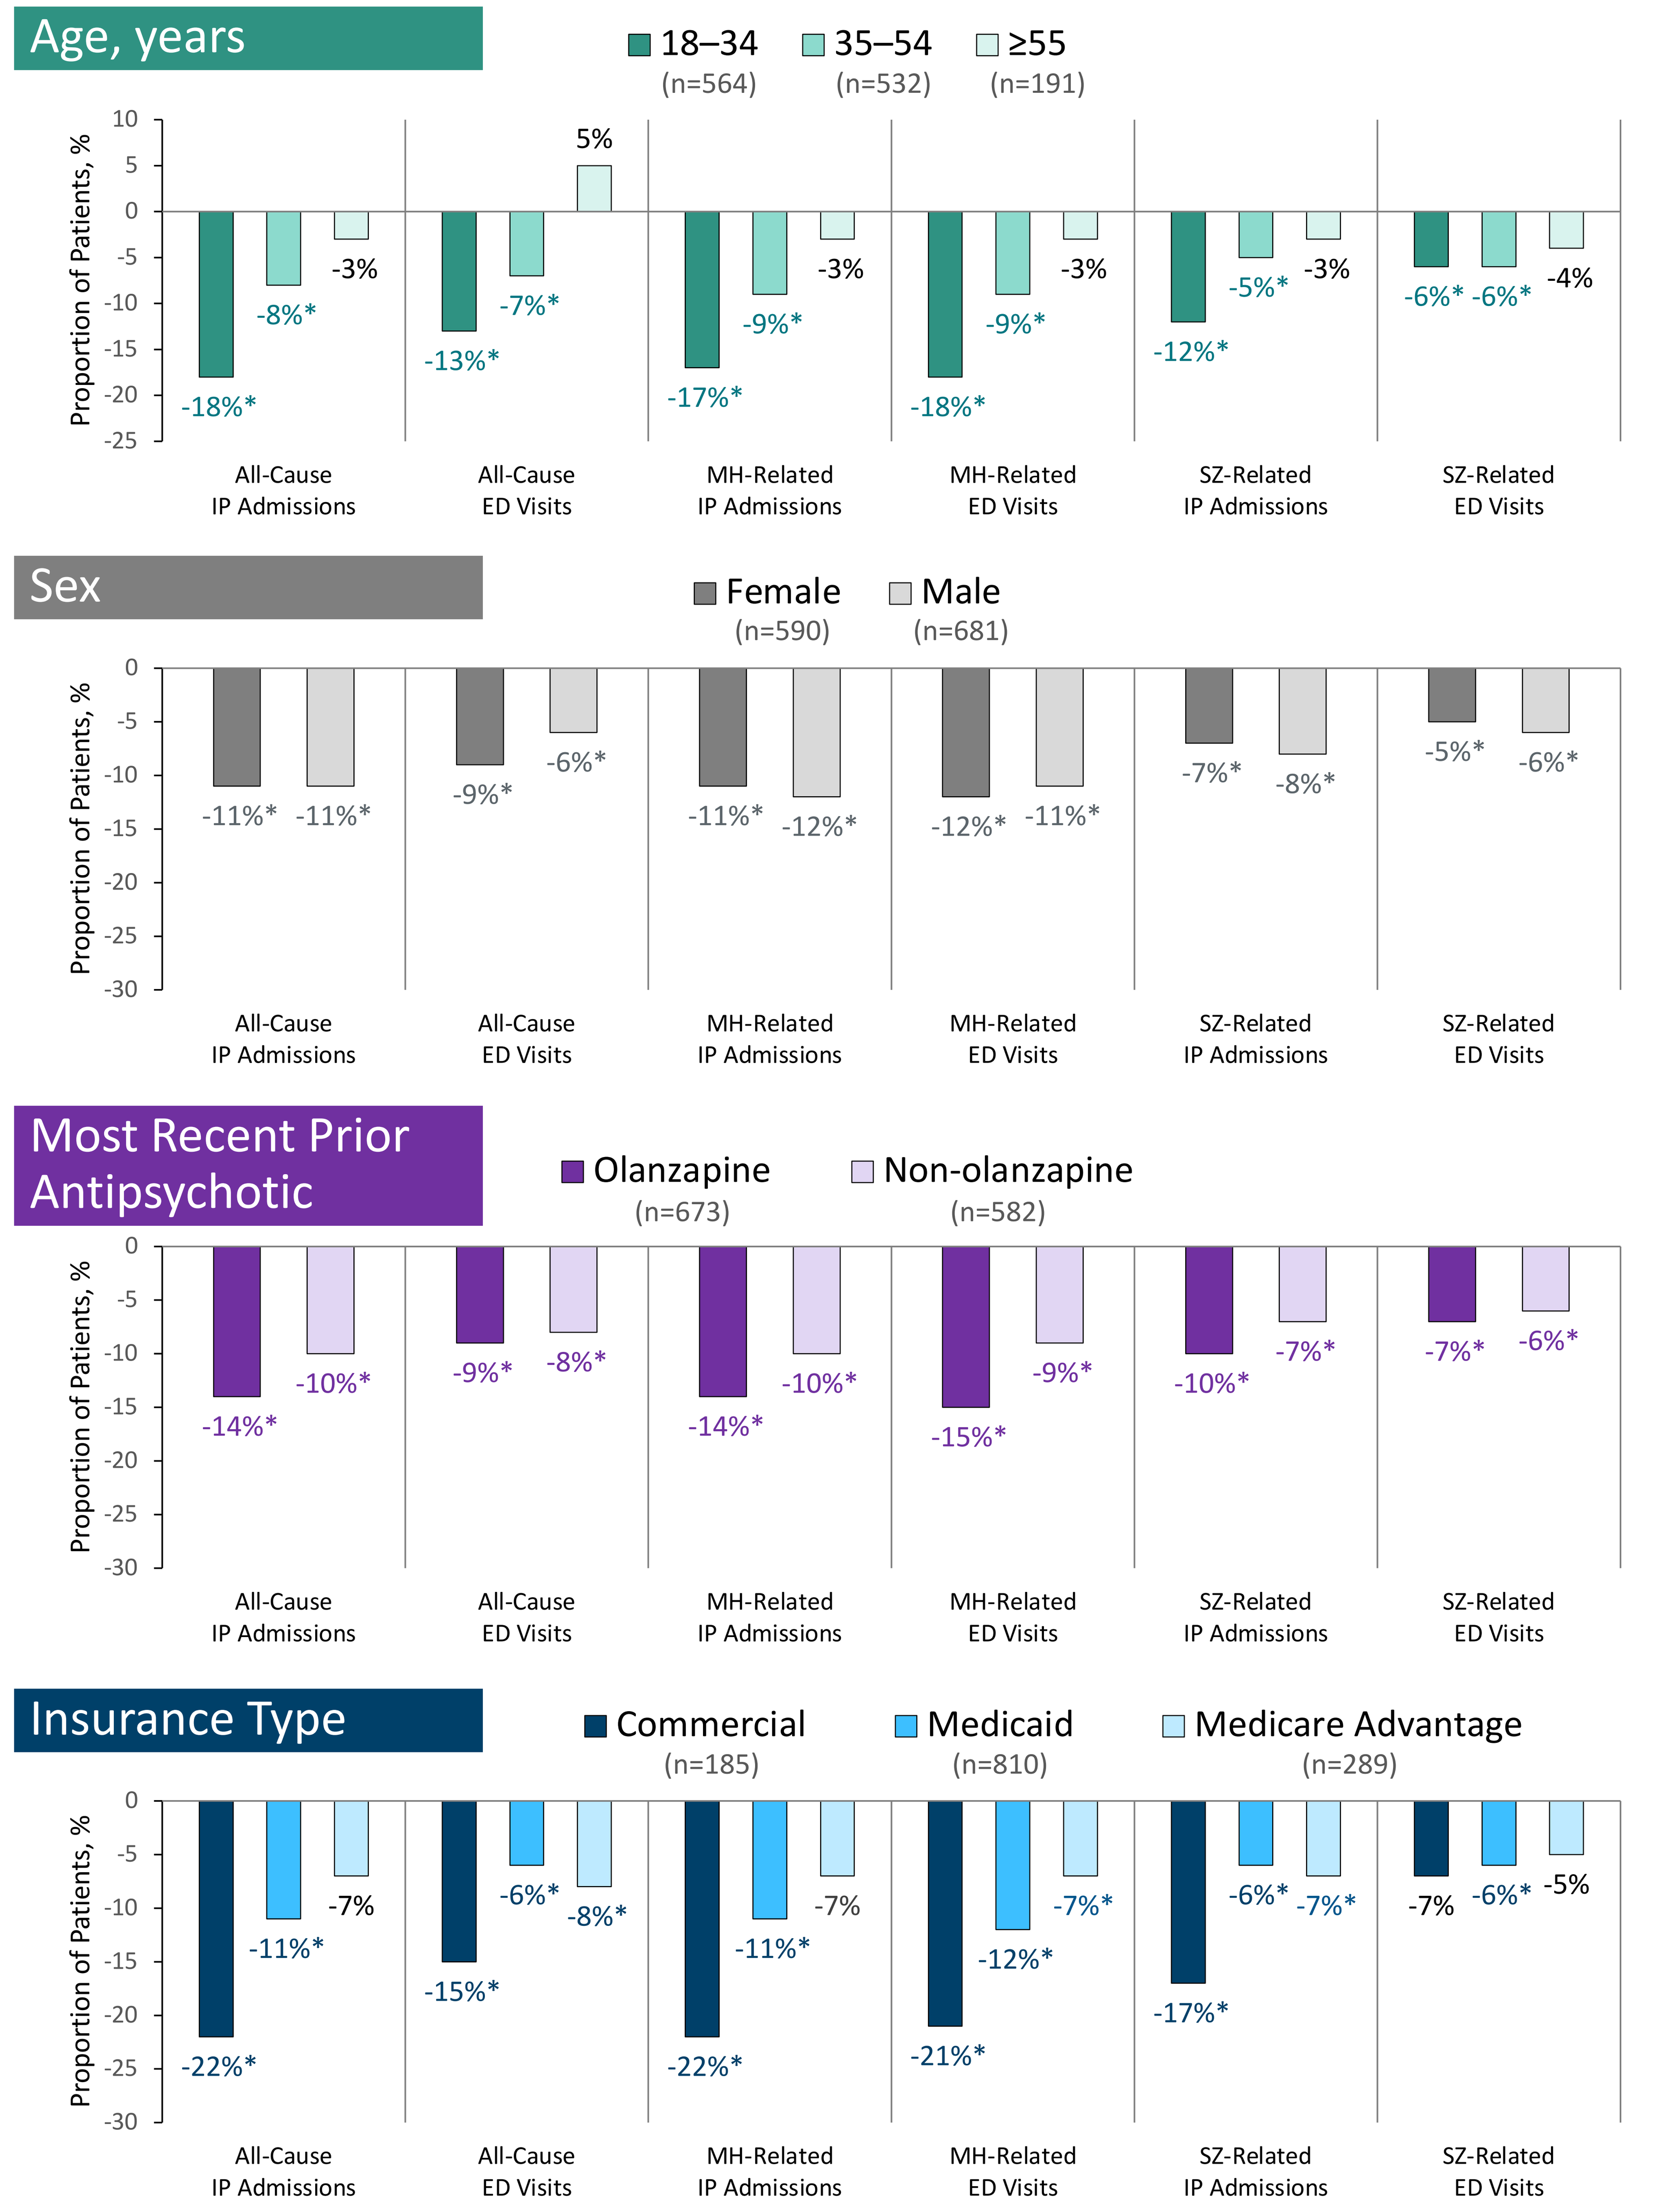
Age, years
18–34 (n=564)
35–54 (n=532)
≥55 (n=191)
Proportion of Patients, %
All-Cause IP Admissions: -18%* -8%* -3%
All-Cause ED Visits: -13%* -7%* 5%
MH-Related IP Admissions: -17%* -9%* -3%
MH-Related ED Visits: -18%* -9%* -3%
SZ-Related IP Admissions: -12%* -5%* -3%
SZ-Related ED Visits: -6%* -6%* -4%
Sex
Female (n=590)
Male (n=681)
All-Cause IP Admissions: -11%* -11%*
All-Cause ED Visits: -9%* -6%*
MH-Related IP Admissions: -11%* -12%*
MH-Related ED Visits: -12%* -11%*
SZ-Related IP Admissions: -7%* -8%*
SZ-Related ED Visits: -5%* -6%*
Most Recent Prior Antipsychotic
Olanzapine (n=673)
Non-olanzapine (n=582)
All-Cause IP Admissions: -14%* -10%*
All-Cause ED Visits: -9%* -8%*
MH-Related IP Admissions: -14%* -10%*
MH-Related ED Visits: -15%* -9%*
SZ-Related IP Admissions: -10%* -7%*
SZ-Related ED Visits: -7%* -6%*
Insurance Type
Commercial (n=185)
Medicaid (n=810)
Medicare Advantage (n=289)
All-Cause IP Admissions: -22%* -11%* -7%
All-Cause ED Visits: -15%* -6%* -8%*
MH-Related IP Admissions: -22%* -11%* -7%
MH-Related ED Visits: -21%* -12%* -7%*
SZ-Related IP Admissions: -17%* -6%* -7%*
SZ-Related ED Visits: -7% -6%* -5%

