Impact of Economic Status Across Race and Ethnicity on Narcolepsy Treatment Access: A Claims Database Analysis

Rakesh Bhattacharjee, MD[1]; Alec B. Platt, MD, MSCE[2]; Heidy Merius, DNP[3,4]; Omavi Bailey, MD, MPH[5]; Jennifer Gudeman, PharmD[6,a]; Johanna Hertel, PhD[7]; Brian Abaluck, MD[7]

[1]University of California, San Diego, San Diego, CA, USA; [2]Respiratory Specialists, Wyomissing, PA, USA; [3]Stony Brook Medicine, Stony Brook, NY, USA; [4]Sleep Medicine Advanced Practice Providers (SMAPP) Secretary and Treasurer, East Hanover, NJ, USA; [5]Biorhythms, El Paso, TX, USA; [6]Avadel Pharmaceuticals, Chesterfield, MO, USA; [7]Alkermes, Inc., Waltham, MA, USA*; [a]At the time of the study

Poster 317

INTRODUCTION

- Narcolepsy is a rare, chronic sleep disorder characterized by excessive daytime sleepiness (EDS) that impacts patients across economic status, race, and ethnicity[1-3]
- Narcolepsy symptom management has included medications approved by the US Food and Drug Administration (FDA; eg, oxybates and wake-promoting agents)[4-11]
- The 2021 American Academy of Sleep Medicine (AASM) clinical practice guidelines included several recommendations for symptom management in patients with narcolepsy[12]:
 - 4 strong recommendations for the treatment of EDS (modafinil, pitolisant, sodium oxybate [SXB], and solriamfetol)
 - 2 strong recommendations for the treatment of cataplexy (pitolisant and SXB)
 - Conditional recommendations for armodafinil, dextroamphetamine, and methylphenidate for the treatment of EDS and/or cataplexy
- There are 3 formulations of oxybate available in the US: immediate-release SXB, immediate-release mixed-salt oxybates, and extended-release SXB[6-7]
- As more therapies become available, it is important to understand medication usage patterns in people with narcolepsy, including across diverse populations

OBJECTIVE

- To describe narcolepsy treatment patterns among patients of different races and ethnicities and income levels

METHODS

DATA SOURCE AND PATIENT POPULATION

- Retrospective analysis of data from the Komodo Health claims database, which comprises data from >330 million individuals in the United States
 - The closed claims data set was used to identify patients with narcolepsy type 1 (NT1) or type 2 (NT2) in 2023
- The patient population was narrowed by defining a high-confidence diagnostic subset (**Table**)
 - High-confidence diagnosis was defined as ≥2 relevant *International Classification of Diseases, 10th Revision* (ICD-10) codes documented ≥30 days apart, with the first of such claims prior to 2023
 - The diagnosis of a different narcolepsy type or idiopathic hypersomnia (IH) was exclusionary
- Treatment was defined as any paid prescription claim linked to a patient in the study cohort with National Drug Codes corresponding to narcolepsy treatments after receiving a diagnosis code
- Self-reported race or ethnicity data and national income data were required

DATA ANALYSIS

- The proportions of patients who received the following narcolepsy treatments were calculated:
 - Strongly recommended narcolepsy treatment per 2021 AASM clinical practice guidelines (modafinil, pitolisant, solriamfetol, and SXB; other oxybates; and armodafinil)
 - Oxybate treatment
 - No indicated treatment for EDS
- All data were stratified by race and ethnicity (Black or African American, Hispanic or Latino, and white), and income level (defined based on national income percentile: lower, 0%-30%; middle, 40%-60%; upper, 70%-90%) and reported descriptively
 - Two-sided z-tests were used to compare Black or African American, or Hispanic or Latino categories to the reference category (white) within each income band for each treatment category
 - Due to very small sample size, patients who did not self-report as Black or African American, Hispanic or Latino, or white were excluded from the analysis

TABLE: Inclusion and Exclusion Criteria for High-Confidence Diagnostic Subset

Inclusion criteria	Exclusion criteria
• Exactly 1 NT1 or NT2 diagnosis prior to 2023: – ≥2 claims with a relevant ICD-10 code ≥30 days apart, with the first such claim prior to 2023: ▪ NT1: G47.411, G47.421 ▪ NT2: G47.419, G47.429 • 100% claim availability (continuously enrolled) in 2023 • ≥1 claim for the diagnosed narcolepsy type in 2023	• Multiple distinct diagnoses of IH, NT1, or NT2 at any time • No medical or prescription claims for the diagnosed narcolepsy type in 2023

ICD-10, *International Classification of Diseases, 10th Revision*; IH, idiopathic hypersomnia; NT1, narcolepsy type 1; NT2, narcolepsy type 2.

RESULTS

PATIENT CHARACTERISTICS

- The Komodo closed claims data set included 30,563 individuals with an NT1 or NT2 diagnosis in 2023; of these, 16,389 had race or ethnicity data and income data available (**Figure 1**)

