Baseline Severity of Illness and Response to Treatment With Aripiprazole Lauroxil Every 2 Months: A Post Hoc Analysis of Phase 3 ALPINE Clinical Trial Data

John M. Kane, MD[1]; Martin Dunbar, PhD[2]; and James A. McGrory, PhD[2]

[1]The Donald and Barbara Zucker School of Medicine at Hofstra/Northwell, Hempstead, NY, USA; [2]Alkermes, Inc., Waltham, MA, USA

BACKGROUND

- In patients with schizophrenia, severity of illness and symptoms related to hostility/excitement or activation may be associated with poor outcomes, including nonadherence to medication or discontinuation,[1,2] complicating treatment
- Aripiprazole lauroxil (AL) is a long-acting injectable (LAI) atypical antipsychotic medication indicated for the treatment of schizophrenia in adults
- The randomized, controlled, phase 3b ALPINE (Aripiprazole Lauroxil and Paliperidone palmitate: INitiation Effectiveness) study evaluated the efficacy and safety of a 2-month formulation of AL[3]
 - Paliperidone palmitate (PP) was included as an active control with known effectiveness[4]; ALPINE was not designed as or powered for a direct comparison between AL and PP
- Primary efficacy results from the ALPINE study were reported previously[3]
 - Within-group improvement from baseline in Positive and Negative Syndrome Scale[5] (PANSS) Total scores was observed at weeks 4, 9, and 25 for AL and PP

OBJECTIVE

- This post hoc analysis examined the efficacy and safety of AL every 2 months by baseline severity of illness in adult patients with schizophrenia enrolled in the ALPINE study

METHODS

Patients

- The ALPINE study (NCT03345979) enrolled acutely ill adults (aged 18–65 years) diagnosed according to *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition*, criteria with schizophrenia and requiring hospitalization
- Enrollment criteria included a PANSS Total score of 80–120, with a score of ≥4 on ≥2 of PANSS Positive scale items 1 (delusions), 2 (conceptual disorganization), 3 (hallucinatory behavior), and 6 (suspiciousness/persecution)