*P≤0.05, indicating a statistically significant difference when comparing the periods before vs after starting OLZ/SAM.
[a]Reductions in acute care events were numerically largest in the commercial insurance subgroup within the schizophrenia cohort. These patients with commercial insurance in the schizophrenia cohort tended to be of younger age, with over 65% in the younger age group (ie, 18-34 years old).
ED, emergency department; IP, inpatient; MH, mental health; OLZ/SAM, combined olanzapine and samidorphan; SZ, schizophrenia.

Acute Care Events in Patients With BD-I

Figure 3. Absolute Changes in Proportions of Patients With BD-I With ≥1 IP Admission or ED Visit After Initiating OLZ/SAM

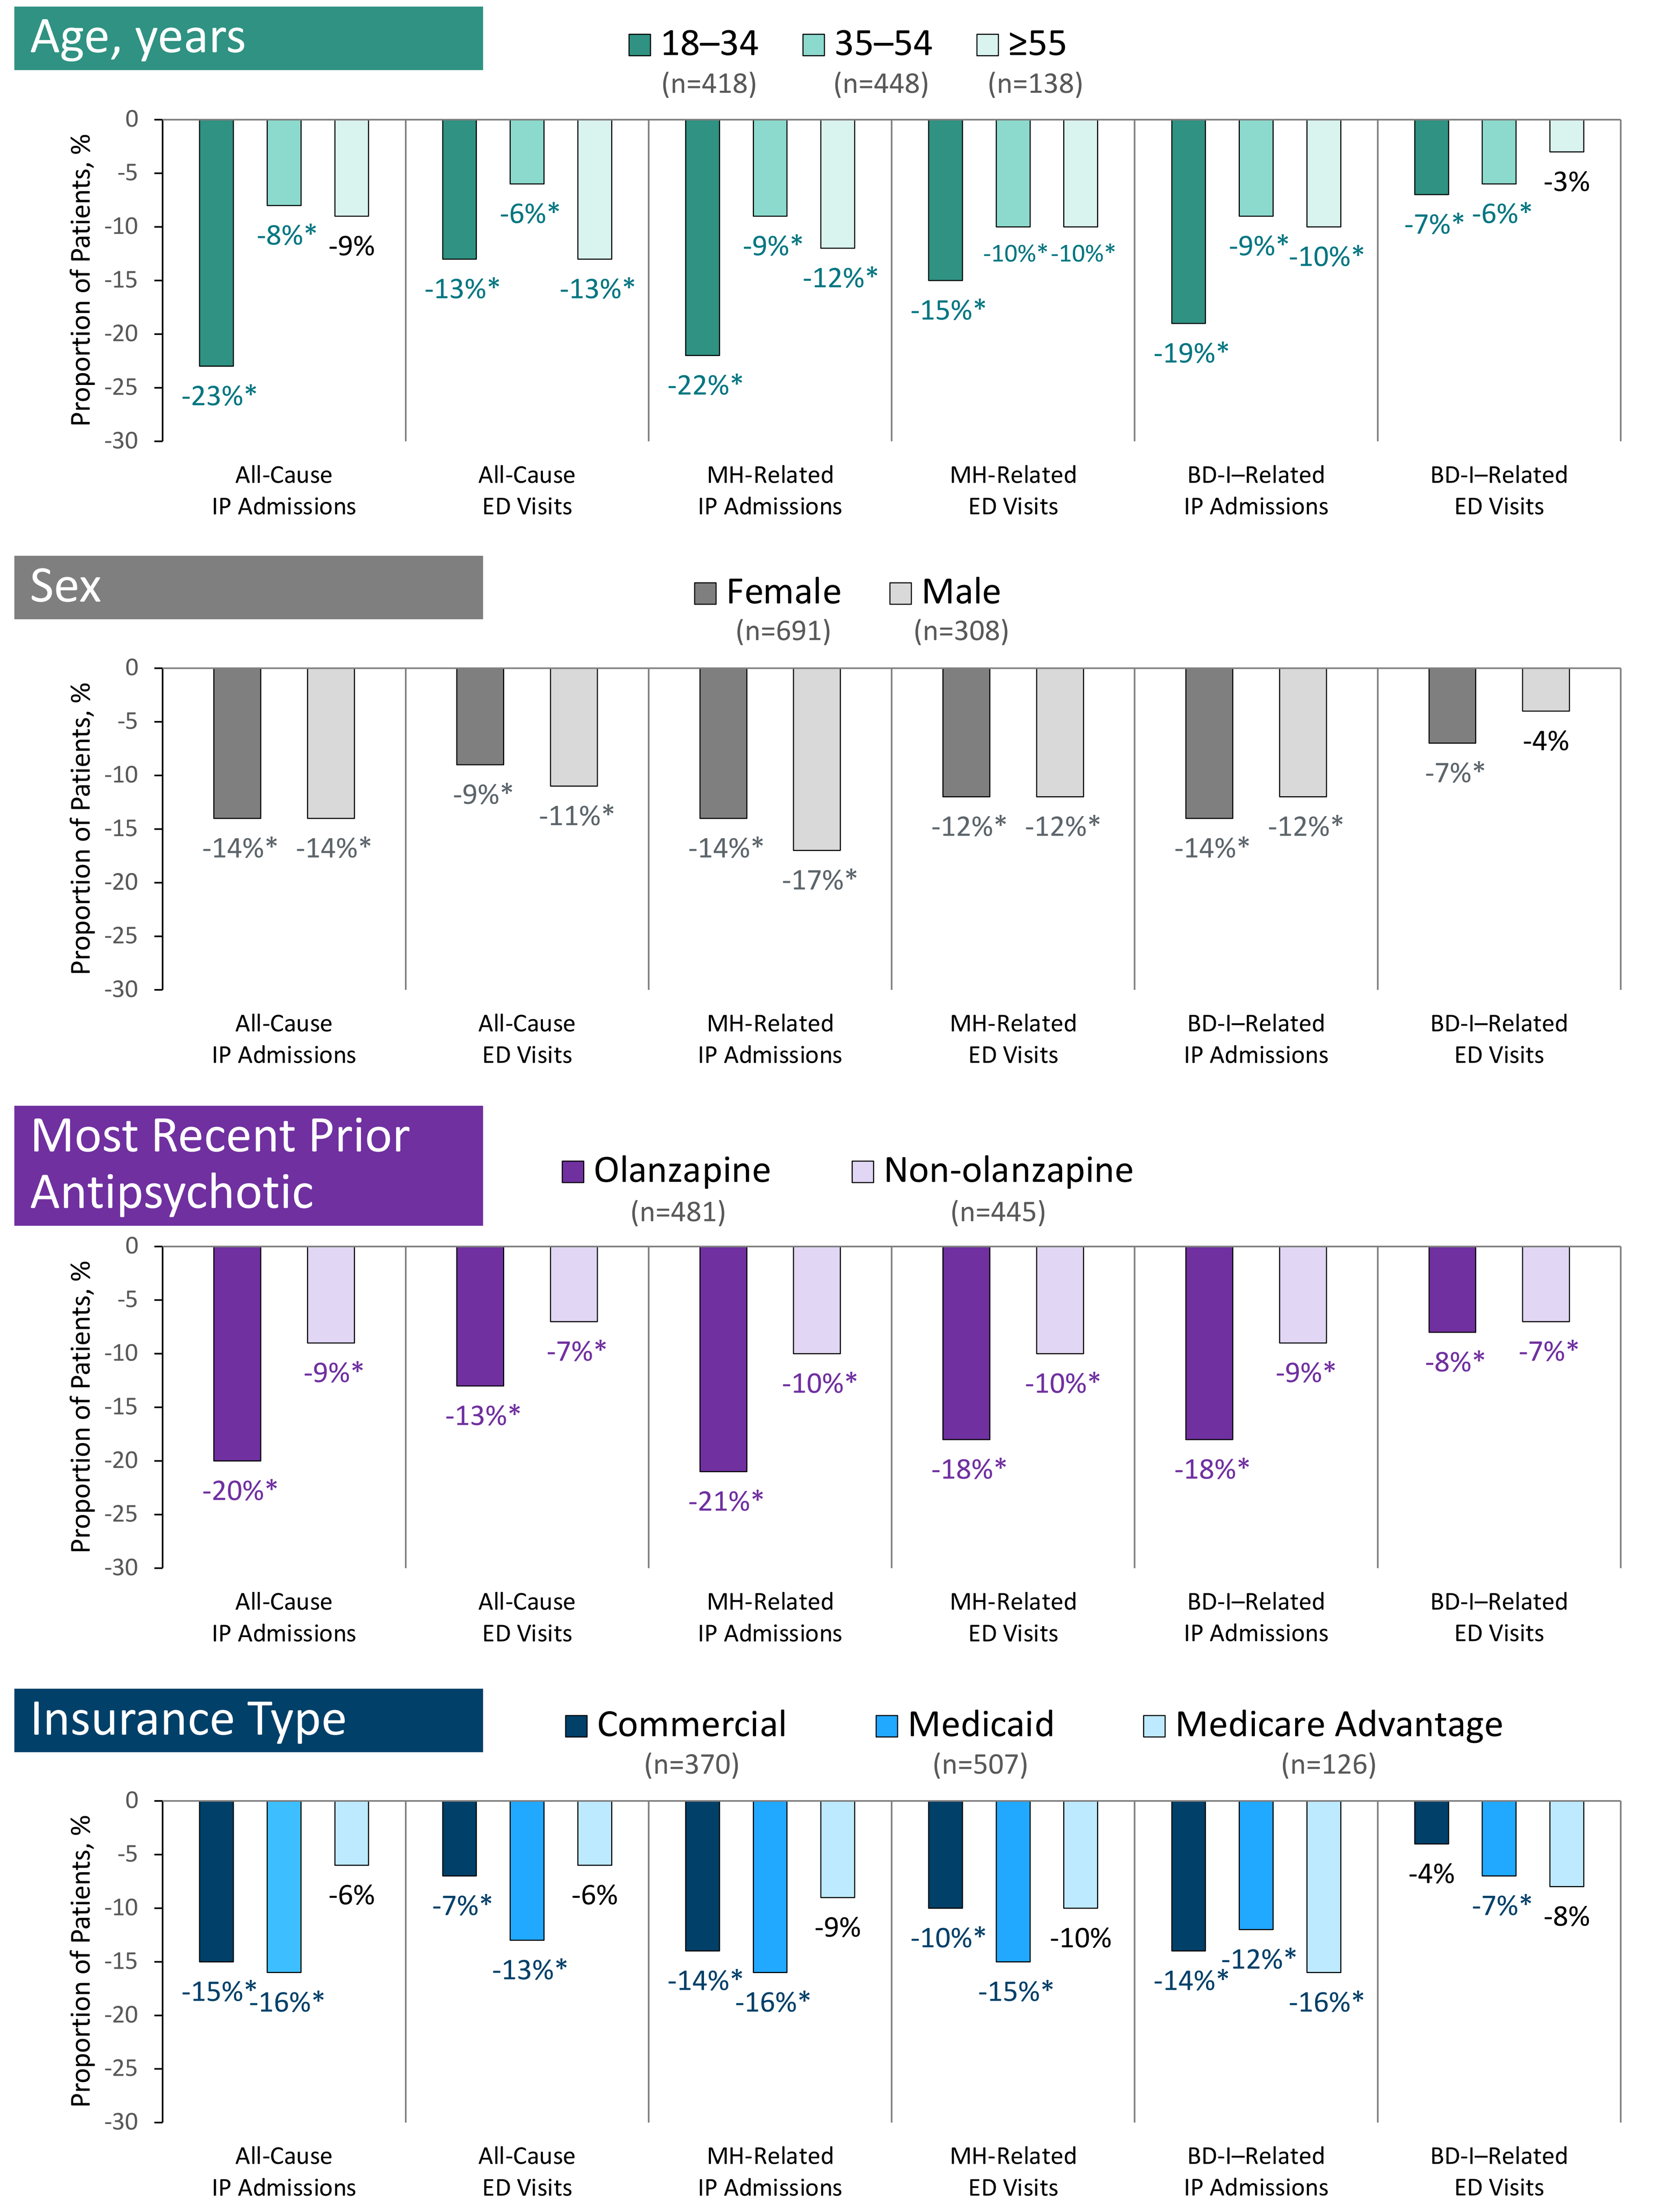
Age, years
18–34 (n=418)
35–54 (n=448)
≥55 (n=138)
Proportion of Patients, %
All-Cause IP Admissions: -23%* -8%* -9%
All-Cause ED Visits: -13%* -6%* -13%*
MH-Related IP Admissions: -22%* -9%* -12%*
MH-Related ED Visits: -15%* -10%* -10%*
BD-I–Related IP Admissions: -19%* -9%* -10%*
BD-I–Related ED Visits: -7%* -6%* -3%
Sex
Female (n=691)
Male (n=308)
All-Cause IP Admissions: -14%* -14%*
All-Cause ED Visits: -9%* -11%*
MH-Related IP Admissions: -14%* -17%*
MH-Related ED Visits: -12%* -12%*
BD-I–Related IP Admissions: -14%* -12%*
BD-I–Related ED Visits: -7%* -4%
Most Recent Prior Antipsychotic
Olanzapine (n=481)
Non-olanzapine (n=445)
All-Cause IP Admissions: -20%* -9%*
All-Cause ED Visits: -13%* -7%*
MH-Related IP Admissions: -21%* -10%*
MH-Related ED Visits: -18%* -10%*
BD-I–Related IP Admissions: -18%* -9%*
BD-I–Related ED Visits: -8%* -7%*
Insurance Type
Commercial (n=370)
Medicaid (n=507)
Medicare Advantage (n=126)
All-Cause IP Admissions: -15%* -16%* -6%
All-Cause ED Visits: -7%* -13%* -6%
MH-Related IP Admissions: -14%* -16%* -9%
MH-Related ED Visits: -10%* -15%* -10%
BD-I–Related IP Admissions: -14%* -12%* -16%*
BD-I–Related ED Visits: -4% -7%* -8%