FIGURE 1: High-Confidence Diagnosis Cohort

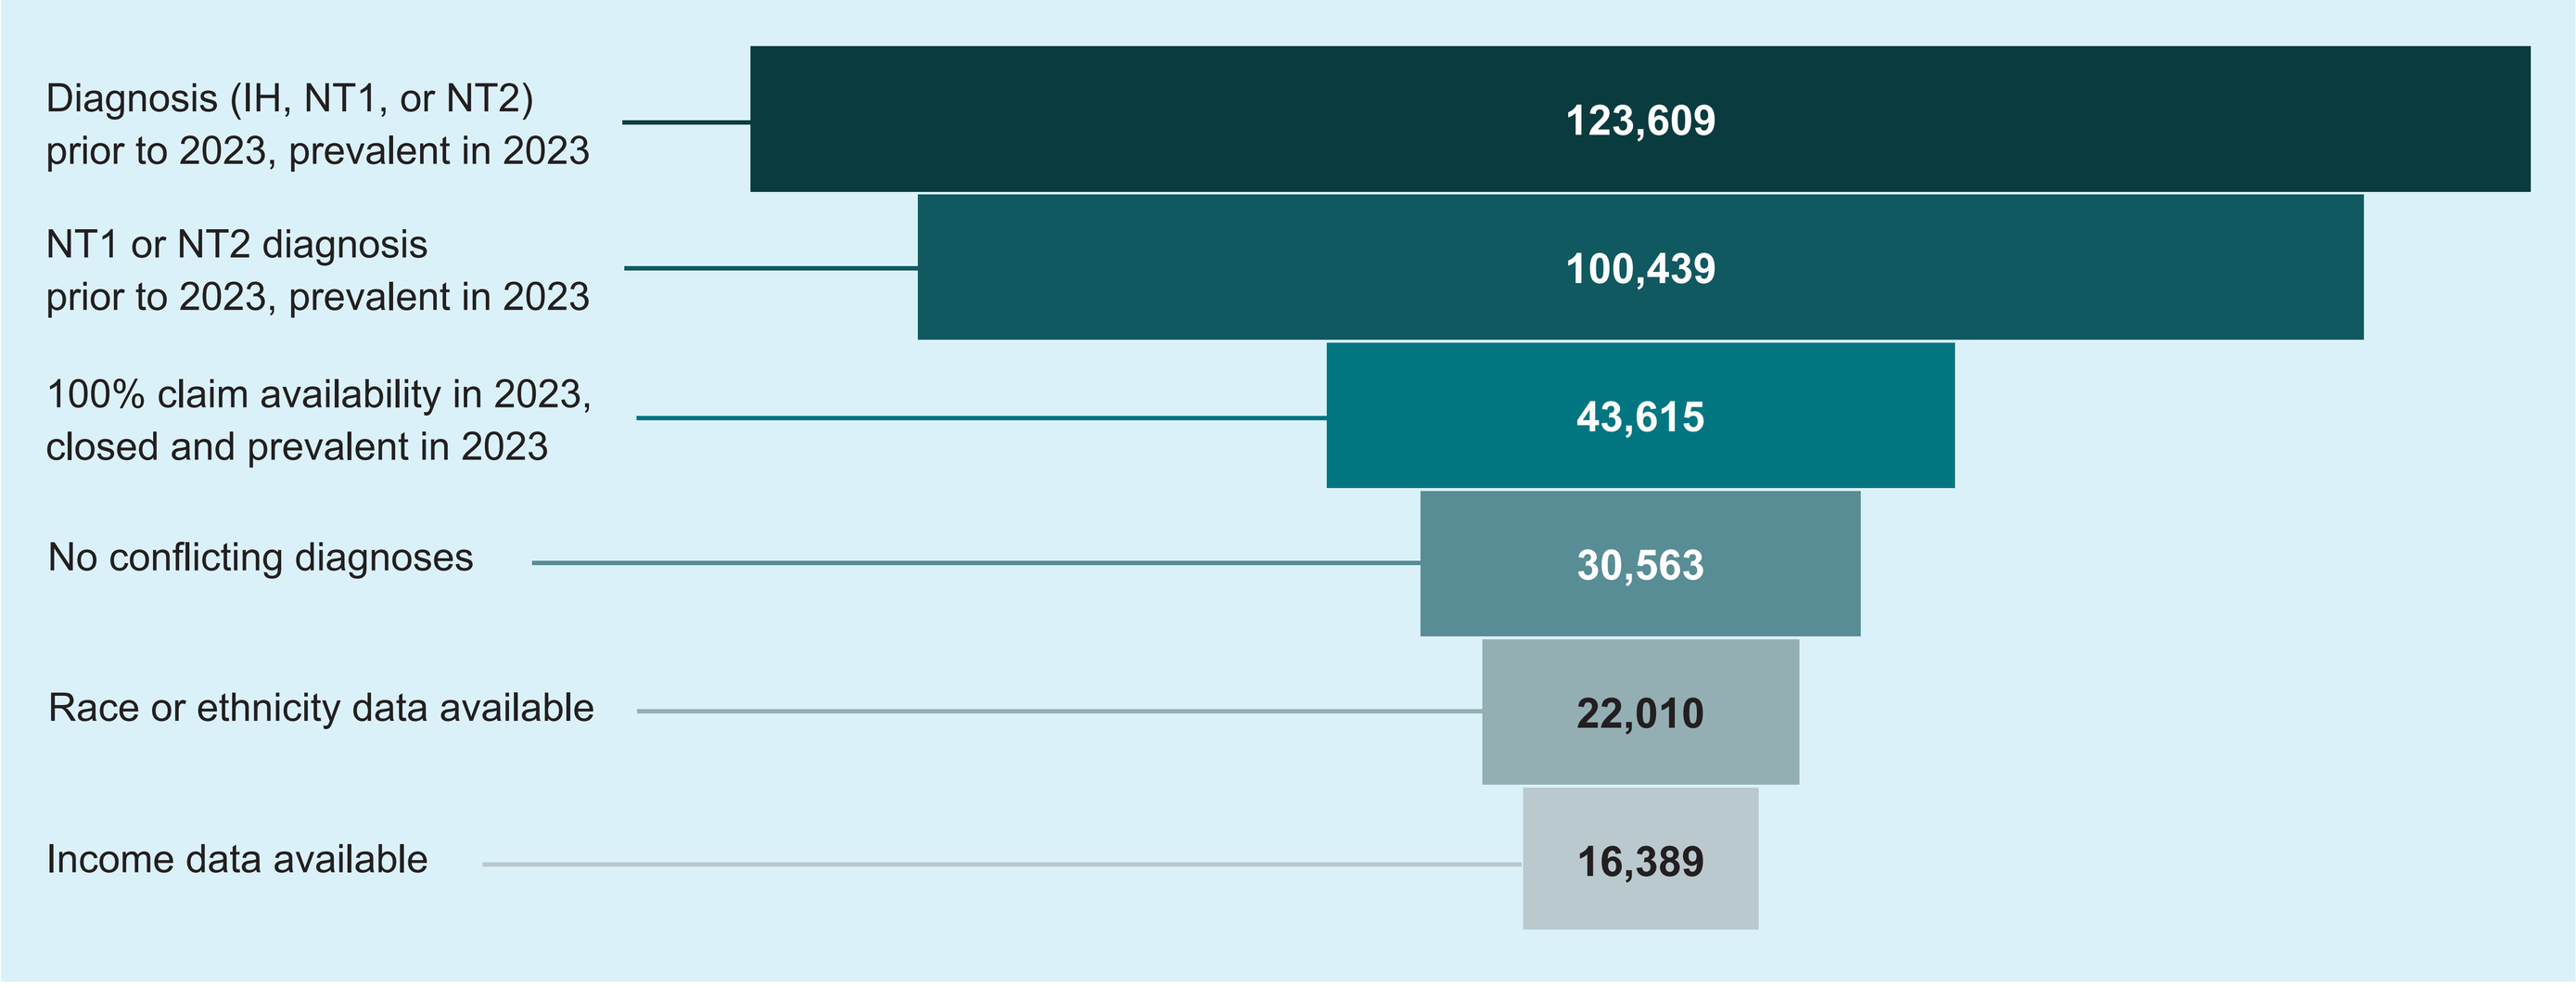
Diagnosis (IH, NT1, or NT2) prior to 2023, prevalent in 2023
123,609
NT1 or NT2 diagnosis prior to 2023, prevalent in 2023
100,439
100% claim availability in 2023, closed and prevalent in 2023
43,615
No conflicting diagnoses
30,563
Race or ethnicity data available
22,010
Income data available
16,389

IH, idiopathic hypersomnia; NT1, narcolepsy type 1; NT2, narcolepsy type 2.

- The majority of patients had an income within the lower and middle bands (69%) and were white (80%; **Figure 2**)

