Opioid Antagonists: Clinical Utility, Pharmacology, and Safety and Tolerability

Roger S. McIntyre,[1] Marni White,[2] Mark S. Todtenkopf,[2] Sarah Akerman,[2] Joshua Burgett[3]
[1]Brain and Cognition Discovery Foundation (BCDF), University of Toronto, Toronto, ON, Canada; [2]Alkermes, Inc., Waltham, MA, USA; [3]Community Bridges, Mesa, AZ, USA

INTRODUCTION

- Opioid antagonists block opioid receptors and are used therapeutically for several different indications
- This poster summarizes relevant information on the receptor pharmacology, clinical utility, safety, and tolerability of US Food and Drug Administration–approved opioid antagonists

Figure 1. Drug Receptor Interactions

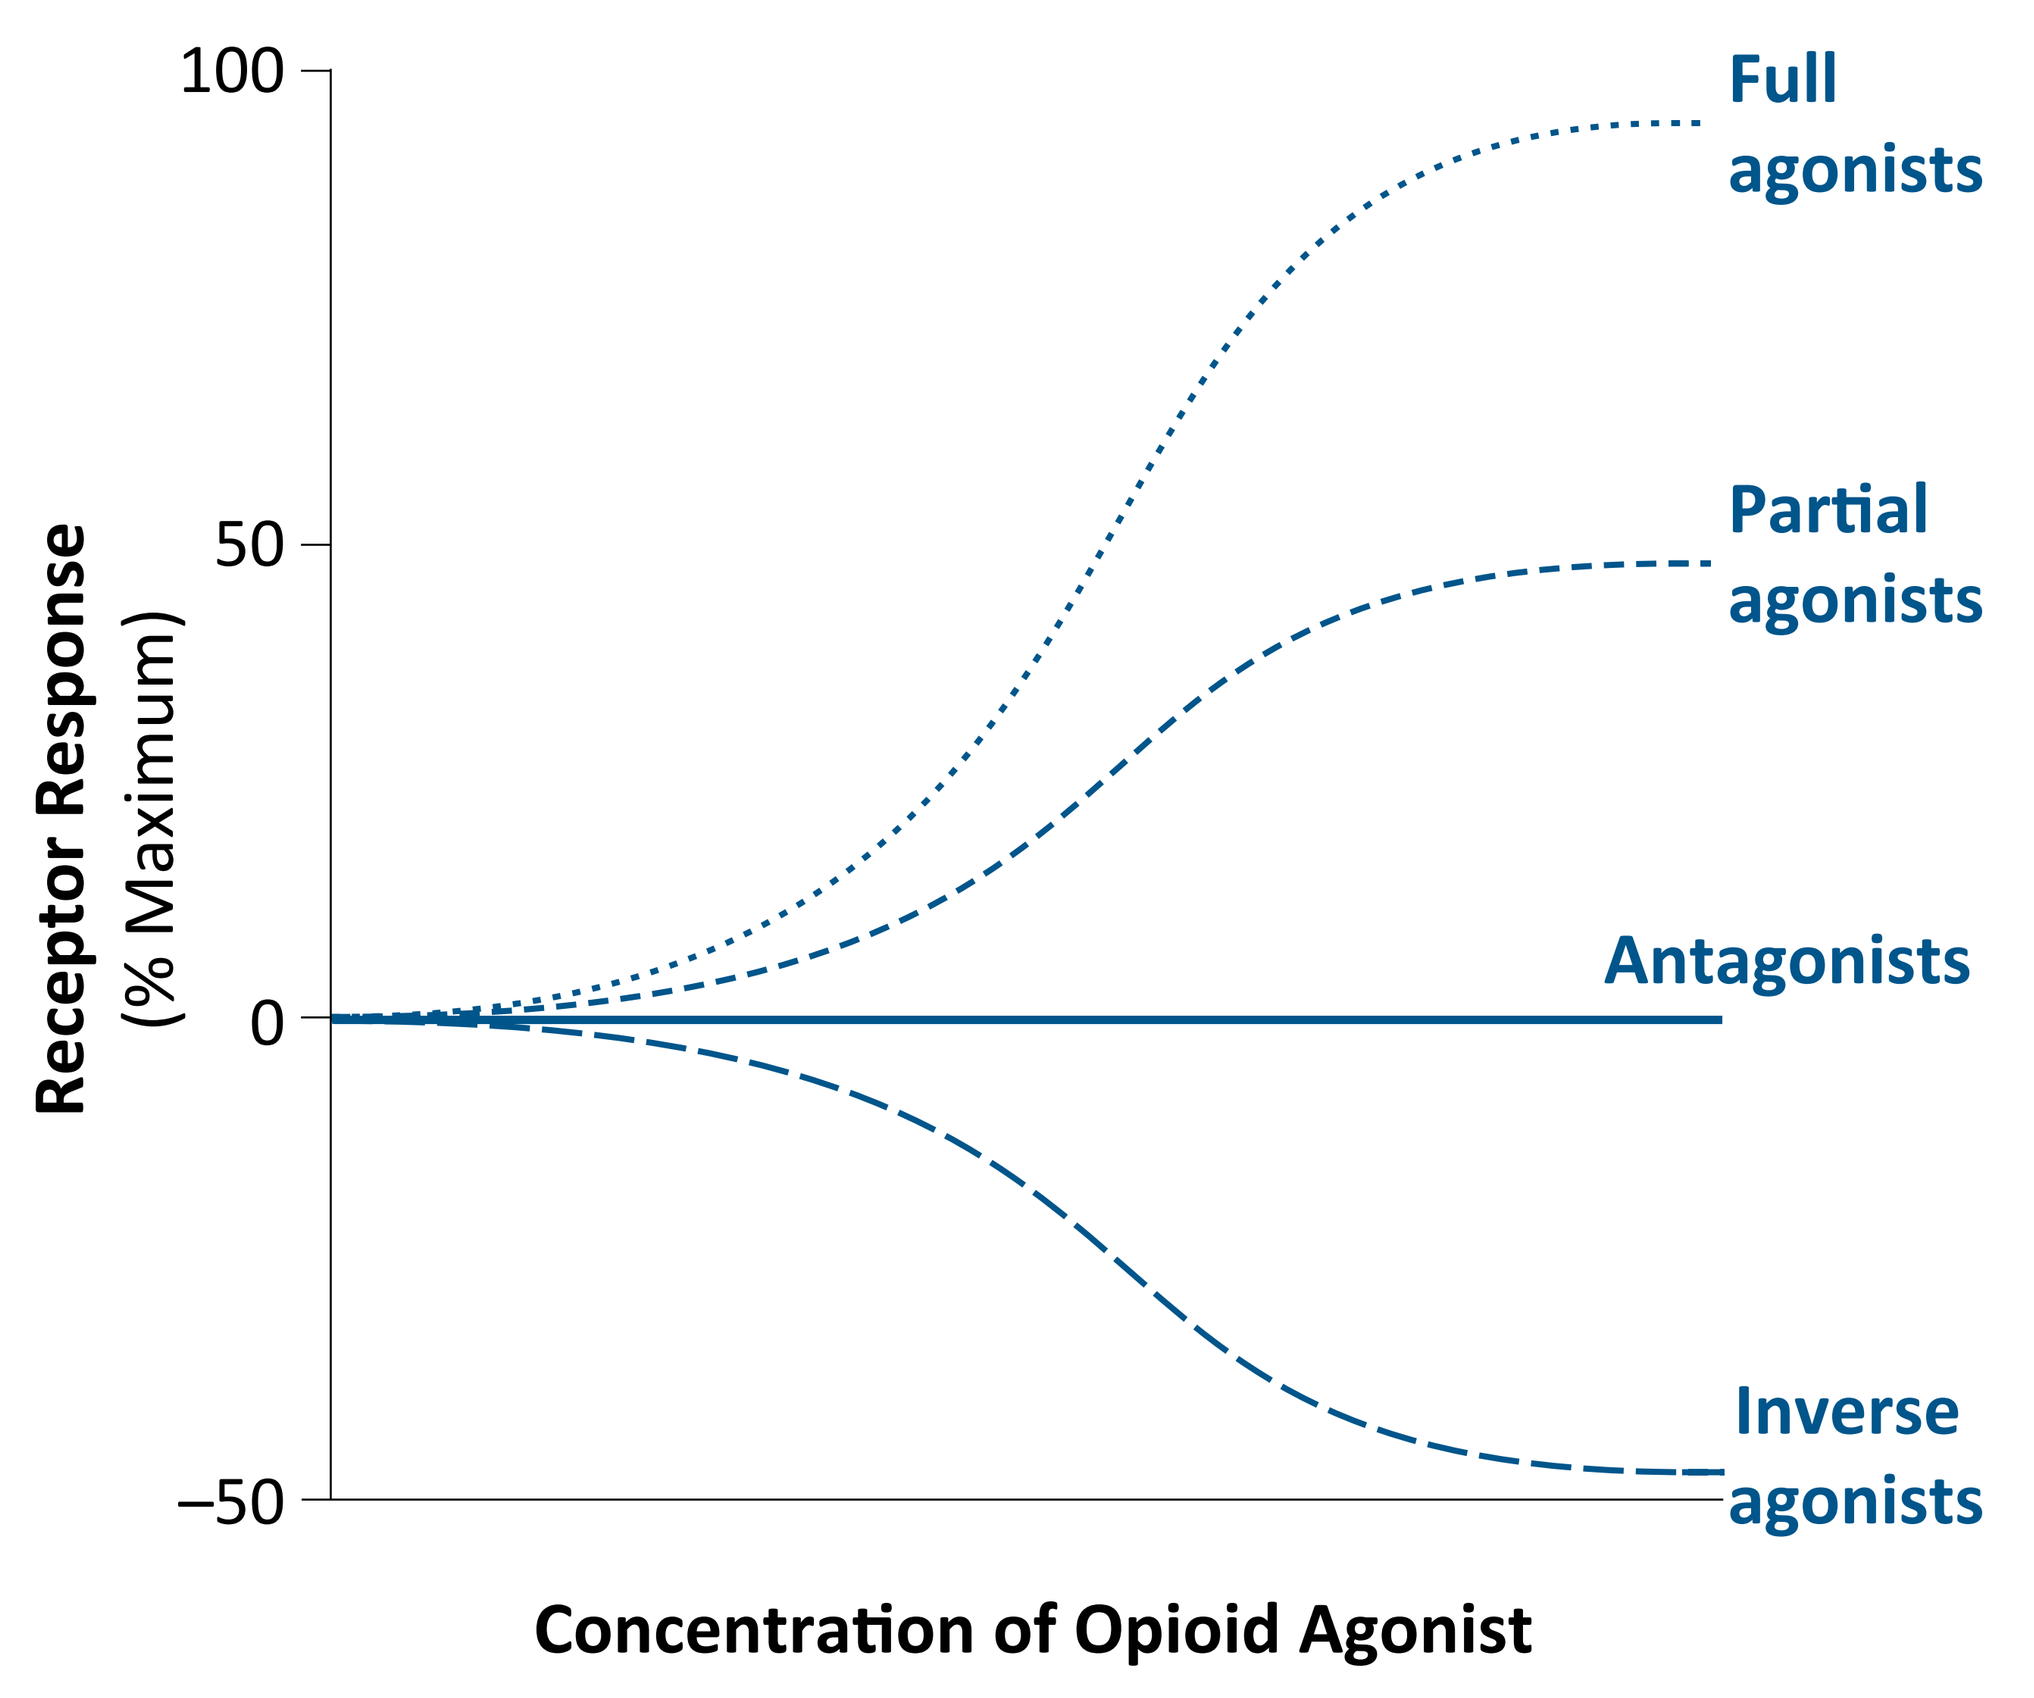
100
50
0
−50
Receptor Response
(% Maximum)
Full agonists
Partial agonists
Antagonists
Inverse agonists
Concentration of Opioid Agonist