*P≤0.05, indicating a statistically significant difference when comparing the periods before vs after starting OLZ/SAM.
BD-I, bipolar I disorder; ED, emergency department; IP, inpatient; MH, mental health; OLZ/SAM, combined olanzapine and samidorphan.

Table 2. Treatment Patterns for Patients With BD-I

Subgroups	N	MPR, mean	Persistence, mean, days	Discontinuation, %
All patients	1004	0.86	173.7	70.1
Age, years				
18–34	418	0.86	173.0	70.1
35–54	448	0.87	176.0	68.5
≥55	138	0.84	168.2	75.4
Sex				
Female	691	0.85	173.0	70.3
Male	308	0.88	177.1	69.2
Most recent antipsychotic				
Olanzapine	481	0.87	180.5	68.4
Non-olanzapine	445	0.86	170.5	70.1
Insurance type				
Commercial	370	0.85	157.2	74.9
Medicaid	507	0.86	185.3	66.9
Medicare Advantage	126	0.89	176.3	69.1

BD-I, bipolar I disorder; MPR, medication possession ratio.

LIMITATIONS

- Results from these cohorts may not be generalizable to uninsured cohorts
- Claims data do not capture disease severity and are subject to data omissions and/or coding inaccuracies
- Presence of a claim for a filled prescription does not indicate that the medication was consumed
- Due to the fixed follow-up time, treatment patterns and acute care events reported may not fully capture the effects of longer-term (>12 months) OLZ/SAM use

CONCLUSIONS

- Treatment patterns were generally similar across all subgroups in both cohorts of patients with schizophrenia or BD-I
- Across most subgroups, initiating OLZ/SAM was associated with significant reductions in proportions of patients with ≥1 all-cause, mental health–related, or disease-related IP admission or ED visit in both the schizophrenia and BD-I cohorts
 – Reductions in disease-related acute care events suggest a reduction in relapse after the initiation of OLZ/SAM
- Greater reductions in young adults (18- to 34-year-old subgroup) across both cohorts suggest that earlier intervention with OLZ/SAM treatment may lead to greater reductions in acute care events
- Patients with commercial insurance in the schizophrenia cohort had the largest reductions in acute care events; over 65% of this population was composed of younger patients (ie, 18-34 years old), again pointing to the association of initiating OLZ/SAM in younger patients with greater reductions in acute care events

REFERENCES

1. Lybalvi [package insert]. Waltham, MA: Alkermes, Inc.; 2025.
2. Correll CU, et al. *Am J Psychiatry*. 2020;177(12):1168-1178. DOI: 10.1176/appi.ajp.2020.19121279.
3. Kahn RS, et al. *J Clin Psychiatry*. 2023;84(3):22m14674. DOI: 10.4088/JCP.22m14674.
4. Olivares JM, et al. *Ann Gen Psychiatry*. 2013;12(1):32. DOI: 10.1186/1744-859X-12-32.
5. Hassan M, Lage MJ. *Am J Health Syst Pharm*. 2009;66(4):358-365. DOI: 10.2146/ajhp080374.
6. Jain R, et al. Poster presented at: Psych Congress 2024; Oct 29–Nov 2, 2024; Boston, MA.

ACKNOWLEDGMENTS

This study was sponsored by Alkermes, Inc. Medical writing and editorial support were provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Alkermes, Inc.