Study Design and Assessments

- Patients were enrolled and randomized as inpatients during an acute exacerbation of schizophrenia, discharged after 2 weeks (if clinically stable), and followed as outpatients for the remainder of the 25 weeks (**Figure 1**)
 - AL was initiated using a 1-day regimen (a single intramuscular [IM] injection of the NanoCrystal Dispersion formulation of AL [675 mg] and a single 30-mg oral aripiprazole tablet) on day 1, followed by AL 1064 mg IM on day 8 and then every 8 weeks thereafter
 - PP was initiated at 234 mg IM on day 1, followed by PP 156 mg IM on day 8 and every 4 weeks thereafter
- PANSS score was assessed at baseline, day 4, and weeks 1, 2, 4, 9, and 25
- In this post hoc analysis, severity of illness was categorized according to baseline Clinical Global Impression–Severity[6] (CGI-S) scores as moderately (CGI-S=4), markedly (CGI-S=5), or severely (CGI-S=6) ill
- Changes from baseline in PANSS Total and PANSS Activation (also referred to as Excitement)[7,8] factor scores were determined at each postbaseline assessment
 - PANSS Activation factor score was calculated as the sum of hostility, anxiety, excitement, tension, and poor impulse control item scores
- Frequency of adverse events (AEs) related to Activation factor symptoms, including anxiety, agitation, and insomnia, was assessed by baseline severity for the AL and PP groups

Figure 1. ALPINE Study Design

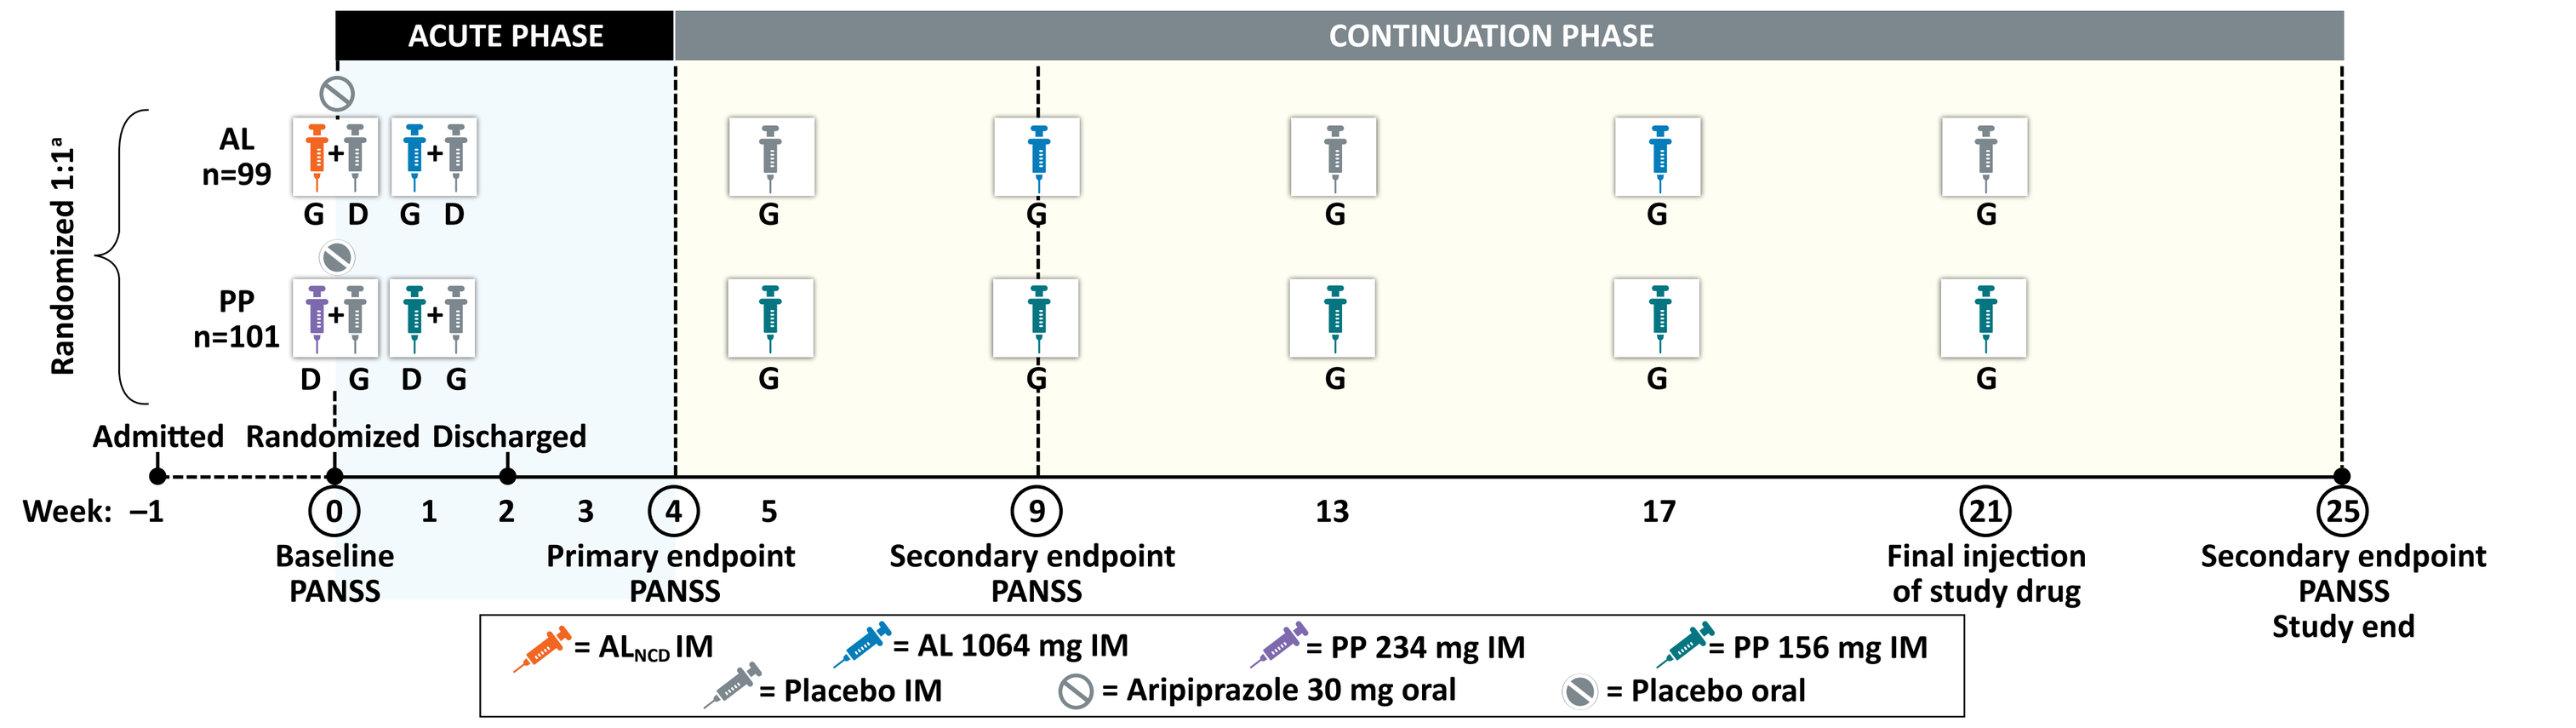
ACUTE PHASE
CONTINUATION PHASE
Randomized 1:1[a]
AL n=99
PP n=101
G D G D
D G D G
Admitted Randomized Discharged
Week: −1 0 1 2 3 4 5 9 13 17 21 25
Baseline PANSS
Primary endpoint PANSS
Secondary endpoint PANSS
Final injection of study drug
Secondary endpoint PANSS Study end
= AL$_{NCD}$ IM
= AL 1064 mg IM
= PP 234 mg IM
= PP 156 mg IM
= Placebo IM
= Aripiprazole 30 mg oral
= Placebo oral