Adapted from https://commons.wikimedia.org/wiki/File:Inverse_agonist_3.svg, Creative Commons Attribution-Share Alike 4.0 International license.

- In general, drugs can be categorized by the physiologic response elicited once bound to receptors[1] (**Figure 1**)
- **Full agonists** activate receptors to their maximum extent, thus initiating a physiologic response
- **Partial agonists** activate receptors to a lesser extent than full agonists, evoking an attenuated physiologic response compared with full agonists
- **Antagonists** prevent or reduce the physiologic response that results from full or partial agonists by blocking receptor binding
- **Inverse agonists** reduce the natural background activation of receptors, resulting in a physiologic response that is opposite to that of full or partial agonists

Figure 2. Distribution of Opioid Receptors

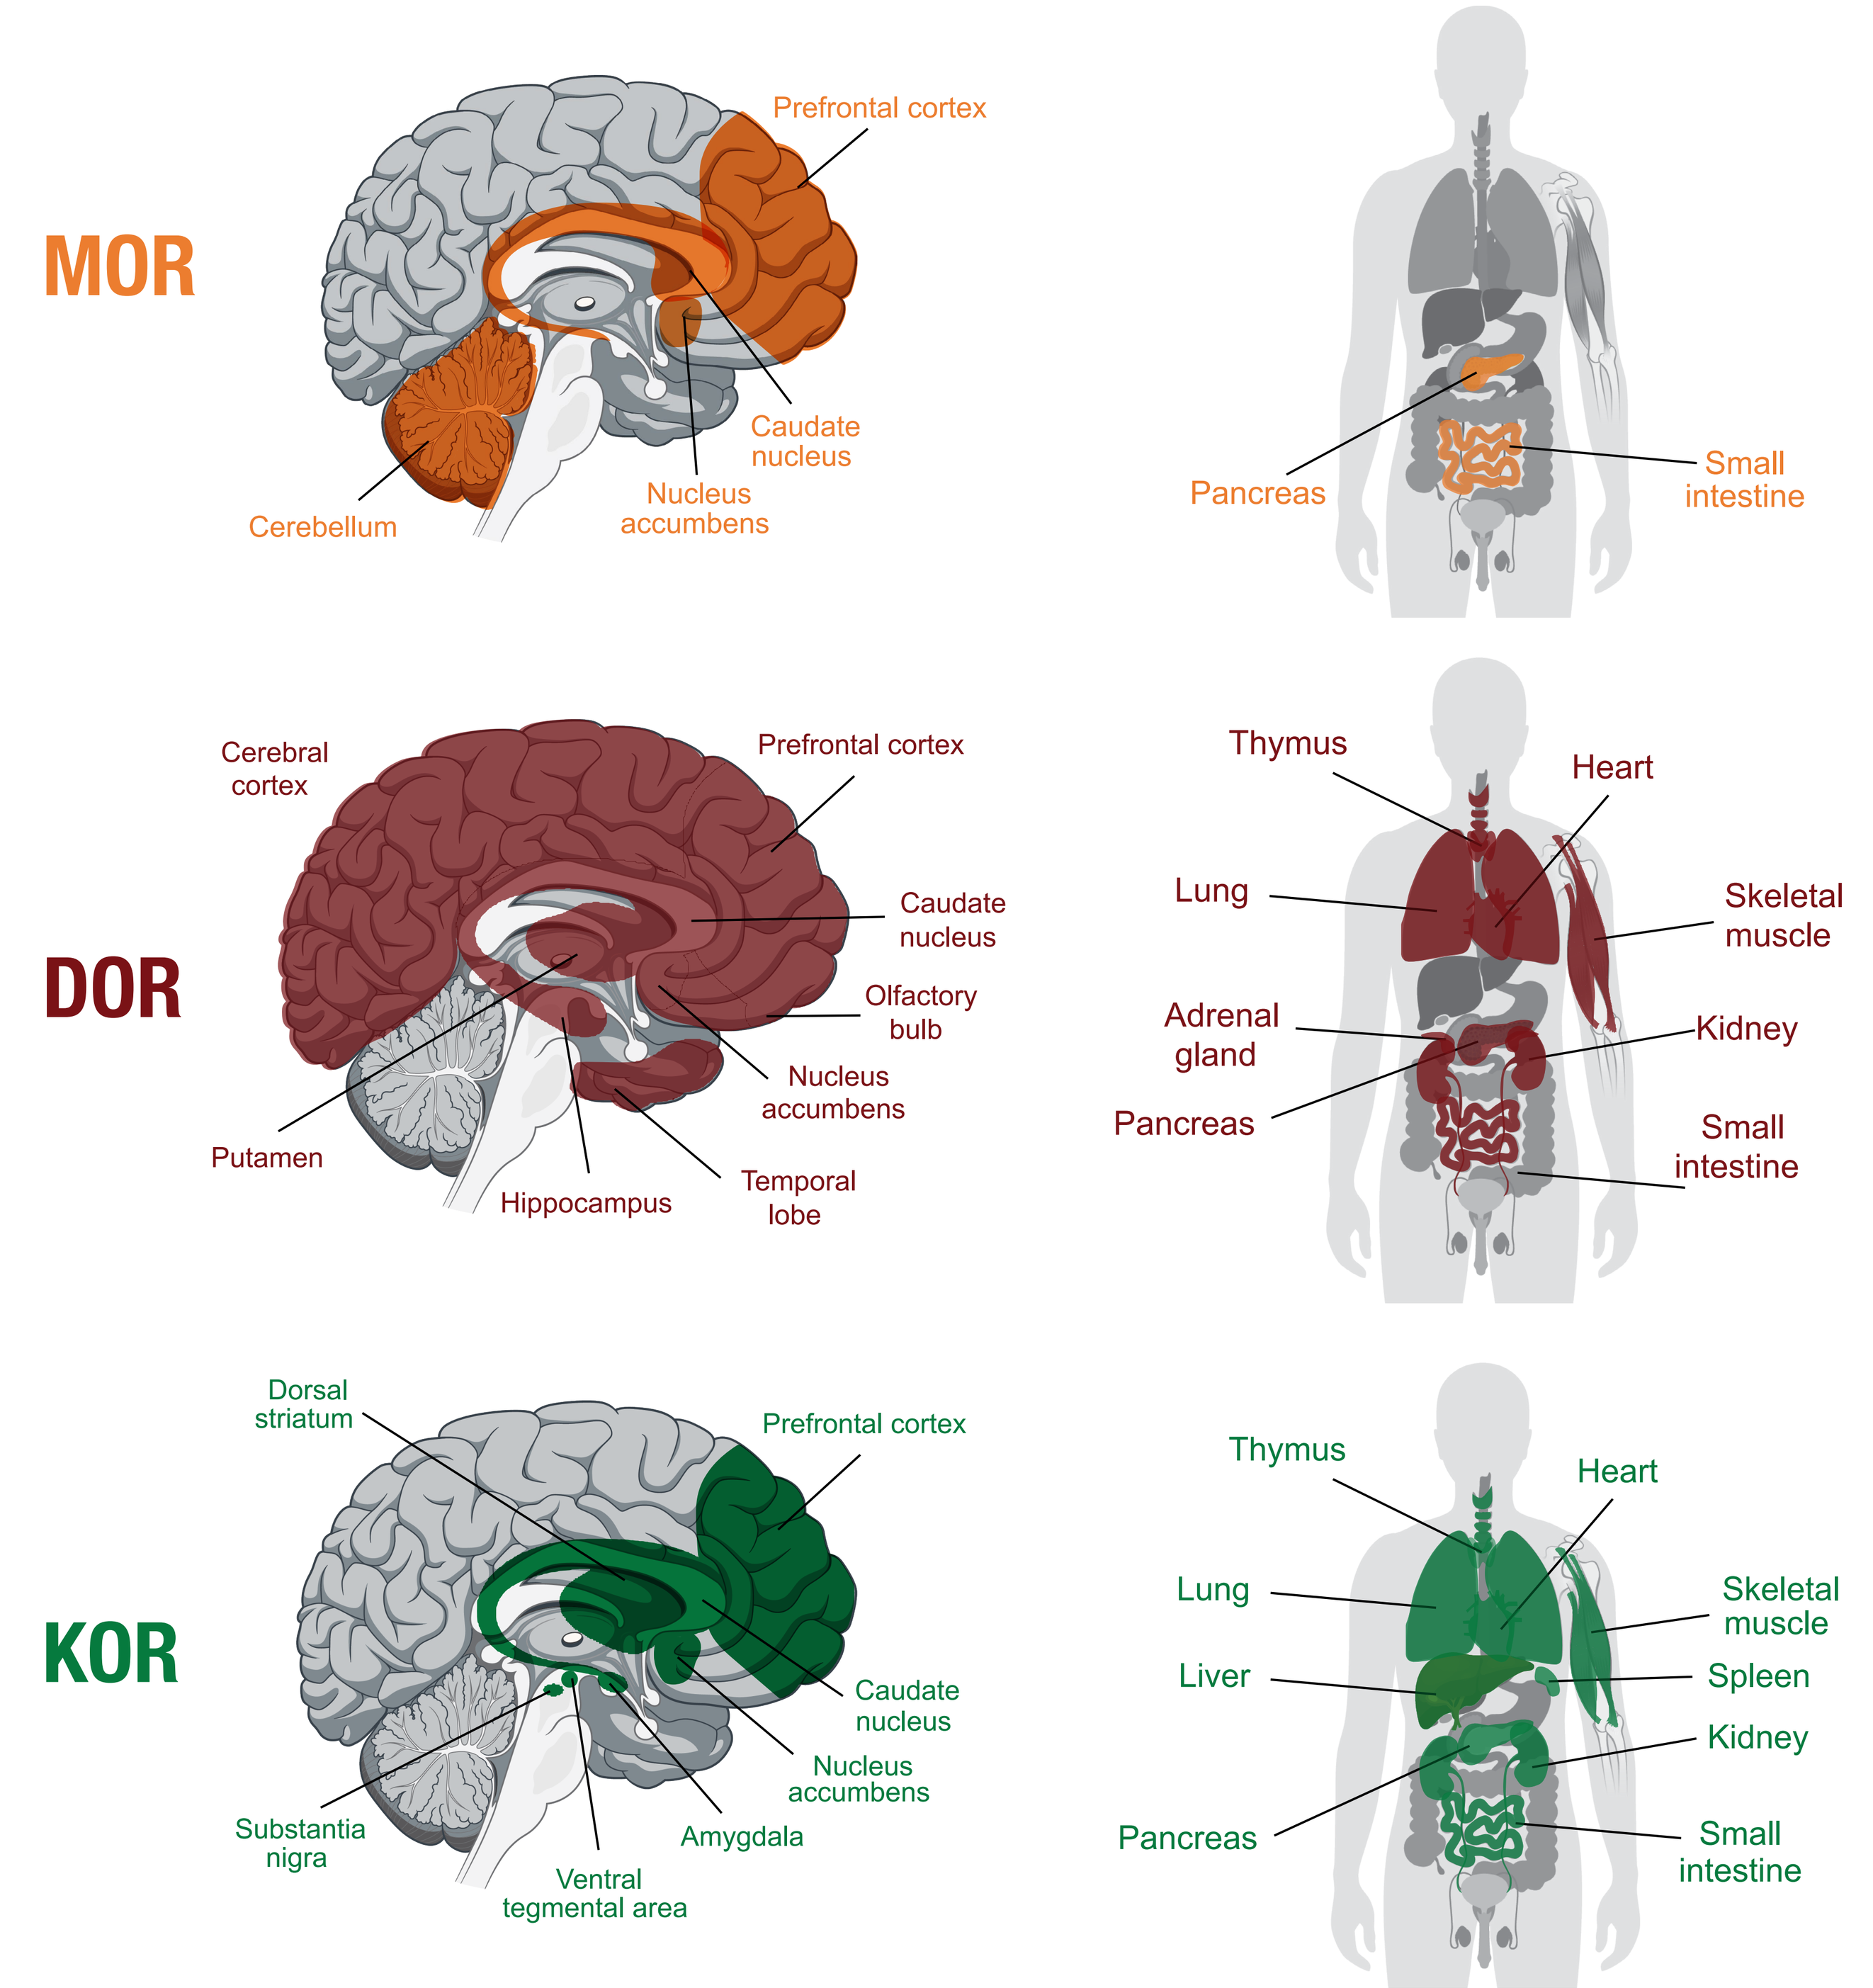
MOR
Prefrontal cortex
Caudate nucleus
Nucleus accumbens
Cerebellum
Pancreas
Small intestine
DOR
Cerebral cortex
Prefrontal cortex
Caudate nucleus
Olfactory bulb
Nucleus accumbens
Temporal lobe
Hippocampus
Putamen
Thymus
Heart
Lung
Skeletal muscle
Adrenal gland
Kidney
Pancreas
Small intestine
KOR
Dorsal striatum
Prefrontal cortex
Caudate nucleus
Nucleus accumbens
Amygdala
Ventral tegmental area
Substantia nigra
Thymus
Heart
Lung
Skeletal muscle
Liver
Spleen
Kidney
Pancreas
Small intestine