FIGURE 2: Patient Distribution by (A) Income Level and (B) Race or Ethnicity

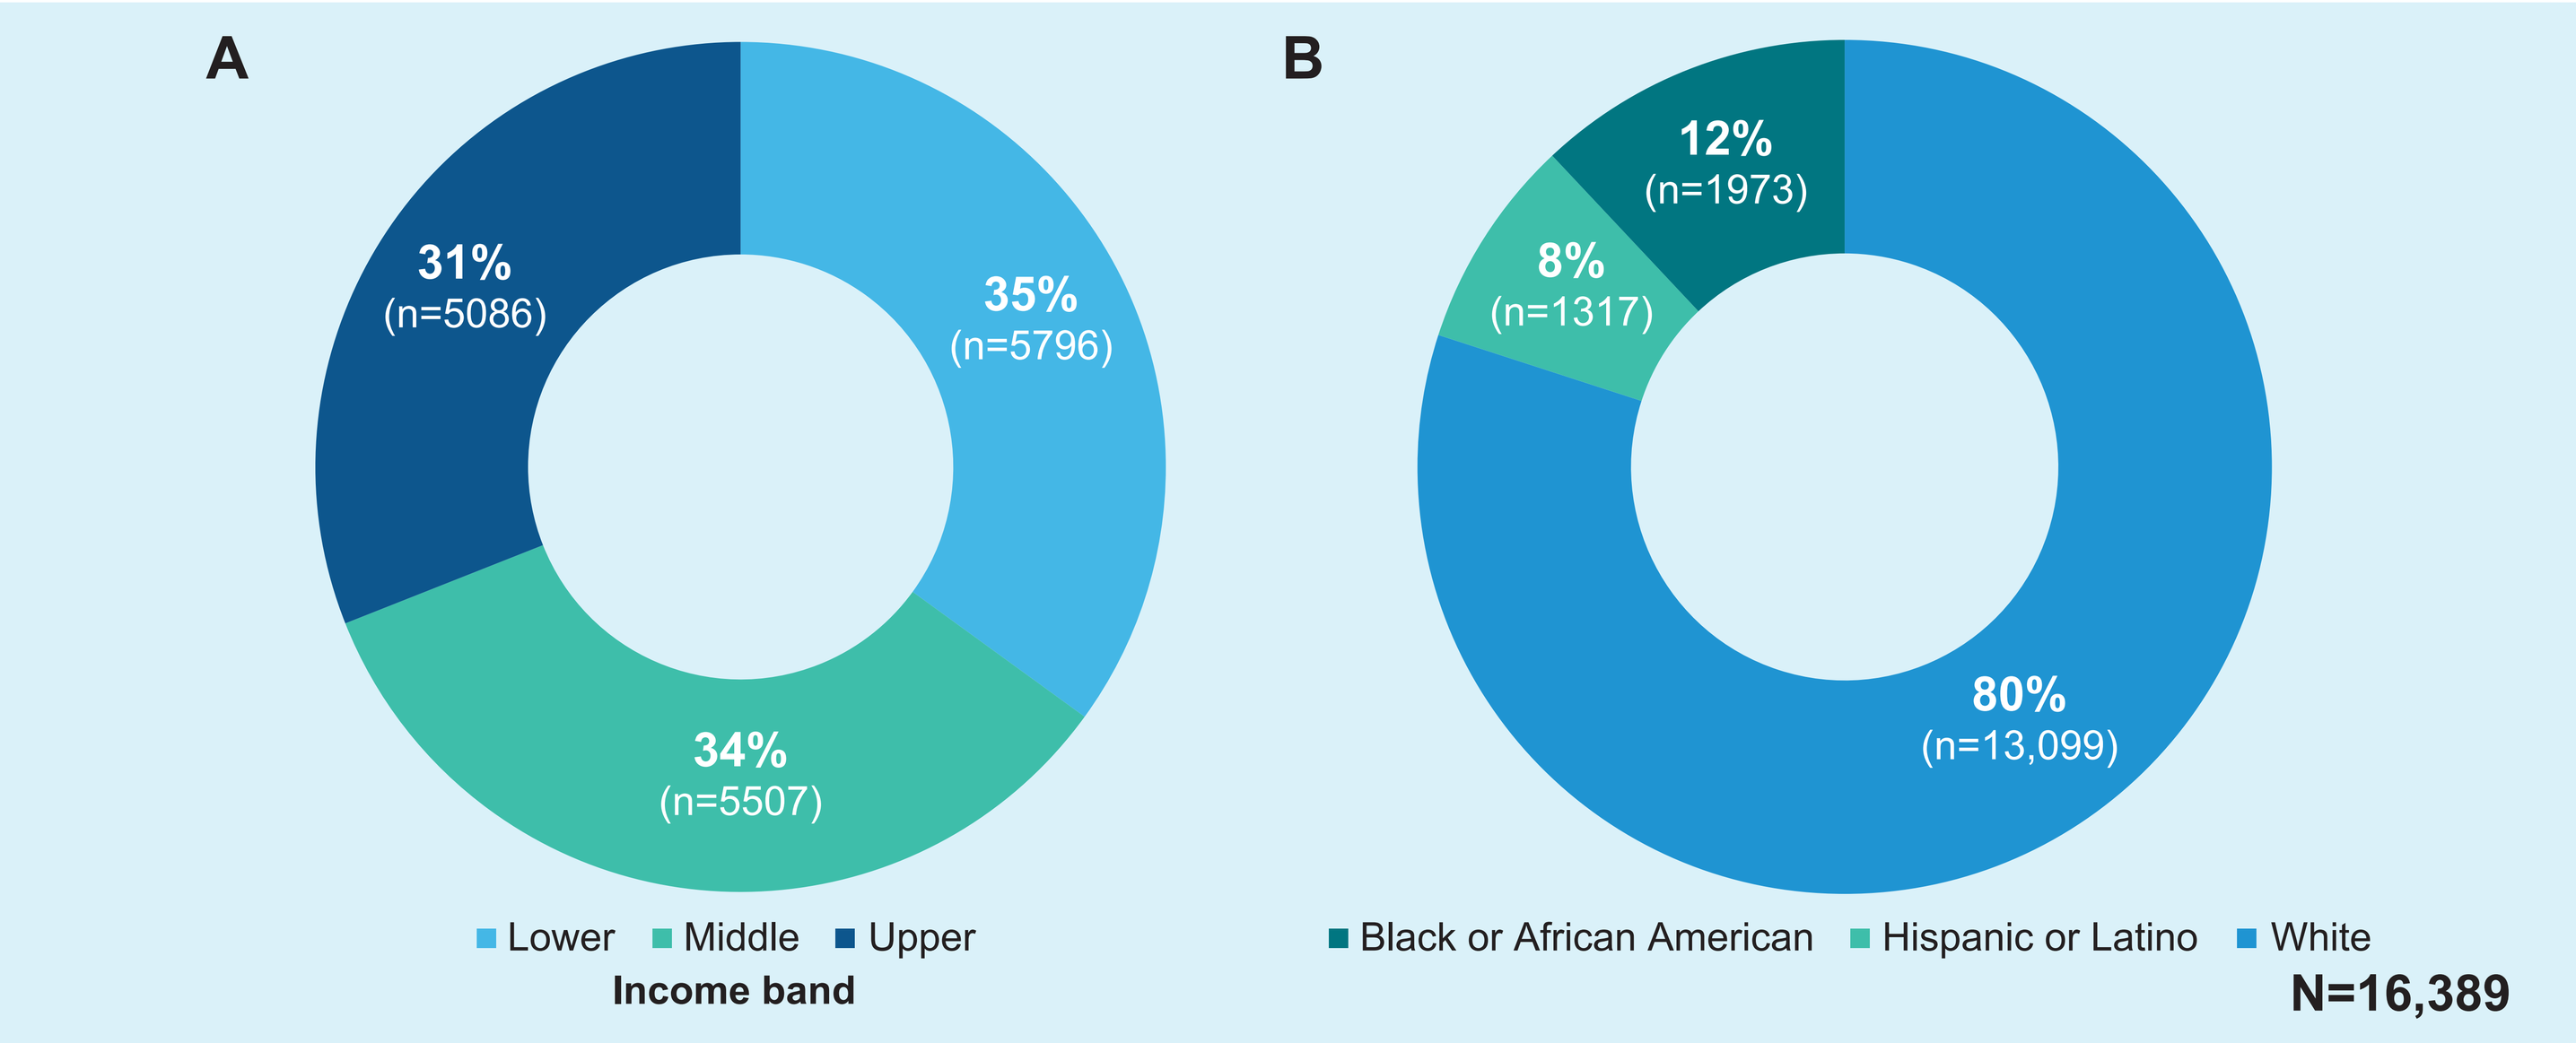
A
31% (n=5086)
35% (n=5796)
34% (n=5507)
Lower Middle Upper
Income band
B
12% (n=1973)
8% (n=1317)
80% (n=13,099)
Black or African American Hispanic or Latino White
N=16,389

NARCOLEPSY TREATMENT PATTERNS

- Overall, 48% (n=7903) of patients received a strongly recommended narcolepsy treatment in 2023 (**Figure 3**)
 - In the subset of those who received a strongly recommended treatment (n=7903), 16% (n=1259) were treated with oxybates
- 23% (n=3802) of patients did not receive any treatment for EDS