[a]Because AL initiation required gluteal injection and PP initiation required deltoid injection, placebo injections were administered in patients' deltoid and gluteal muscles, respectively, during initiation to maintain blinding. The AL group also received a placebo injection at weeks 5, 13, and 21 to match the PP dosing schedule, and the PP group received an oral placebo tablet on day 1 to match the oral dose of aripiprazole in the AL initiation regimen.
AL, aripiprazole lauroxil; AL$_{NCD}$, NanoCrystal Dispersion formulation of AL; ALPINE, Aripiprazole Lauroxil and Paliperidone palmitate: INitiation Effectiveness; D, deltoid; G, gluteal; IM, intramuscular; PANSS, Positive and Negative Syndrome Scale; PP, paliperidone palmitate.

Statistical Analysis

- In this post hoc analysis, changes from baseline in PANSS Total and PANSS Activation factor scores were summarized descriptively for AL and PP separately by baseline severity for the AL and PP groups
- No formal statistical testing was conducted

RESULTS

Patients

Table 1. Demographics and Baseline Clinical Characteristic Scores by Baseline Severity Group[a]

Aripiprazole Lauroxil			
Characteristics	Moderately Ill (n=31)	Markedly Ill (n=54)	Severely Ill (n=14)
Age, mean (SD), years	46.4 (9.9)	42.5 (8.9)	41.1 (11.2)
BMI, mean (SD), kg/m^2	29.0 (4.8)	28.5 (5.9)	25.3 (5.0)
PANSS Total score,[b] mean (SD)	87.1 (5.5)	95.3 (7.3)	106.1 (7.2)
PANSS Activation factor score,[b] mean (SD)	12.3 (3.0)	14.4 (3.4)	16.3 (3.0)

Paliperidone Palmitate			
Characteristic	Moderately Ill (n=26)	Markedly Ill (n=59)	Severely Ill (n=16)
Age, mean (SD), years	44.3 (12.8)	42.4 (10.1)	45.3 (10.4)
BMI, mean (SD), kg/m^2	28.4 (5.2)	28.1 (5.1)	25.9 (4.8)
PANSS Total score,[c] mean (SD)	88.7 (5.1)	94.6 (7.6)	104.6 (6.1)
PANSS Activation factor score,[c] mean (SD)	12.9 (3.0)	14.4 (3.5)	16.0 (2.1)

[a]Moderately ill, CGI-S=4; markedly ill, CGI-S=5; severely ill, CGI-S=6. [b]Efficacy analysis population for AL: moderately ill, n=31; markedly ill, n=52; severely ill, n=13. [c]Efficacy analysis population for PP: moderately ill, n=26; markedly ill, n=57; severely ill, n=16.
AL, aripiprazole lauroxil; BMI, body mass index; CGI-S, Clinical Global Impression–Severity; PANSS, Positive and Negative Syndrome Scale; PP, paliperidone palmitate.

Efficacy

Figure 2. Changes From Baseline in PANSS Total Score by Baseline Severity Group[a]