(Reprinted with permission from reference 2.)
DOR, delta opioid receptor; KOR, kappa opioid receptor; MOR, mu opioid receptor.

- The opioid system is composed of opioid receptors that bind both endogenous ligands and exogenous drugs (**Figure 2**)[2]
- Opioid receptors are located both centrally and peripherally; as a result, the opioid system impacts the regulation of several bodily functions, including reward, mood, body weight, metabolism, and pain[2]
- These effects are mediated through 3 main types of opioid receptors: **MORs, DORs,** and **KORs**[2]; opioid agonists, such as morphine, activate these receptors and elicit physiologic responses that include euphoria, analgesia, respiratory depression, and decreased gastrointestinal (GI) motility[3,4]
- **Opioid antagonists** generally function as **neutral antagonists**, producing limited physiologic effects on their own, but block the physiologic effects when an opioid agonist is present
 - They block 1 or more of these receptor types and therefore have several clinical uses[2]

Table 1. Centrally Acting Opioid Antagonists

	Clinical Uses	Receptor Profile	Formulation(s)	Oral Bioavailability, %	Parenteral/ Other Bioavailability, %	Half-life, hours
Alcohol use disorder/opioid use disorder						
Naltrexone[5-7]	Treatment of alcohol use disorder and opioid use disorder	MOR and KOR antagonist, lower affinity for DOR	Oral Injection	5–40 NA	NA NA	4 120-240
Naloxone + buprenorphine[1,8-11]	Treatment of opioid use disorder with naloxone used as an abuse deterrent	Naloxone: MOR antagonist, affinity for DORs and KORs	Sublingual film and tablets	Naloxone: 3	NA	Naloxone: 2–12
Opioid agonist overdose reversal						
Naloxone[4,9,12,13]	Reversing physiologic effects of opioid overdose, such as respiratory depression	MOR antagonist, affinity for DORs and KORs	Intranasal Injection	<3 NA	44-54 98	1–2
Nalmefene[14-16]	Reversing physiologic effects of opioid overdose, such as respiratory depression	MOR and DOR antagonist, partial KOR agonist	Injection	41	IM: 102 SC: 100	11
Body weight						
Naltrexone + bupropion[2,17]	Chronic weight management in adults who are overweight with weight-related comorbidity, or in those who are obese	Naltrexone: MOR and KOR antagonist, lower affinity for DORs	Oral	5–40	Undetermined	Naltrexone: 5
Samidorphan + olanzapine[18]	Provides the antipsychotic efficacy of olanzapine in patients with schizophrenia or bipolar I disorder while mitigating olanzapine-associated weight gain	Samidorphan: MOR antagonist, partial DOR and KOR agonist	Oral	69	Undetermined	Samidorphan: 7–11

DOR, delta opioid receptor; KOR, kappa opioid receptor; MOR, mu opioid receptor; NA, not applicable.

- Centrally acting opioid antagonists preferentially bind to opioid receptors in the brain and spinal cord (**Table 1**)[19,20]
- Each of these medications is a MOR antagonist, with different binding profiles at DOR and KOR
- AEs associated with centrally acting opioid antagonists are generally mild and may include nausea and vomiting for certain medications

Table 2. Peripherally Acting Opioid Antagonists

	Clinical Use	Receptor Profile	Formulation(s)	Oral Bioavailability, %	Parenteral/ Other Bioavailability, %	Half-life, hours
Opioid-induced constipation						
Naloxegol[19,20]	Opioid-induced constipation	MOR and DOR antagonist, partial KOR agonist	Oral	Undetermined	NA	6–11
Methylnaltrexone[21,22]	Opioid-induced constipation	MOR and KOR antagonist, low affinity with DORs	Oral Injection	Undetermined NA	NA 82	8–15
Naldemedine[23,24]	Opioid-induced constipation	MOR, DOR, and KOR antagonist	Oral	20–56	NA	11
GI recovery after bowel surgery						
Alvimopan[25,26]	Accelerate recovery after certain GI surgeries	MOR antagonist, low affinity for DORs and KORs	Oral	6	NA	10–17

DOR, delta opioid receptor; GI, gastrointestinal; KOR, kappa opioid receptor; MOR, mu opioid receptor; NA, not applicable.

- Peripherally acting opioid antagonists preferentially bind to opioid receptors located throughout the body (**Table 2**)
- Each of these medications is a MOR antagonist with a different binding profile at DOR and KOR
- OIC is a common and bothersome AE associated with opioid agonist use[27]
 - Stimulation of opioid receptors in the gut decreases GI motility and may lead to bowel dysfunction
 - Peripherally acting opioid antagonists preferentially block opioid receptors located in the gut, thus alleviating symptoms of OIC
 - Because of their limited ability to reach the brain, they can treat OIC without compromising the analgesia that opioid agonists provide
- AEs associated with peripherally acting opioid antagonists are mild and may include abdominal pain, nausea, diarrhea, and dyspepsia

Figure 3. Safety Considerations When Using Opioid Antagonists

Safety Considerations

- Generally safe and well tolerated
- No abuse potential
- Not associated with physiologic tolerance or dependence
- May precipitate withdrawal in patients taking an opioid agonist
- Risk of overdose if attempts are made to overcome receptor blockade of centrally acting opioid antagonist
- Peripherally acting opioid antagonists should not be used in patients with or at risk for gastrointestinal obstruction

- Opioid antagonists are safe and well tolerated when used according to their approved indication (**Figure 3**)
- They are not associated with physiologic tolerance or dependence, nor are they associated with abuse
- As a class, opioid antagonist medications may precipitate opioid agonist withdrawal
 - Symptoms of opioid agonist withdrawal may include abdominal cramps, restlessness, and nausea
 - Some opioid antagonists (eg, naltrexone and samidorphan) should be used only after a period of abstinence from opioid agonists
- Centrally acting opioid antagonists confer a risk of opioid agonist overdose if attempts are made to overcome receptor blockade

CONCLUSIONS

- Opioid antagonists are available for clinical use for several indications
- Their differential pharmacologic profiles at the 3 main types of opioid receptors, and differential abilities to cross the blood–brain barrier, lead to unique clinical uses among these agents
- As a class, opioid antagonists are generally safe and well tolerated with no abuse potential, but they may precipitate opioid agonist withdrawal
- Their multiple clinical indications make opioid antagonists important tools in the treatment armamentarium
- Knowledge of their pharmacologic properties, clinical uses, and safety profiles enable their safe and effective use

References, acknowledgments, disclosure information, and copies of this poster can be obtained through this QR (Quick Response) code. These materials are for personal use only and may not be reproduced without permission of Alkermes. For permission, contact USMedInfo@Alkermes.com

Supported by funding from Alkermes, Inc.