FIGURE 3: Narcolepsy Treatment Use

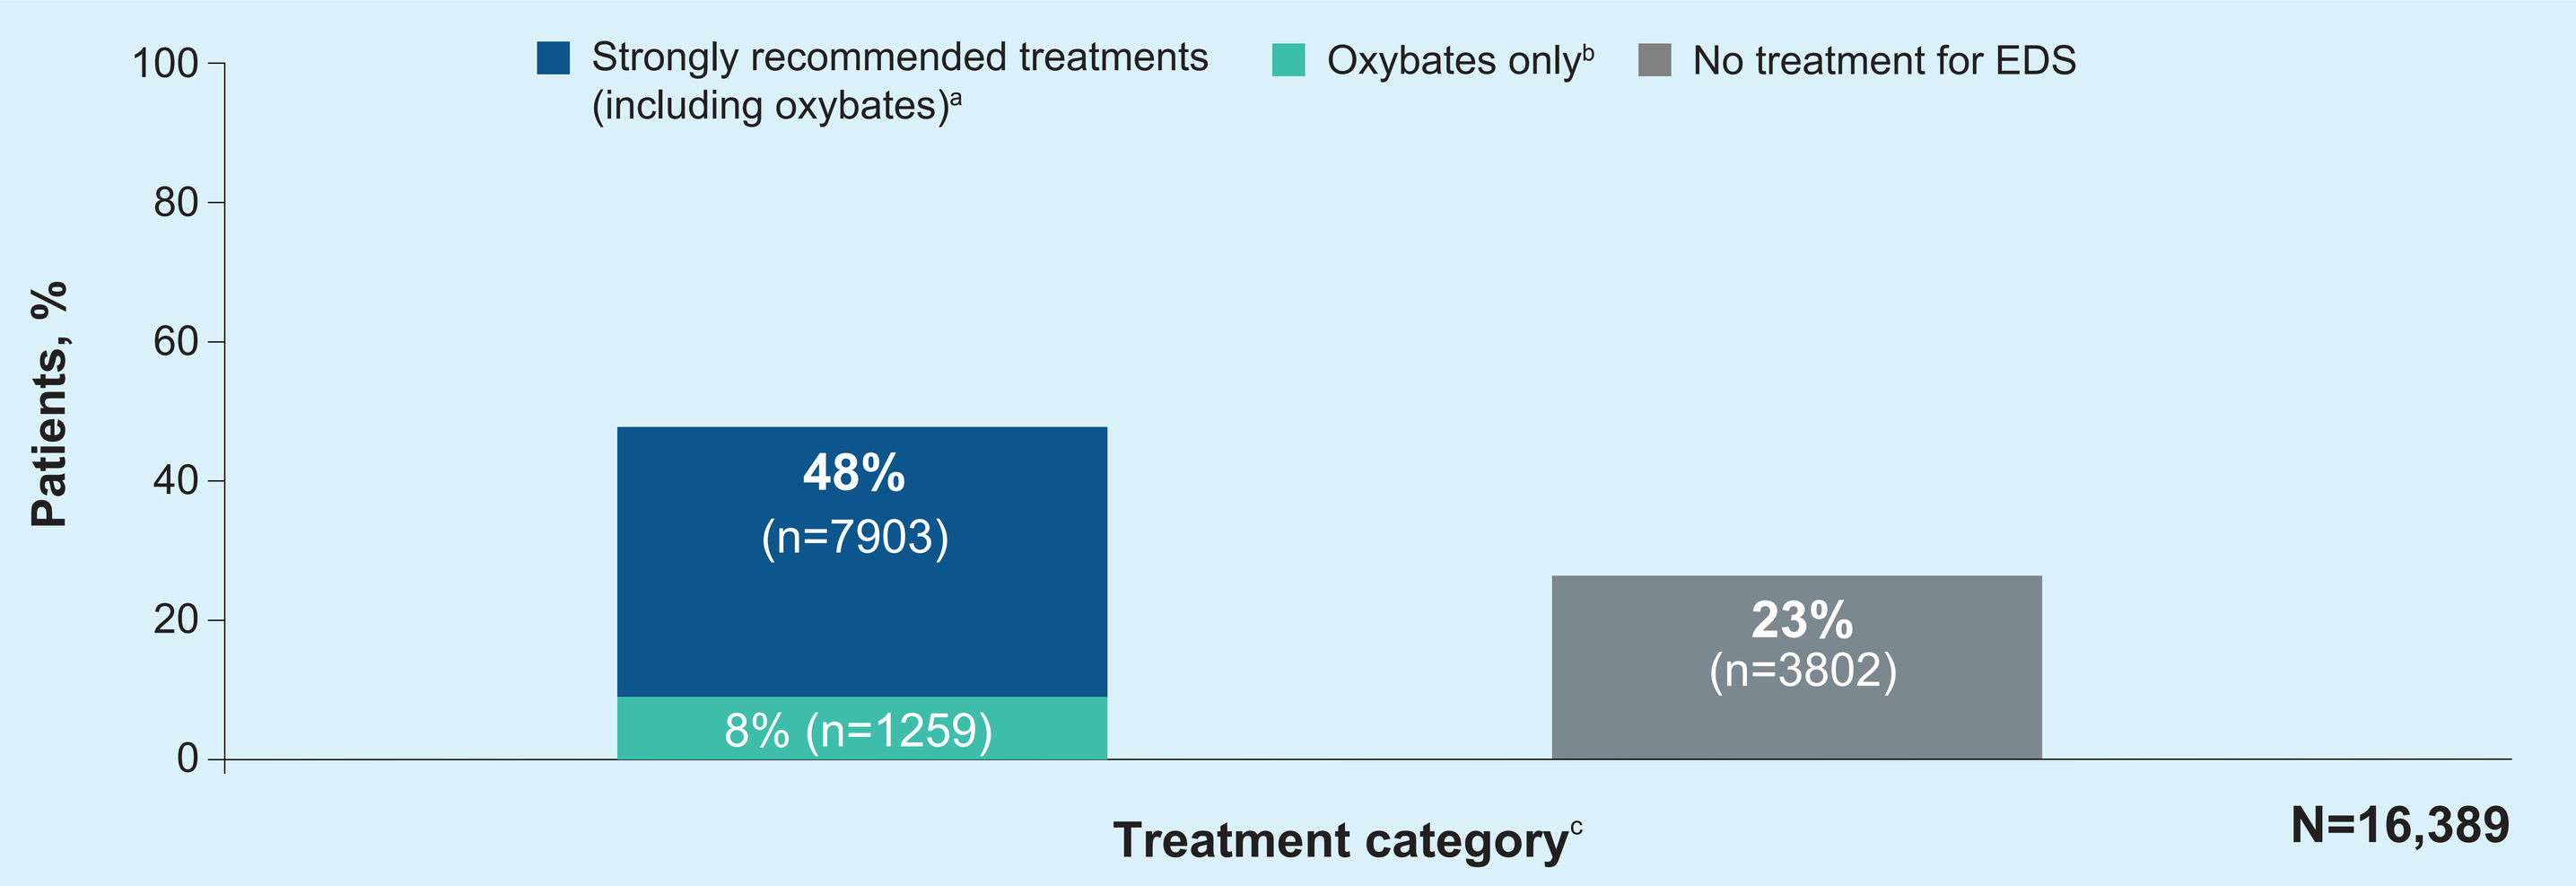
Strongly recommended treatments (including oxybates)[a]
Oxybates only[b]
No treatment for EDS
Patients, %
100
80
60
40
20
0
48% (n=7903)
8% (n=1259)
23% (n=3802)
Treatment category[c]
N=16,389

EDS, excessive daytime sleepiness.
[a]Strongly recommended treatments included modafinil, pitolisant, solriamfetol, armodafinil, and oxybates.
[b]Patients who received oxybates are a subset of those who received strongly recommended narcolepsy treatments.
[c]Patients receiving medications outside of those categorized as strongly recommended are not depicted.

- Across all income levels, Black or African American patients were numerically less likely to receive a strongly recommended narcolepsy treatment compared with patients of other races or ethnicities (**Figure 4**)
 - Within the lower and middle income bands, Black or African American patients were significantly less likely to receive a strongly-recommended treatment compared with white patients
- Receipt of strongly recommended narcolepsy treatments was numerically lowest among Black or African American and Hispanic or Latino patients in the lower income band

FIGURE 4: Strongly Recommended Narcolepsy Treatment Use by Race or Ethnicity and Income Level