A. Aripiprazole Lauroxil

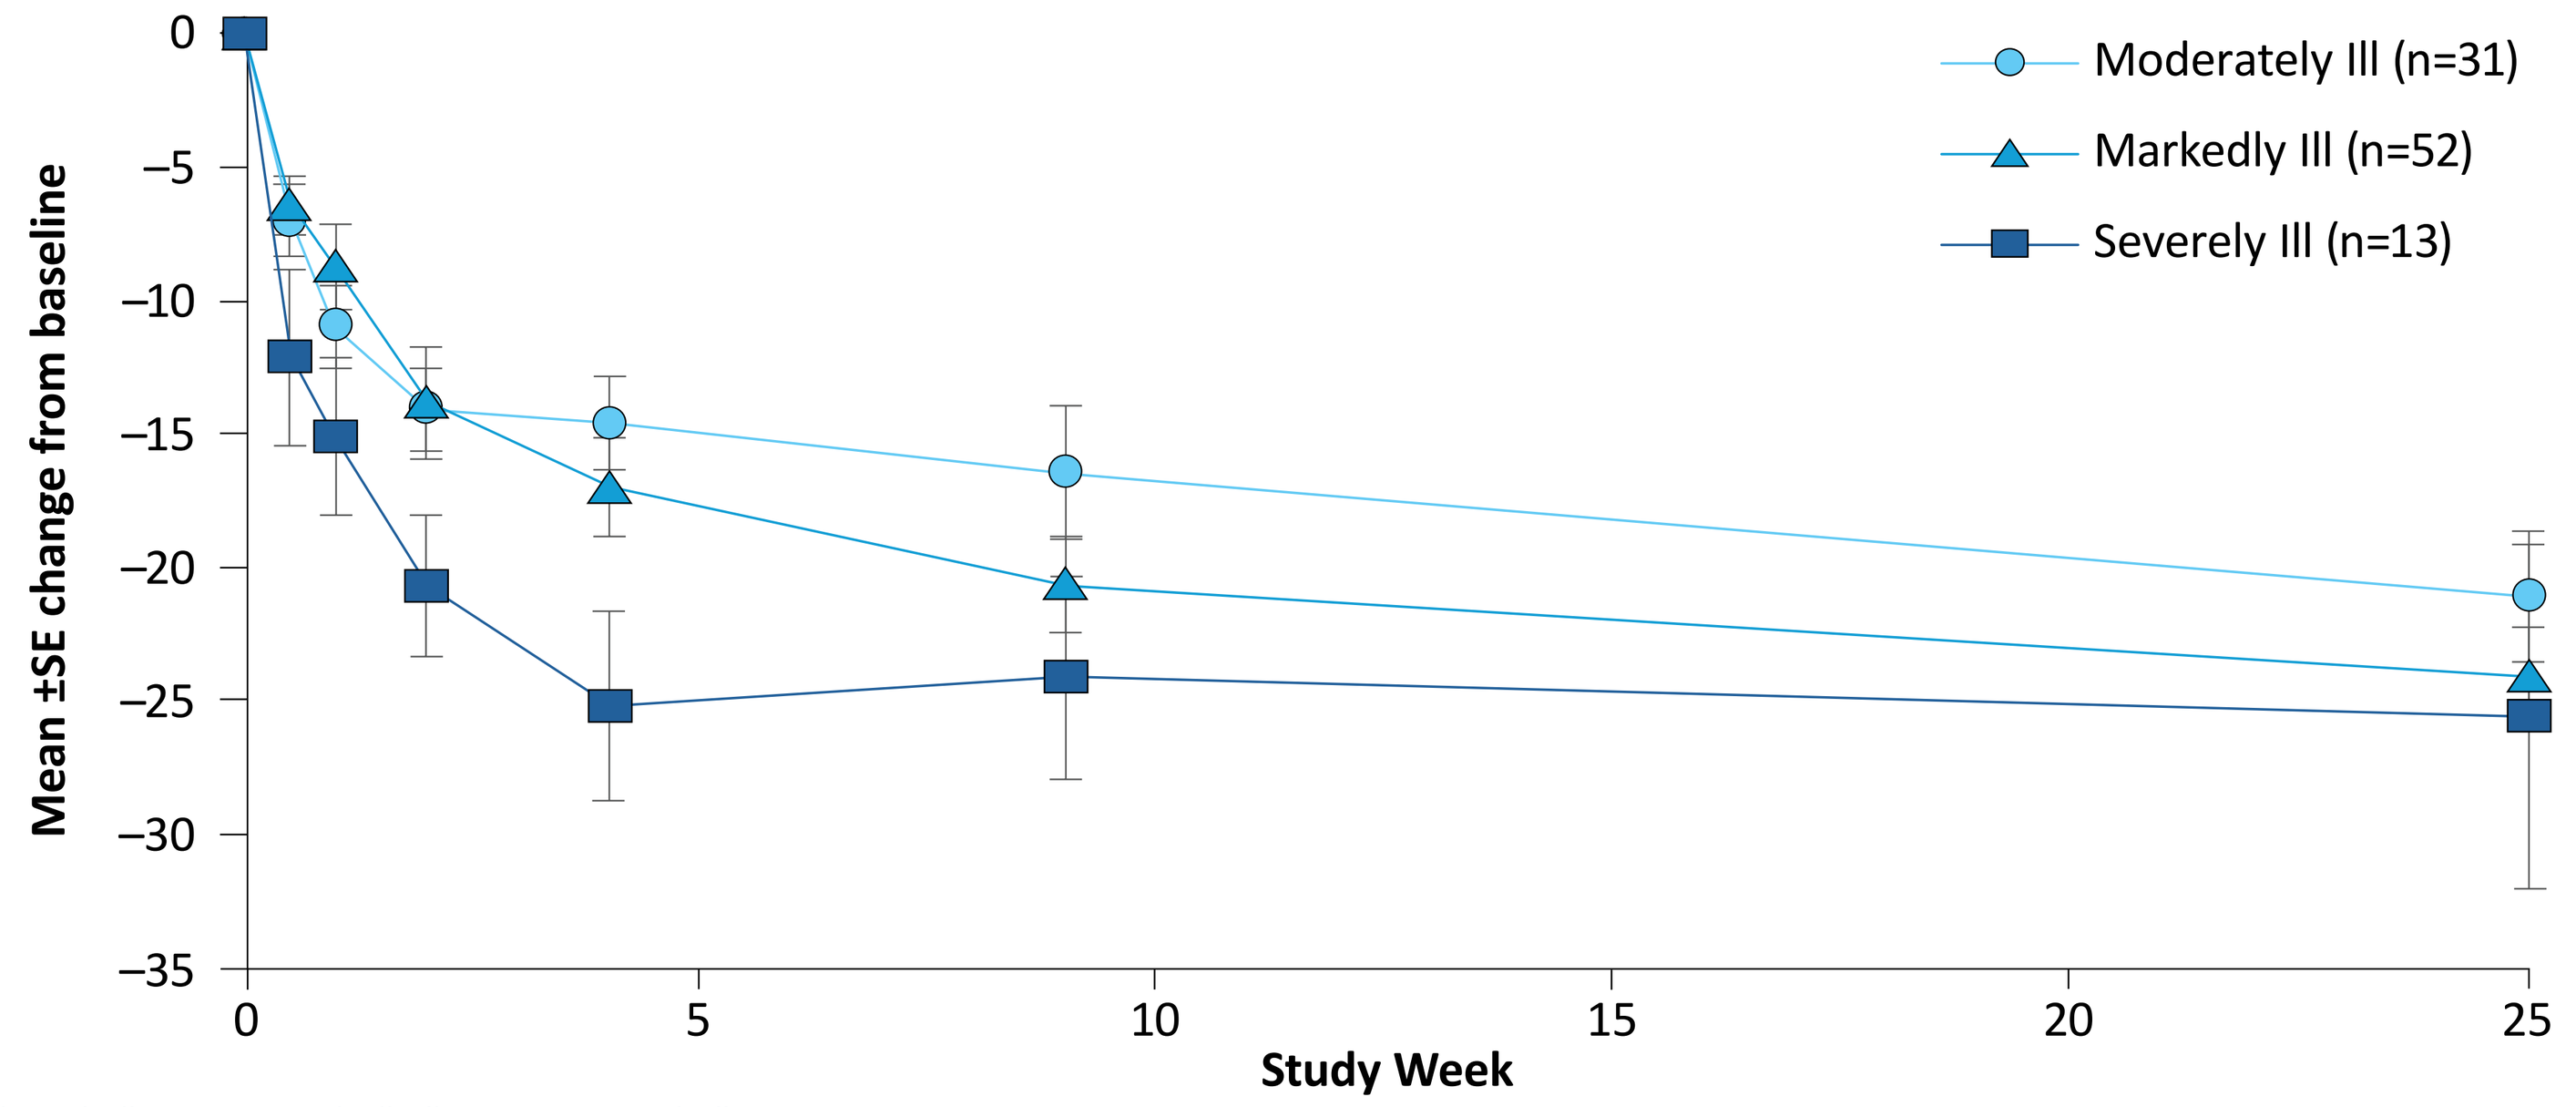
Moderately Ill (n=31)
Markedly Ill (n=52)
Severely Ill (n=13)
Mean ±SE change from baseline
0
−5
−10
−15
−20
−25
−30
−35
0 5 10 15 20 25
Study Week