Disclosure information and copies of this poster can be obtained through this QR (Quick Response) code. These materials are for personal use only and may not be reproduced without permission of Alkermes. For permission, contact USMedInfo@Alkermes.com.

Supported by funding from Alkermes, Inc.

Acute Care Events and Treatment Patterns 12 Months After Initiating Olanzapine/Samidorphan: Subgroup Analyses of Patients With Schizophrenia or Bipolar I Disorder

Hemangi R. Panchmatia, MSc,[1] Andrew J. Cutler, MD,[2,3] Noah S. Webb, PhD,[4] Alejandro G. Hughes, MPH,[4] Michael J. Doane, PhD,[1] Hara E. Oyedeji, DNP, PMHNP-BC,[5] Rakesh Jain, MD, MPH[6]

[1]Alkermes, Inc., Waltham, MA, USA; [2]Department of Psychiatry, SUNY Upstate Medical University, Syracuse, NY, USA; [3]Neuroscience Education Institute, Lakewood Ranch, FL, USA; [4]Optum, Inc., Eden Prairie, MN, USA; [5]Fortitude Behavioral Health, Baltimore, MD, USA; [6]Department of Psychiatry, Texas Tech University School of Medicine-Permian Basin, Midland, TX, USA

DISCLOSURES

HRP and **MJD** are or were employees of Alkermes, Inc., and may own stock/options in the company.

AJC has been a consultant or on an advisory board for AbbVie, Acadia, Alfasigma, Alkermes, Anavex Life Sciences, Autobahn Therapeutics, Axsome, Biogen, Biohaven, BioXcel, Boehringer Ingelheim, Bristol Myers Squibb, Cerevel, Collegium Pharmaceutical, Corium, Delpor, 4M Therapeutics, Intra-Cellular Therapies, J&J Innovative Medicine, Jazz Pharma, Karuna, Knight Therapeutics, LivaNova, Lundbeck, Luye Pharma, MapLight Therapeutics, Mentavi, Neumora, Neurocrine, NeuroSigma, Noven, Otsuka, PaxMedica, Relmada, Sage Therapeutics, Supernus, Teva, Thynk, Tris Pharma, Vanda Pharmaceuticals, and VistaGen; is on the speakers bureau for AbbVie, Alfasigma, Alkermes, Axsome, Boehringer Ingelheim, Bristol Myers Squibb, Collegium Pharmaceutical, Corium, Innovative Medicine, Intra-Cellular Therapies, J&J, Knight Therapeutics, Lundbeck, Neurocrine, Noven, Otsuka, Supernus, Teva, Tris Pharma, and Vanda Pharmaceuticals; is on a data safety monitoring board for COMPASS Pathways; holds stock options from 4M Therapeutics; and receives no royalties.

NSW and **AGH** are or were employees of Optum, Inc., a health services innovation company that received funding from Alkermes, Inc., to conduct this study and analyze the data used for this publication.

HEO has been a consultant for Alkermes, Biogen, Bristol Myers Squibb, Intra-Cellular Therapies, Janssen, Karuna, Neurocrine, Otsuka, Sage Therapeutics, and Sunovion; is on the speakers bureau and has received honoraria from Alkermes, Bristol Myers Squibb, Intra-Cellular Therapies, Lundbeck, Neurocrine, Otsuka, and Teva; receives no royalties; and holds no stock options.

RJ has been a consultant for AbbVie, Acadia, Adamas, Alfasigma, Alkermes, Almatica, Axsome, Biogen, Boehringer Ingelheim, Cingulate Therapeutics, Corium, Eisai, Evidera, Impel, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Neurocrine Biosciences, Osmotica, Otsuka, Pamlab, Pfizer, Sage Therapeutics, Shire, Sunovion, Supernus, Takeda, Teva, Transcend Therapeutics, and Viatris; received speaker/promotional honoraria from AbbVie, Alkermes, Almatica, Axsome, Corium, Eisai, Intra-Cellular Therapies, Ironshore Pharmaceuticals, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Otsuka, Pamlab, Pfizer, Shire, Sunovion, Takeda, Tris Pharmaceuticals, and Viatris; served on an advisory board for Adamas, Alkermes, Corium, Eisai, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Neurocrine Biosciences, Otsuka, Pamlab, Pfizer, Sage Therapeutics, Shire, Sunovion, Supernus, Takeda, and Teva; and received research funding from AbbVie, Lilly, Lundbeck, Otsuka, Pfizer, Shire, and Takeda.

Supported by funding from Alkermes, Inc.