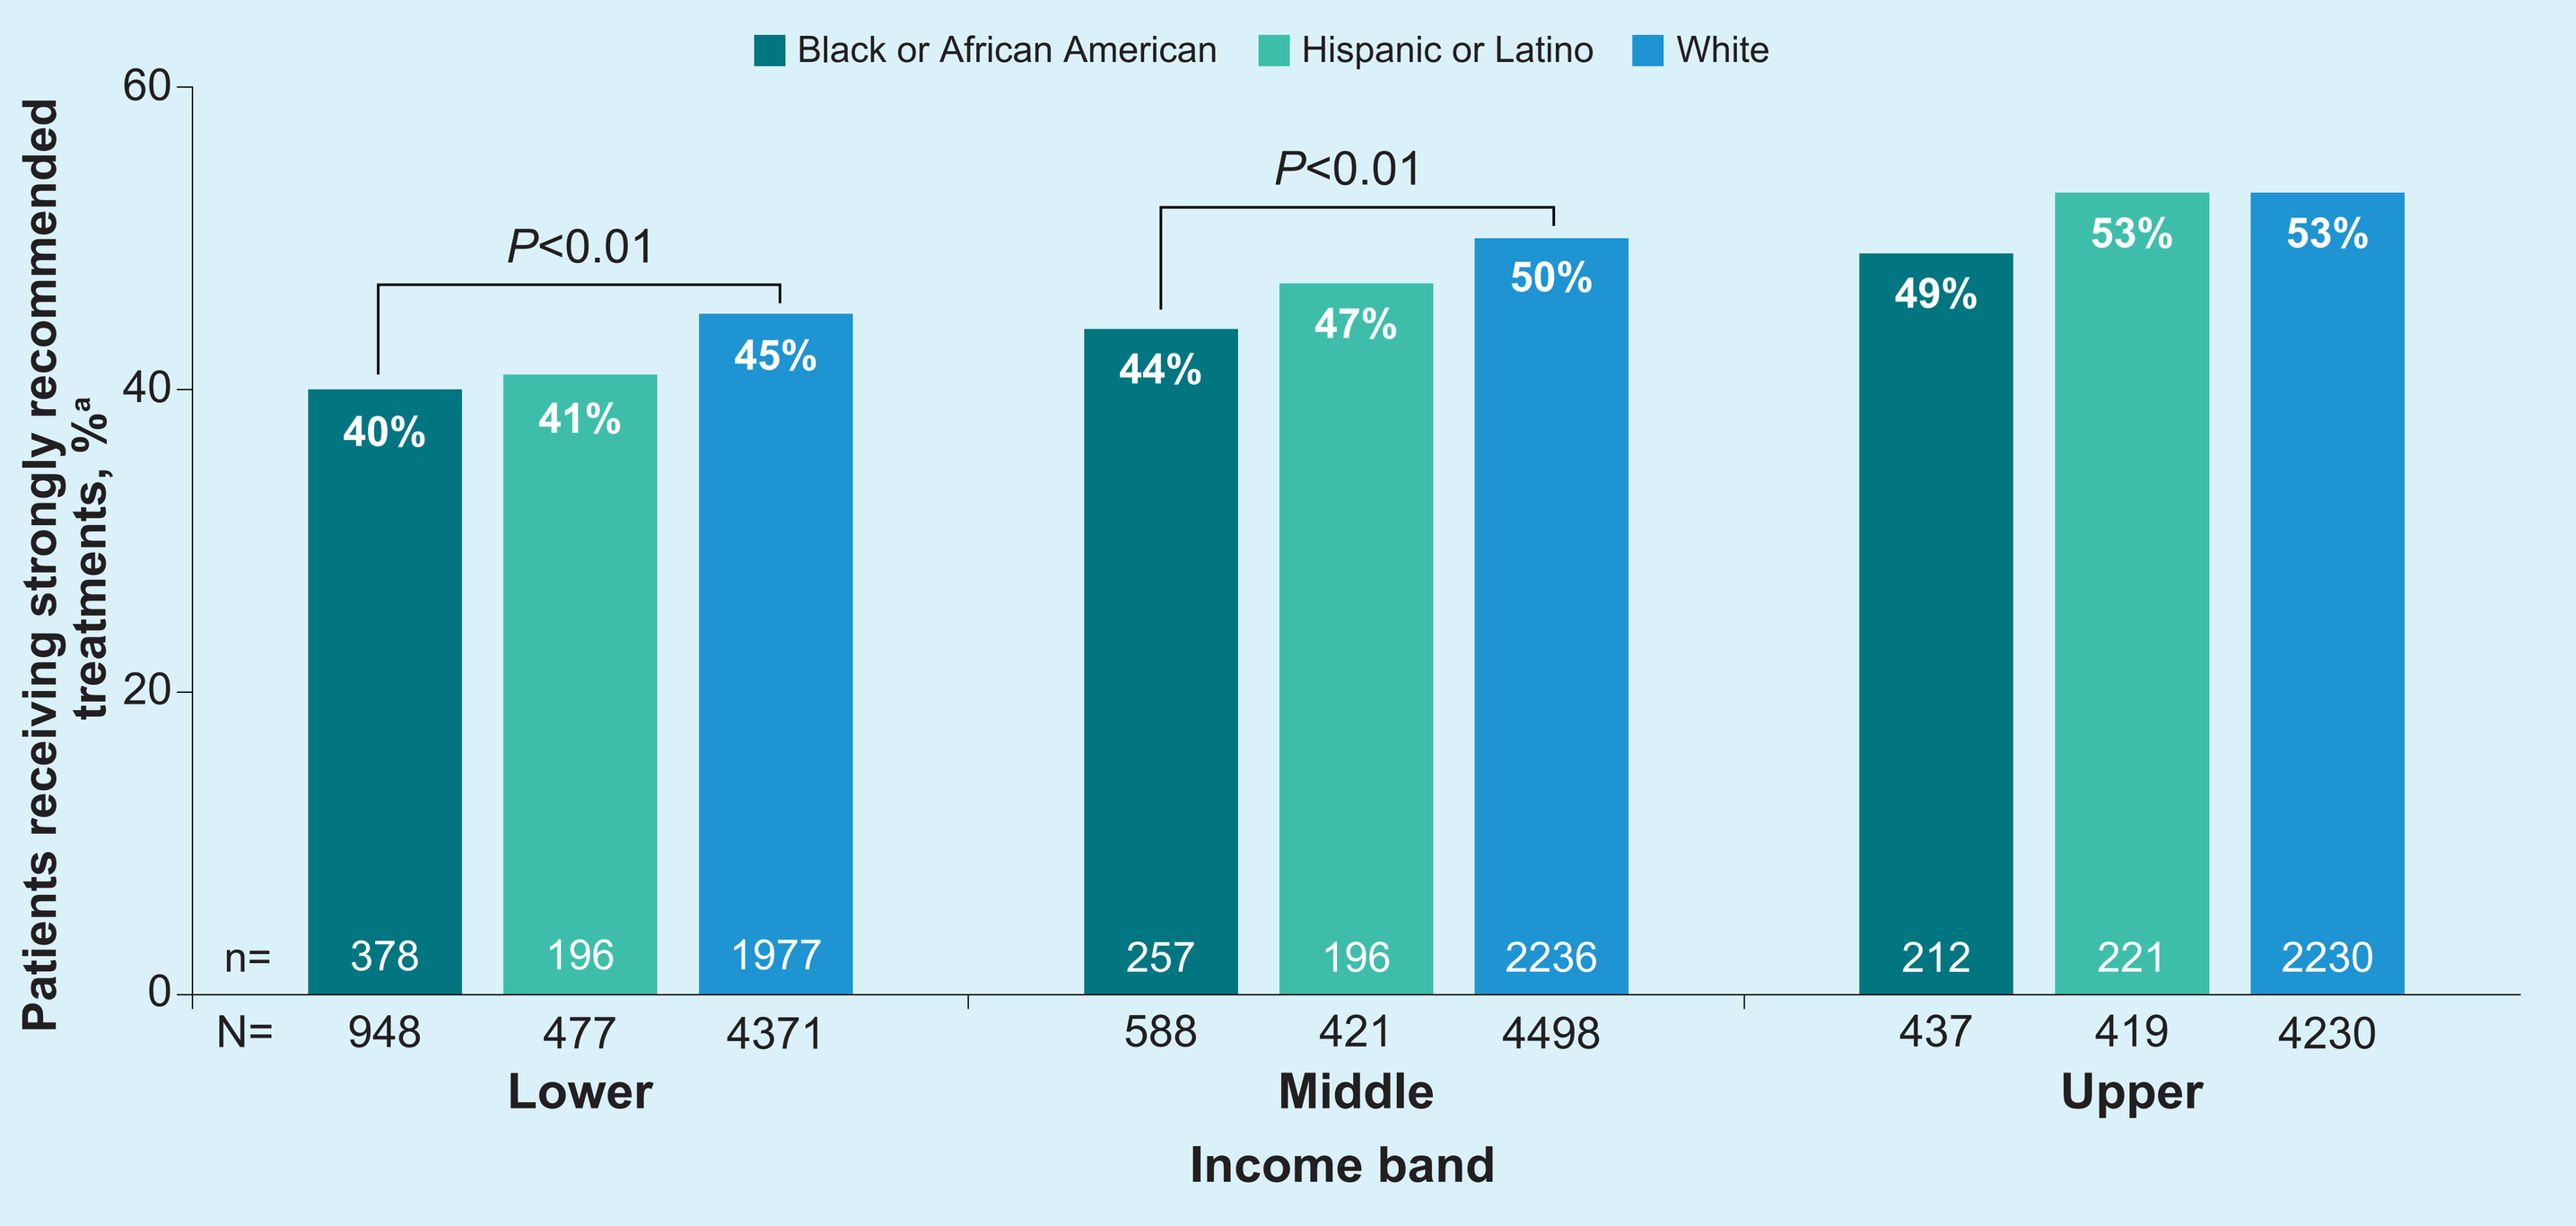
Black or African American Hispanic or Latino White
Patients receiving strongly recommended treatments, %[a]
60
40
20
0
P<0.01
40% 41% 45%
n= 378 196 1977
N= 948 477 4371
Lower
P<0.01
44% 47% 50%
257 196 2236
588 421 4498
Middle
49% 53% 53%
212 221 2230
437 419 4230
Upper
Income band

[a]Strongly recommended treatments included modafinil, pitolisant, solriamfetol, armodafinil, and oxybates.