B. Paliperidone Palmitate

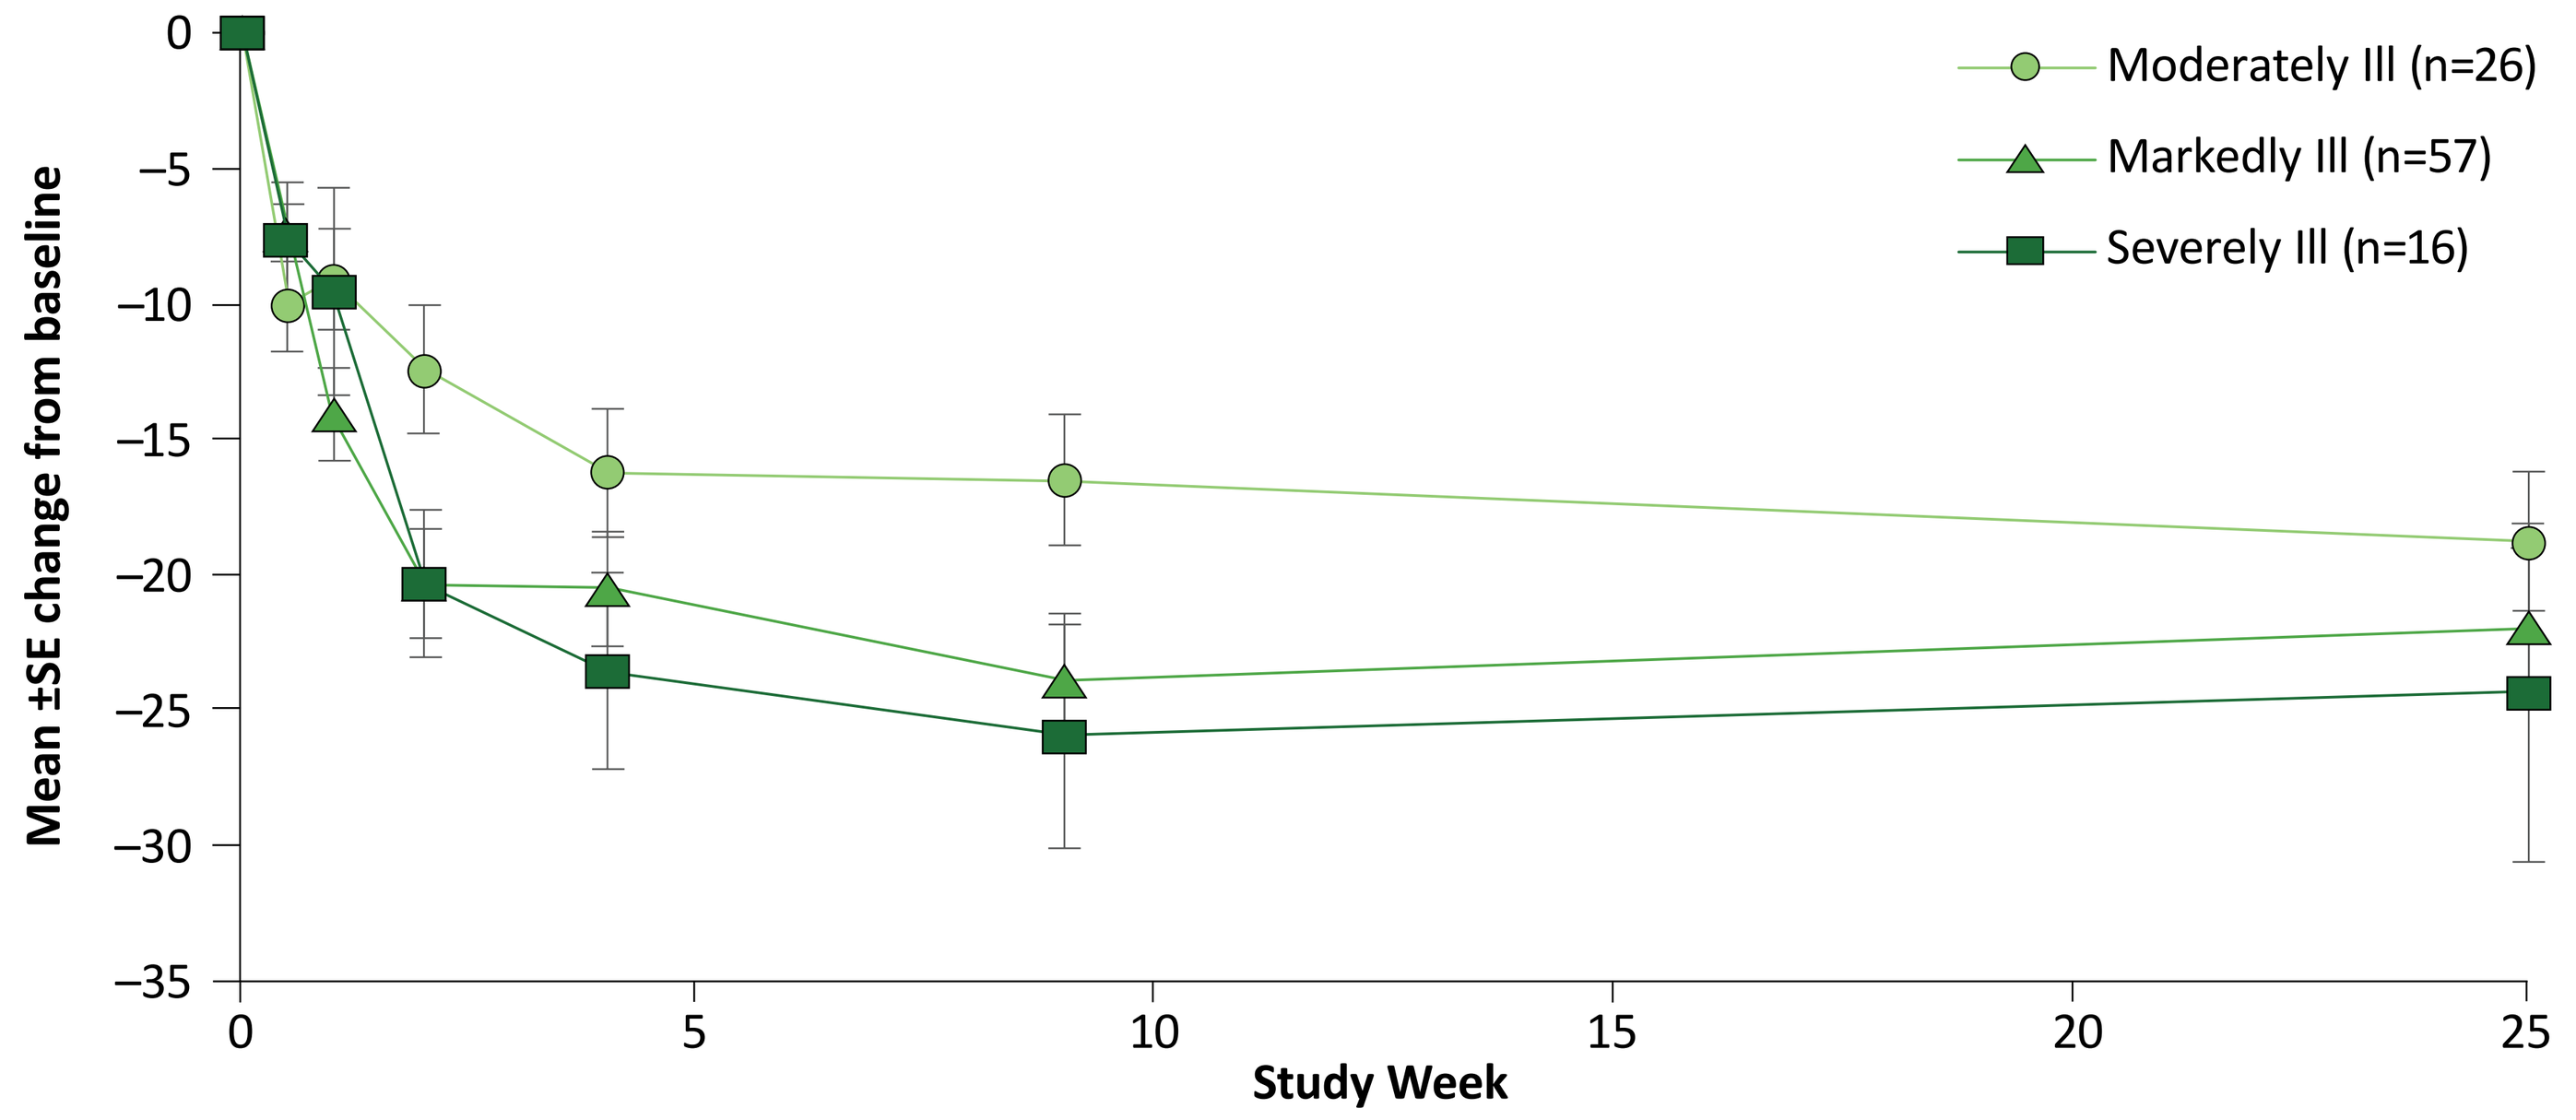
Moderately Ill (n=26)
Markedly Ill (n=57)
Severely Ill (n=16)
Mean ±SE change from baseline
0
−5
−10
−15
−20
−25
−30
−35
0 5 10 15 20 25
Study Week