- Within each race or ethnicity group, oxybate use was numerically lowest among patients with lower income levels (**Figure 5**)
- Within the lower and upper income bands, Black or African American patients were significantly less likely to be receiving oxybates compared with white patients

FIGURE 5: Oxybate Use by Race or Ethnicity and Income Level

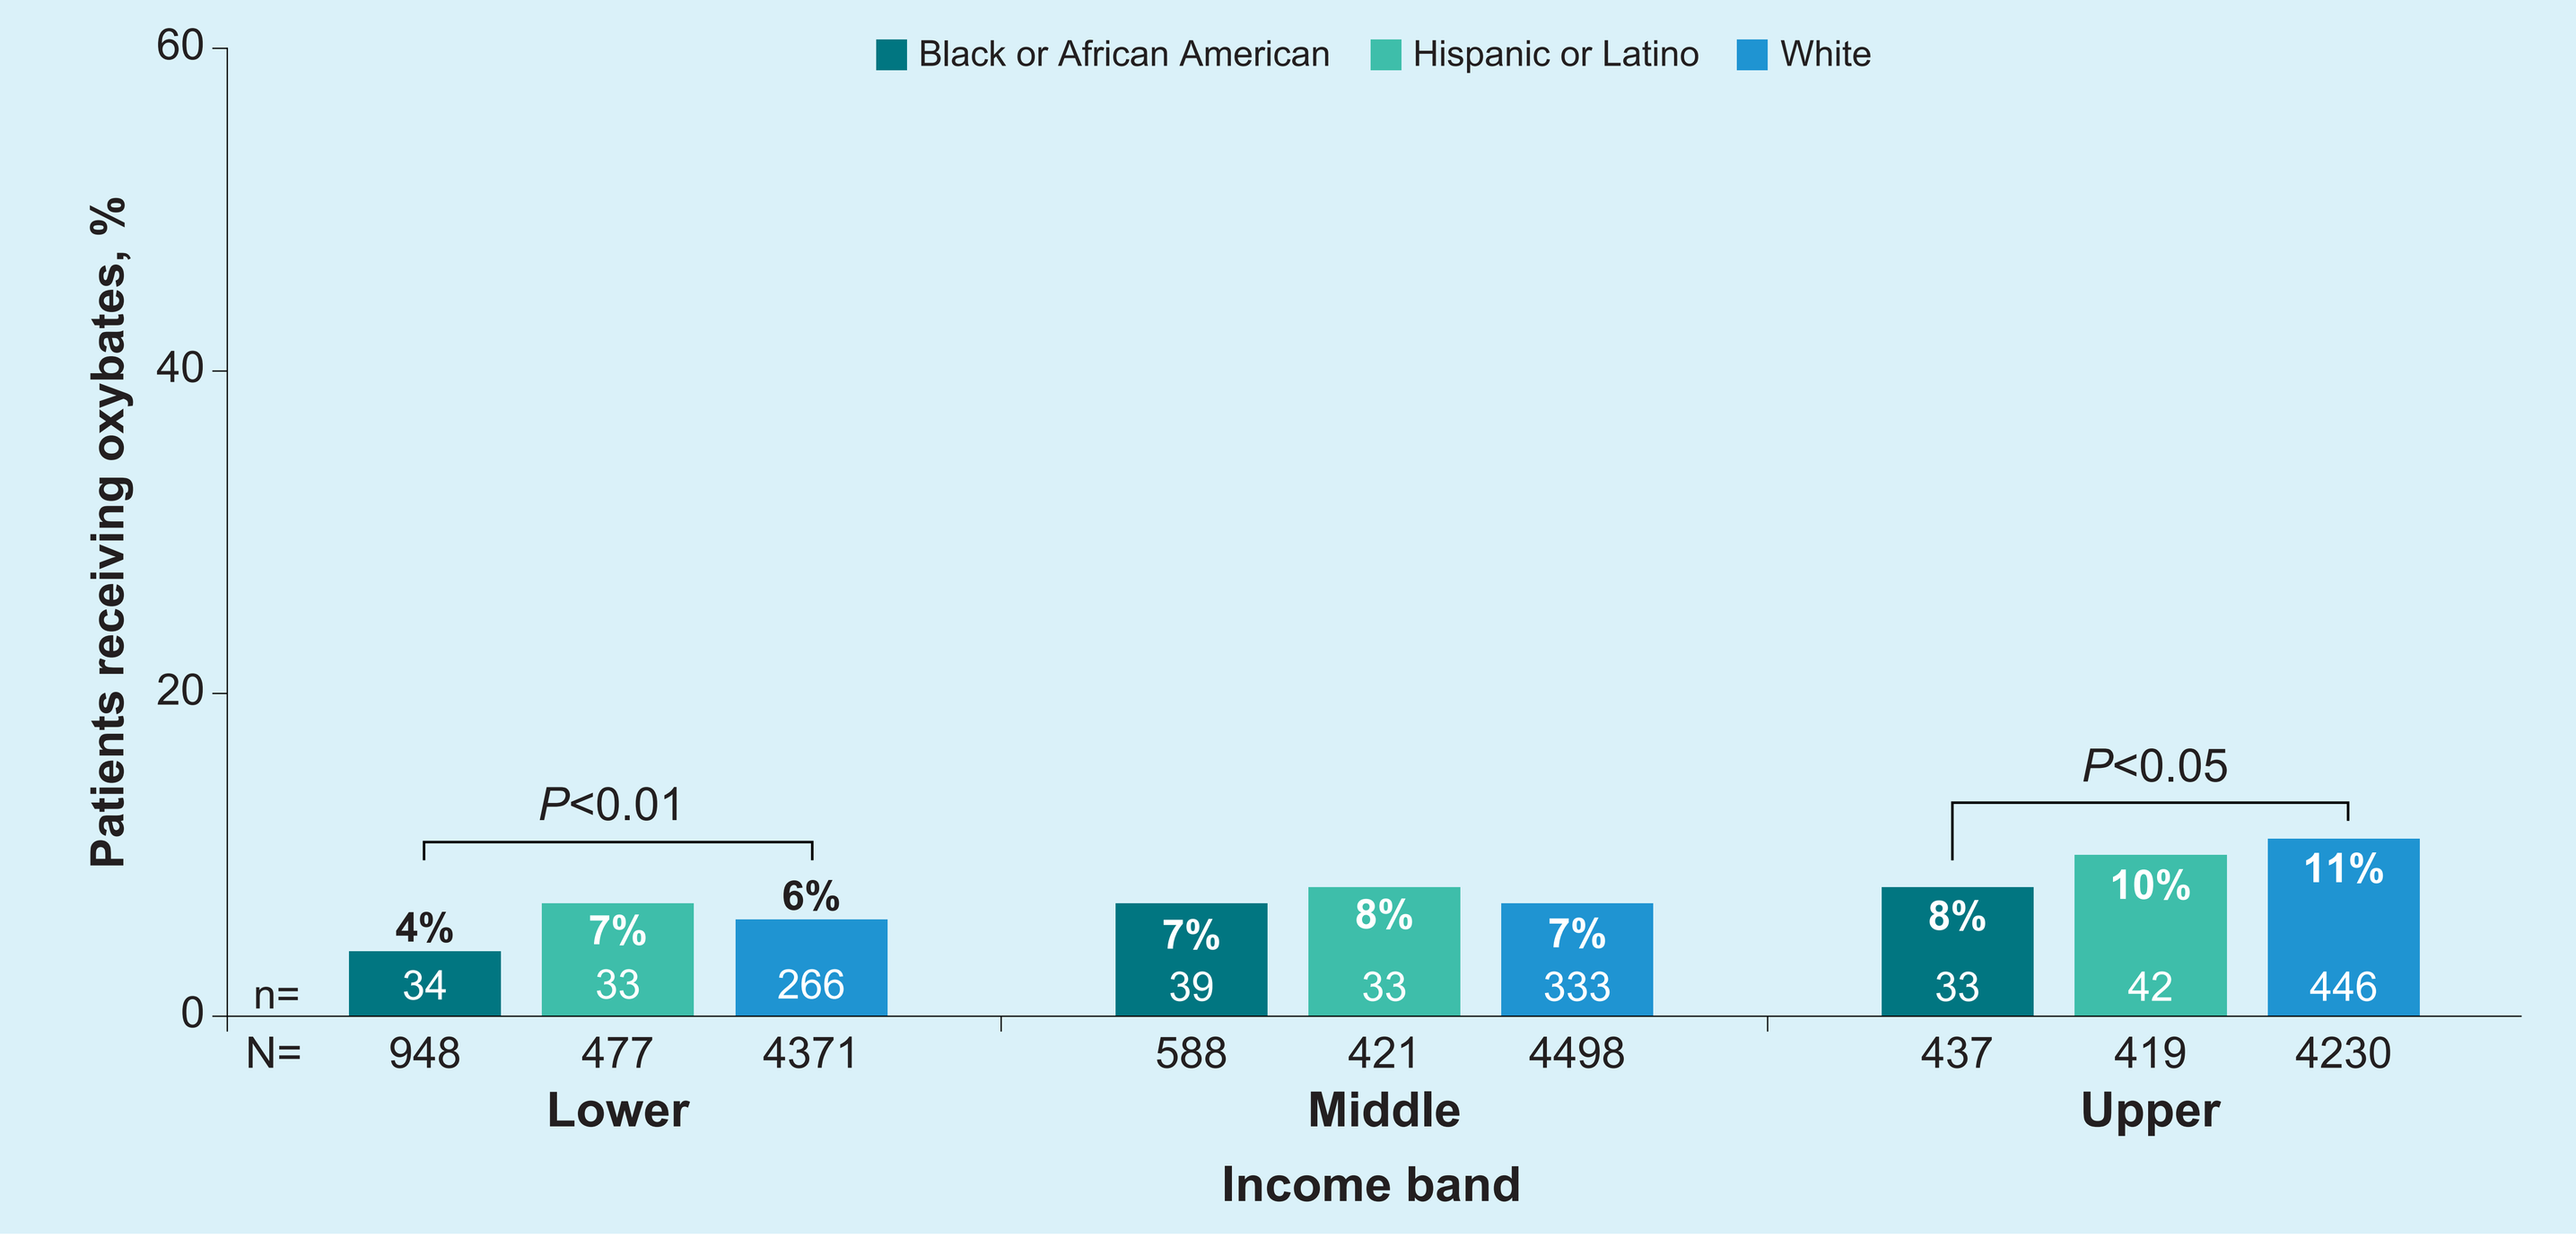
Black or African American Hispanic or Latino White
Patients receiving oxybates, %
60
40
20
0
P<0.01
4% 7% 6%
n= 34 33 266
N= 948 477 4371
Lower
7% 8% 7%
39 33 333
588 421 4498
Middle
P<0.05
8% 10% 11%
33 42 446
437 419 4230
Upper
Income band

- Across all income levels, Black or African American patients were significantly less likely to receive a treatment for EDS compared with white patients (**Figure 6**)
 - Within the lower and middle income bands, Hispanic or Latino patients were significantly less likely to receive a narcolepsy treatment compared with white patients
- Within each race or ethnicity group, the proportion of patients not receiving a narcolepsy treatment increased as income level decreased

FIGURE 6: Patients Not Receiving Treatment for EDS by Race or Ethnicity and Income Level

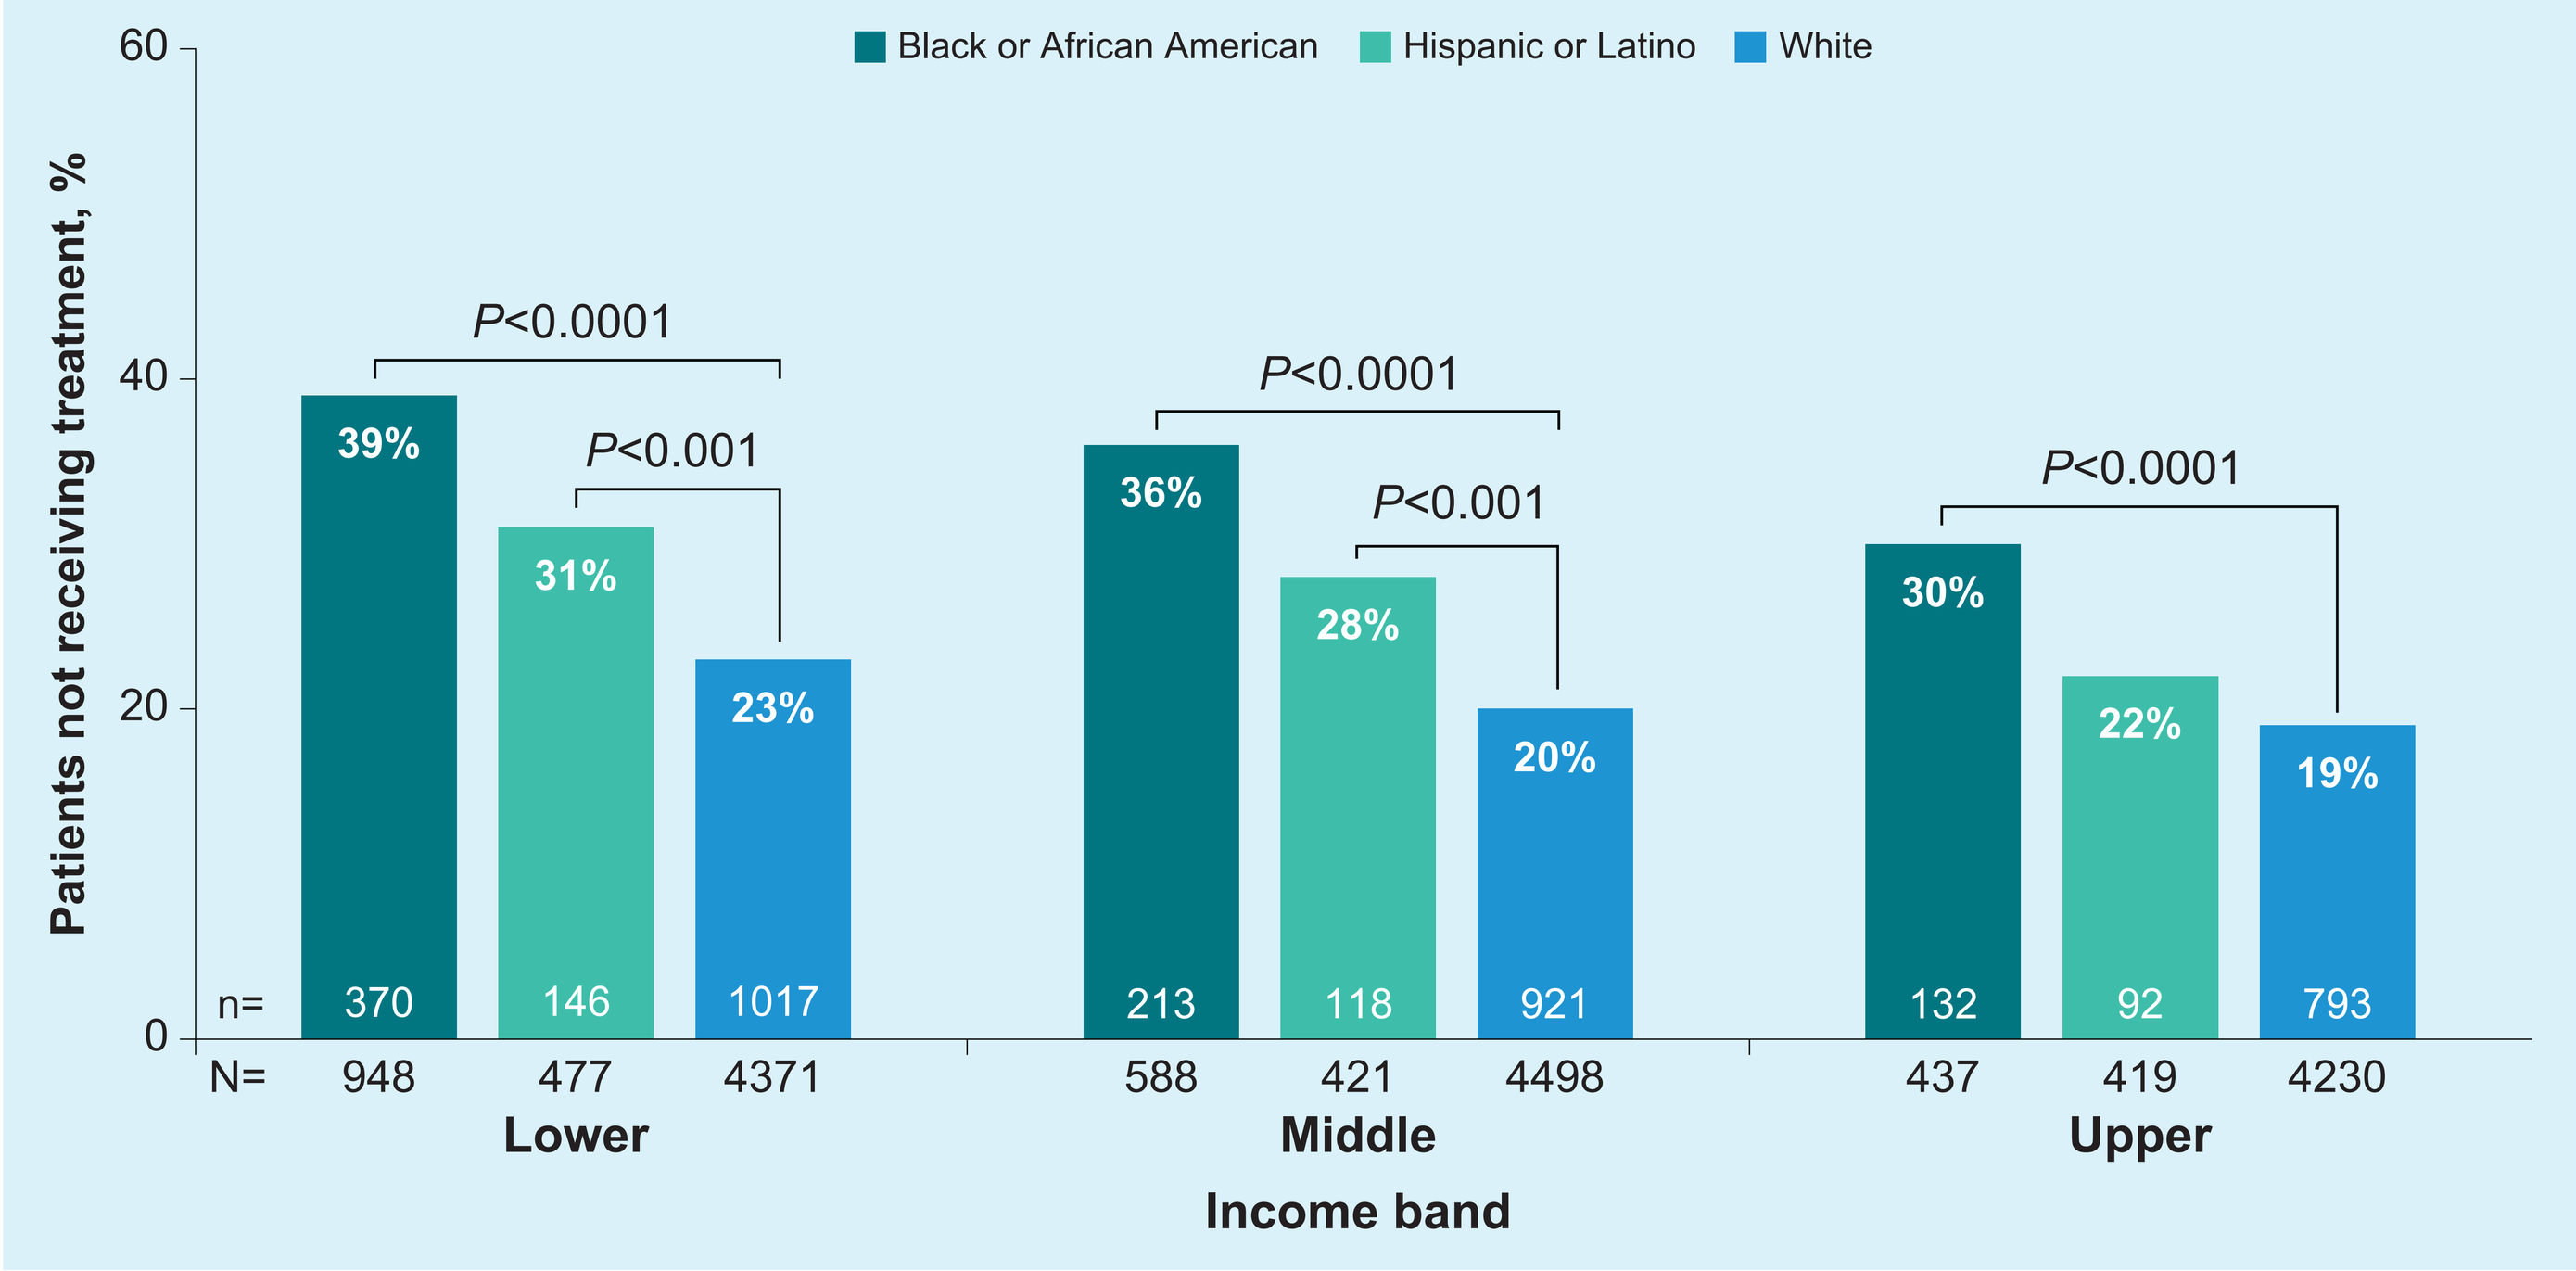
Black or African American Hispanic or Latino White
Patients not receiving treatment, %
60
40
20
0
P<0.0001
P<0.001
39% 31% 23%
n= 370 146 1017
N= 948 477 4371
Lower
P<0.0001
P<0.001
36% 28% 20%
213 118 921
588 421 4498
Middle
P<0.0001
30% 22% 19%
132 92 793
437 419 4230
Upper
Income band