[a]Moderately ill, CGI-S=4; markedly ill, CGI-S=5; severely ill, CGI-S=6.
CGI-S, Clinical Global Impression–Severity; PANSS, Positive and Negative Syndrome Scale.

- In the overall population, observed mean change from baseline in PANSS Total score at week 25, as reported in the primary ALPINE publication,[3] was –23.3 for the AL group and –21.7 for the PP group

Figure 3. Changes From Baseline in PANSS Activation Factor Score by Baseline Severity Group[a]

A. Aripiprazole Lauroxil

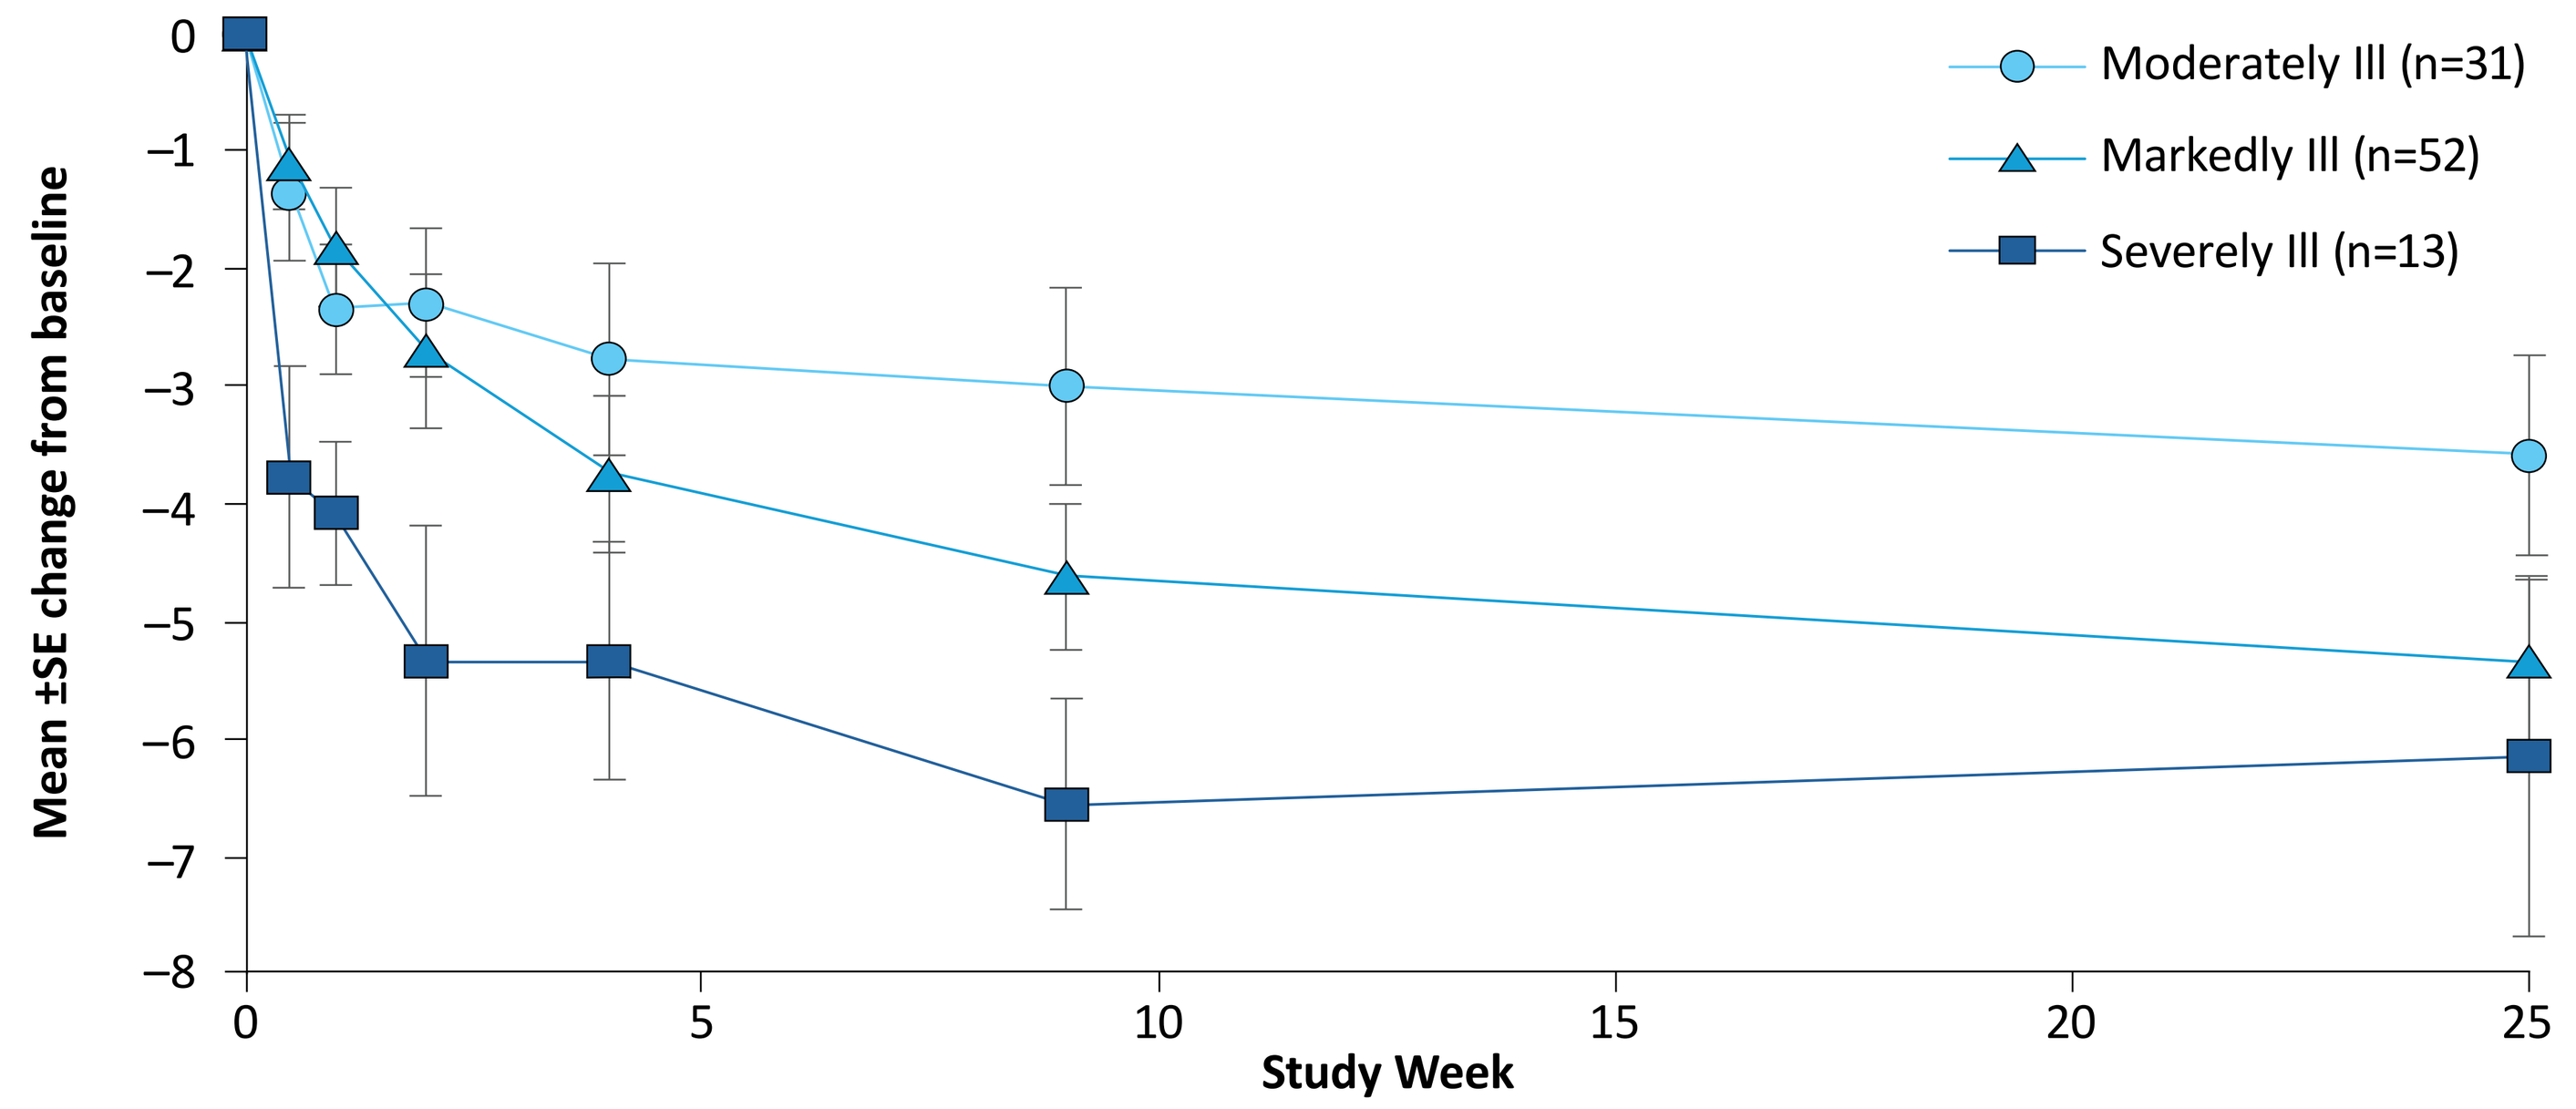
Moderately Ill (n=31)
Markedly Ill (n=52)
Severely Ill (n=13)
Mean ±SE change from baseline
0
−1
−2
−3
−4
−5
−6
−7
−8
0 5 10 15 20 25
Study Week

B. Paliperidone Palmitate

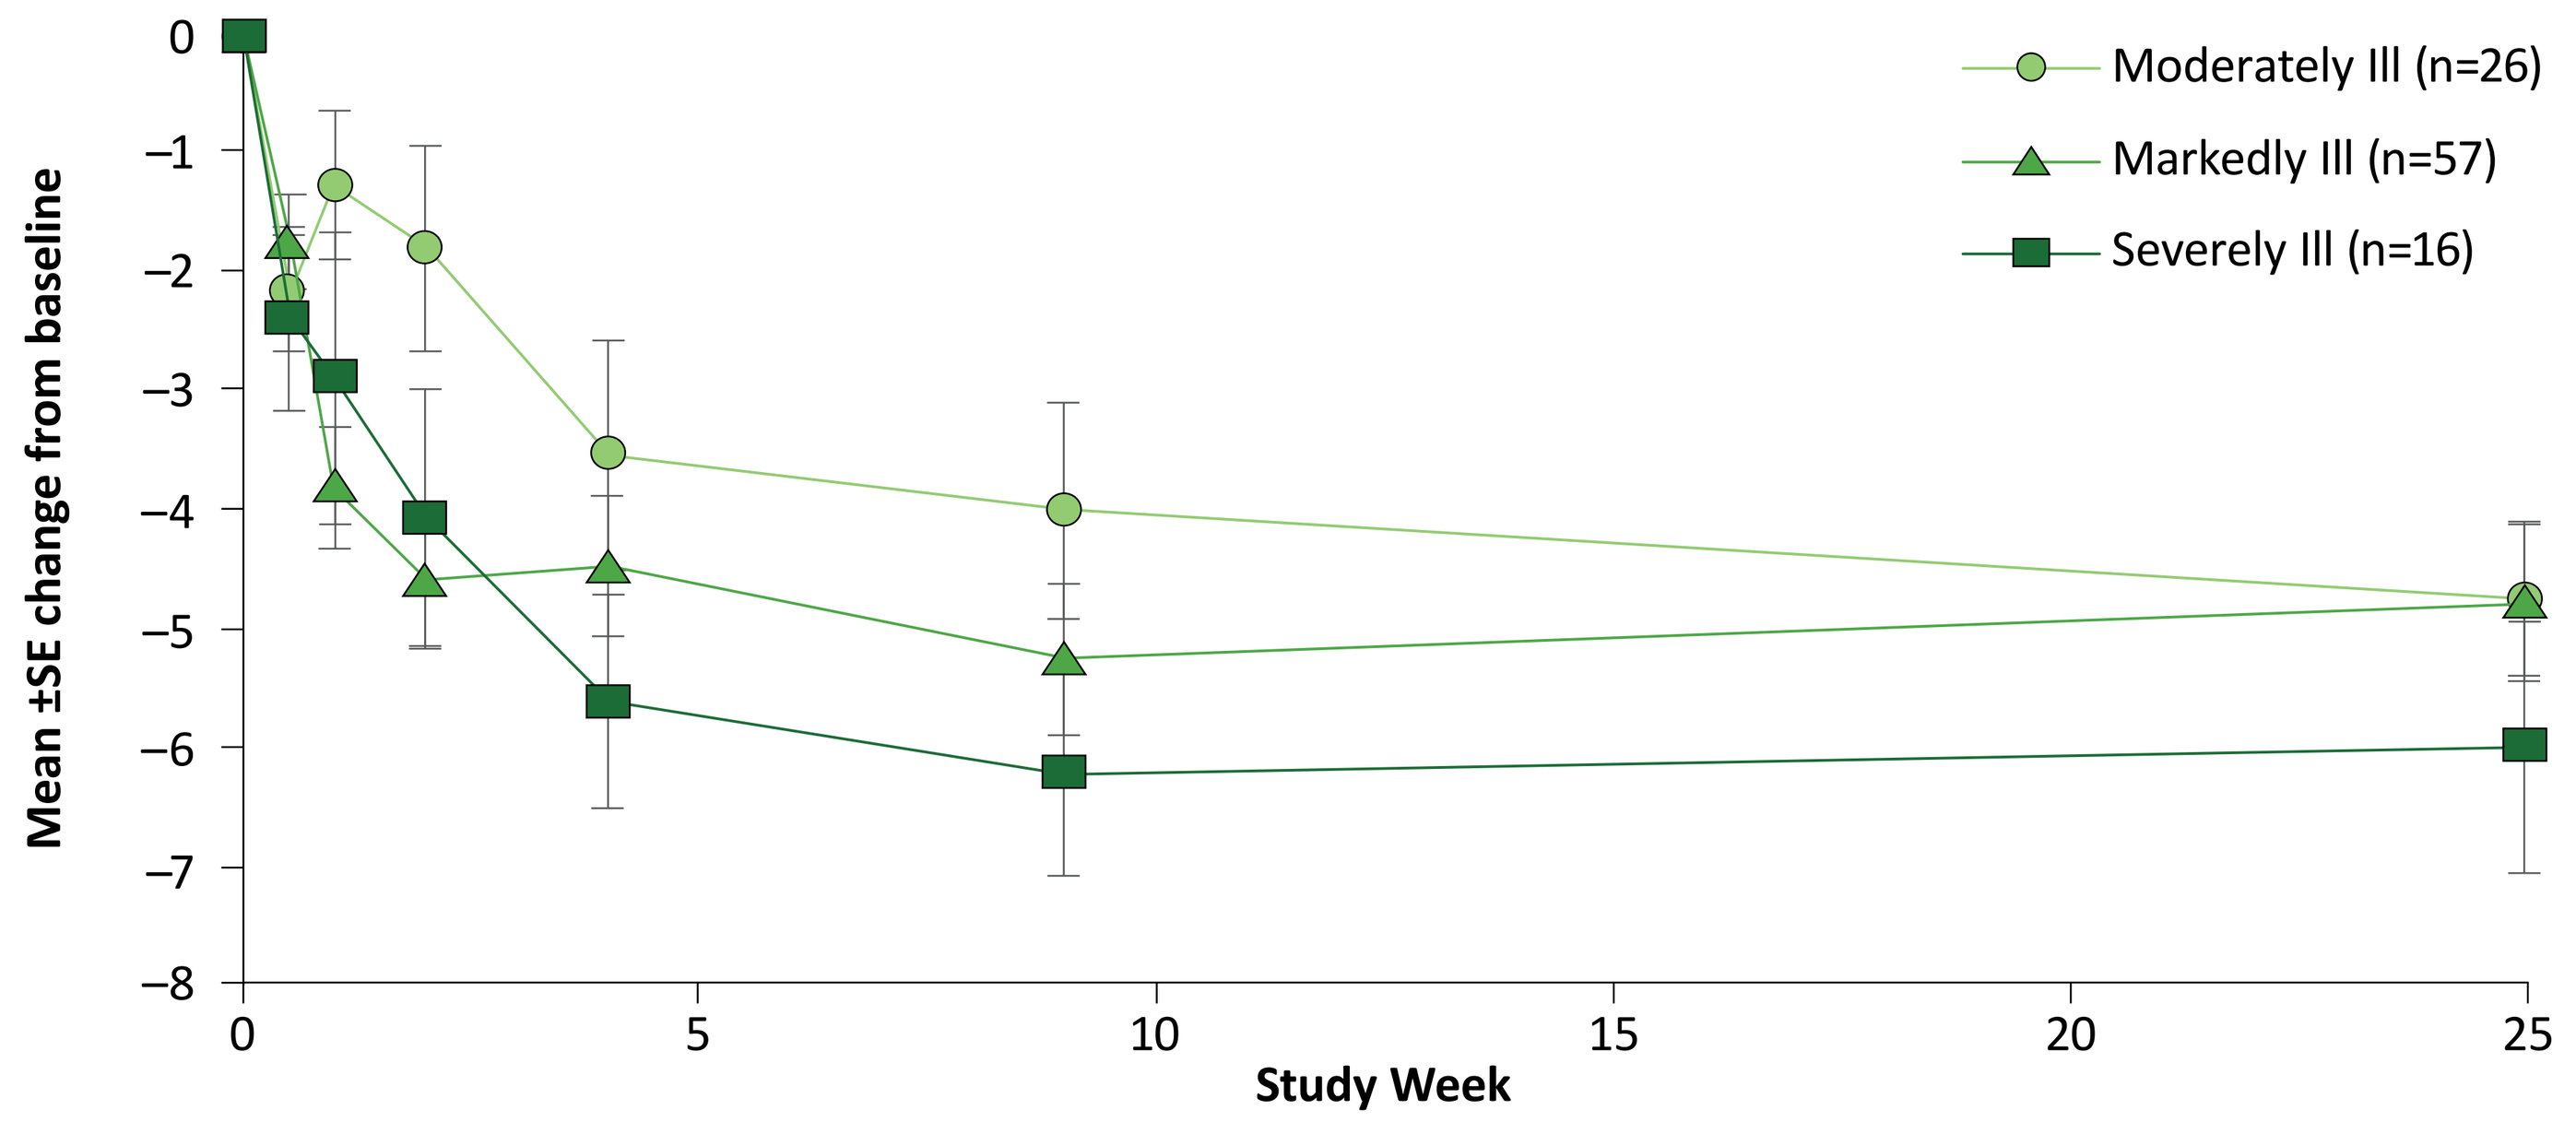
Moderately Ill (n=26)
Markedly Ill (n=57)
Severely Ill (n=16)
Mean ±SE change from baseline
0
−1
−2
−3
−4
−5
−6
−7
−8
0 5 10 15 20 25
Study Week