EDS, excessive daytime sleepiness.

STUDY LIMITATIONS

- Reliance on ICD-10 codes without clinical validation may have resulted in misclassification of NT1, NT2, and IH diagnoses; however, select patients with stable diagnoses over time may have mitigated some misclassification concerns
- Additional clinical and demographic factors that were not included in this analysis (eg, disease severity, symptom burden, comorbidities, age) may influence treatment selection
- As a claims database analysis, demographic characteristics of this study population may not be representative of the overall population of patients with narcolepsy
- Paid claims do not confirm medication initiation, adherence, or persistence
- The limited timeframe of this analysis (2023) did not allow longitudinal assessment of treatment patterns
- Requirement for continuous claim availability may have excluded patients who were more vulnerable or intermittently insured and may have led to underestimated differences based on race and ethnicity, or income level
- The healthcare-seeking behaviors of the patient population in this study could have affected the differences observed across race and ethnicity, or income level

CONCLUSIONS

- These data demonstrate that across income levels, Black or African American patients were numerically less likely to receive a strongly recommended narcolepsy treatment and numerically less likely to receive any treatment for EDS compared with patients of other races or ethnicities
- Despite being approved by the FDA[5-7] for treatment of cataplexy or EDS for >2 decades and being strongly recommended by the AASM for the treatment of EDS in narcolepsy since 2007,[14] oxybate use was consistently low (≤11%) across race and ethnicity groups and income levels, highlighting a potential opportunity to improve patient care

ACKNOWLEDGMENTS

Medical writing support was provided by Marina Wylie, PhD, from Citrus Health Group, Inc. (Chicago, IL) and was funded by Avadel Pharmaceuticals (Chesterfield, MO).

FUNDING

This study was funded by Avadel Pharmaceuticals (Chesterfield, MO). *Avadel Pharmaceuticals Limited (formerly Avadel Pharmaceuticals plc) is an affiliate of Alkermes plc.

DISCLOSURES

RB has served as a consultant, speaker, and/or on advisory boards for Avadel Pharmaceuticals, Harmony Biosciences, and Jazz Pharmaceuticals. **ABP** served as a local principal investigator in the PMLUM-2402 open-label, observational study of once-nightly sodium oxybate (LUMRYZ®) for the treatment of narcolepsy. The study was sponsored by Avadel Pharmaceuticals. **HM** has participated on a Delphi panel and has served as a consultant and on advisory boards for Avadel Pharmaceuticals. **OB** has served as a speaker for Harmony Biosciences, Idorsia, and Lilly; is a principal investigator with Centessa Pharmaceuticals and Neteera; and is Chief Executive Officer of the Sleep Doctor PLLC. **JG** was an employee of Avadel Pharmaceuticals and is a consultant to Alkermes, Inc. **JH** and **BA** are employees of Alkermes, Inc.

REFERENCES

1. American Academy of Sleep Medicine. *International Classification of Sleep Disorders*. 3rd ed. American Academy of Sleep Medicine; 2014.
2. Romero RG, et al. *Sleep*. 2024;47(Supplement_1):A287-A287.
3. Flores NM, et al. *J Clin Sleep Med*. 2016;12(3):401-407.
4. Thorpy MJ, Dauvilliers Y. *Sleep Med*. 2015;16(1):9-18.
5. Xyrem® (sodium oxybate oral solution, CIII). Full Prescribing Information. Jazz Pharmaceuticals, Inc; 2025.
6. Xywav® (calcium, magnesium, potassium, and sodium oxybate). Full Prescribing Information. Jazz Pharmaceuticals, Inc; 2025.
7. LUMRYZ® (sodium oxybate for extended-release oral suspension, CIII). Full Prescribing Information. Avadel Pharmaceuticals; 2025.
8. Wakix (pitolisant). Full Prescribing Information. Harmony Biosciences, LLC; 2025.
9. Provigil (modafinil). Full Prescribing Information. Teva Pharmaceuticals; 2022.
10. DAYVIGO® (lemborexant). Full Prescribing Information. Eisai Inc.; 2025.
11. QUVIVIQ (daridorexant). Full Prescribing Information. Idorsia Pharmaceuticals US Inc.; 2022.
12. Maski K, et al. *J Clin Sleep Med*. 2021;17(9):1881-1893.
13. Maski K, et al. *J Clin Sleep Med*. 2021;17(9):1895-1945.
14. Morgenthaler TI, et al. *Sleep*. 2007;30(12):1705-1711.

Copies of this poster can be obtained through this QR (Quick Response) code

PRESENTED AT SLEEP 2026 • JUNE 14-17, 2026 • BALTIMORE, MD, USA

THESE MATERIALS ARE FOR PERSONAL USE ONLY AND MAY NOT BE REPRODUCED WITHOUT PERMISSION OF ALKERMES. FOR PERMISSION, CONTACT USMEDINFO@ALKERMES.COM.