[a]Moderately ill, CGI-S=4; markedly ill, CGI-S=5; severely ill, CGI-S=6.
CGI-S, Clinical Global Impression–Severity; PANSS, Positive and Negative Syndrome Scale.

Selected Adverse Events

Table 2. AEs Related to Activation Factor Symptoms by Baseline Severity Group[a]

Aripiprazole Lauroxil			
AEs, n (%)	Moderately Ill (n=31)	Markedly Ill (n=54)	Severely Ill (n=14)
Anxiety	2 (6.5)	0	0
Agitation	0	2 (3.7)	1 (7.1)
Insomnia	1 (3.2)	1 (1.9)	0
Paliperidone Palmitate			
AEs, n (%)	Moderately Ill (n=26)	Markedly Ill (n=59)	Severely Ill (n=16)
Anxiety	4 (15.4)	0	3 (18.8)
Agitation	0	1 (1.7)	0
Insomnia	1 (3.8)	1 (1.7)	1 (6.3)

[a]Moderately ill CGI-S=4; markedly ill, CGI-S=5; severely ill, CGI-S=6.
AE, adverse event; CGI-S, Clinical Global Impression–Severity.

LIMITATIONS

- Patients were grouped according to their baseline CGI-S score; CGI-S scores are less precise and may under- or overestimate symptom severity compared with a full PANSS assessment
 - There was overlap in PANSS Total scores across severity groups, particularly among patients in the moderately and markedly ill categories; however, the mean baseline PANSS Total score was higher in each successive severity category
- Numbers of patients in some of the severity-of-illness subgroups were small, which may limit interpretability of the results
- The ALPINE study was not powered for a direct comparison between the AL and PP treatment groups; the PP arm provided an active control with known efficacy
- Results from the ALPINE study may not be generalizable to the real-world population of patients with schizophrenia who are started on LAI antipsychotic treatment

CONCLUSIONS

- In this post hoc analysis of data from the ALPINE study, acutely ill patients with schizophrenia who initiated AL or PP experienced improvement in schizophrenia symptoms, as measured by changes in PANSS Total scores, regardless of severity of illness at baseline
 - Change from baseline in PANSS Total score at week 25 by baseline severity group was comparable with that observed in the overall population
- Improvement from baseline in PANSS Activation factor score with AL or PP treatment was also observed among patients with moderate, marked, or severe symptoms at baseline
- For AEs of anxiety, agitation, and insomnia, no clear pattern of occurrence across baseline severity groups was observed with AL or PP treatment

REFERENCES

1. Olivares JM, Fagiolini A. *Front Psychiatry*. 2022;13:877867. DOI: 10.3389/fpsyt.2022.877867
2. Lindenmayer JP, et al. *J Clin Psychiatry*. 2009;70(7):990-6. DOI: 10.4088/JCP.08m04221
3. Weiden PJ, et al. *J Clin Psychiatry*. 2020;81(3):19m13207. DOI: 10.4088/JCP.19m13207
4. Pandina GJ, et al. *J Clin Psychopharmacol*. 2010;30(3):235-44. DOI: 10.1097/JCP.0b013e3181dd3103
5. Kay SR, et al. *Schizophr Bull*. 1987;13(2):261-76. DOI: 10.1093/schbul/13.2.261
6. Guy W. ECDEU Assessment Manual for Psychopharmacology. Rockville, MD: US Department of Health, Education, and Welfare, National Institute of Mental Health; 1976:217-22.
7. Van den Oord EJ, et al. *Schizophr Res*. 2006;82(2-3):213-23. DOI: 10.1016/j.schres.2005.09.002
8. Kumar A, Khess CR. *Indian J Psychiatry*. 2012;54(3):233-8. DOI: 10.4103/0019-5545.102419

Disclosure information, acknowledgments, and copies of this poster can be obtained through this QR (Quick Response) code. These materials are for personal use only and may not be reproduced without permission of Alkermes. For permission, contact USMedInfo@Alkermes.com

Supported by funding from Alkermes, Inc.

Baseline Severity of Illness and Response to Treatment With Aripiprazole Lauroxil Every 2 Months: A Post Hoc Analysis of Phase 3 ALPINE Clinical Trial Data

John M. Kane, MD[1]; Martin Dunbar, PhD[2]; and James A. McGrory, PhD[2]

[1]The Donald and Barbara Zucker School of Medicine at Hofstra/Northwell, Hempstead, NY, USA; [2]Alkermes, Inc., Waltham, MA, USA

DISCLOSURES

JMK has been a consultant for or received honoraria from Alkermes, Allergan, Boehringer Ingelheim, Cerevel, Click Therapeutics, Dainippon Sumitomo, HLS, Indivior, Intra-Cellular Therapies, Janssen, Johnson & Johnson, Karuna, LB Pharmaceuticals, Lundbeck, Merck, Minerva, Neumora, Neurocrine Biosciences, Newron, Novartis, Otsuka, Reviva, Roche, Saladax Biomedical, Sunovion, Takeda, and Teva; has received grant support from Janssen, Lundbeck, Otsuka, and Sunovion; and is a shareholder of LB Pharmaceuticals and the Vanguard Research Group.

MD and **JAM** are or were employees of Alkermes, Inc., and may own stock/options in the company.

ACKNOWLEDGMENTS

This study was sponsored by Alkermes, Inc. (Waltham, MA, USA). Medical writing and editorial support were provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Alkermes, Inc.

Supported by funding from Alkermes, Inc.