Opioid Antagonists: Clinical Utility, Pharmacology, and Safety and Tolerability

Roger S. McIntyre,[1] Marni White,[2] Mark S. Todtenkopf,[2] Sarah Akerman,[2] Joshua Burgett
[1]Brain and Cognition Discovery Foundation (BCDF), University of Toronto, Toronto, ON, Canada; [2]Alkermes, Inc., Waltham, MA, USA

REFERENCES

1. Katzung BG, et al. Basic and Clinical Pharmacology. New York, NY: McGraw Hill Lange; 2012.
2. Mcintyre RS, et al. *CNS Spectr.* 2022;28(3):288-99. DOI: 10.1017/s1092852922000116.
3. Le Merrer J, et al. *Physiol Rev.* 2009;89(4):1379-412. DOI: 10.1152/physrev.00005.2009.
4. Choi YS, Billings JA. *J Pain Symptom Manage.* 2002;24(1):71-90. DOI: 10.1016/s0885-3924(02)00424-4.
5. Revia [package insert]. Pomona, NY: Duramed Pharmaceuticals, Inc.; 2013.
6. Center for Substance Abuse Treatment. Incorporating Alcohol Pharmacotherapies Into Medical Practice. Rockville, MD: Substance Abuse and Mental Health Services Administration; 2009.
7. Vivitrol [package insert]. Waltham, MA: Alkermes, Inc; 2021.
8. Suboxone [package insert]. North Chesterfield, VA: Indivior Inc.; 2023.
9. Miller RD, et al. Miller's Anesthesia. Philadelphia, PA: Churchill Livingstone; 2009.
10. Coe MA, et al. *J Addict Med.* 2019;13(2):93-103. DOI: 10.1097/adm.0000000000000457.
11. Kuhlman JJ, Jr, et al. *J Anal Toxicol.* 1996;20(6):369-78. DOI: 10.1093/jat/20.6.369.
12. Narcan [package insert]. Radnor, PA: Adapt Pharma; 2015.
13. Zimhi [package insert]. San Diego, CA: Adamis Pharmaceuticals; 2021.
14. Revex [package insert]. Deerfield, IL: Baxter Healthcare; 2006.
15. Selincro [summary of product characteristics]. Valby, Denmark: H. Lundbeck; 2022.
16. Nalmefene hydrochloride [package insert]. Stamford, CT: Purdue Pharma; 2021.
17. Contrave [package insert]. Morristown, NJ: Currax Pharmaceuticals LLC; 2020.
18. Lybalvi [package insert]. Waltham, MA: Alkermes, Inc.; 2021.
19. Movantik [package insert]. Wilmington, DE: AstraZeneca Pharmaceuticals; 2014.
20. Moventig assessment report. 2014. Available at: https://www.ema.europa.eu/en/documents/assessment-report/moventig-epar-public-assessment-report_en.pdf.
21. Relistor [package insert]. Raleigh, NC: Salix Pharmaceuticals, Inc.; 2020.
22. Relistor [summary of product characteristics]. Dublin, Ireland: Bausch Health Ireland; 2022.
23. Symproic [package insert]. Raleigh, NC: BioDelivery Sciences International, Inc.; 2020.
24. Rizmoic [summary of product characteristics]. Amsterdam, Netherlands: Shionogi; 2023.
25. Entereg [package insert]. Rahway, NJ: Merck Sharp & Dohme; 2022.
26. Neary P, Delaney CP. *Expert Opin Investig Drugs.* 2005;14(4):479-88. DOI: 10.1517/13543784.14.4.479.
27. Pergolizzi JV, Jr, et al. *Drug Des Devel Ther.* 2020;14:1009-25. DOI: 10.2147/dddt.s221278.

AUTHOR DISCLOSURES

RSM has received research grant support from CIHR/GACD/National Natural Science Foundation of China (NSFC) and the Milken Institute and speaker/consultation fees from AbbVie, Alkermes, Atai Life Sciences, Axsome, Bausch Health, Biogen, Boehringer Ingelheim, Eisai, Intra-Cellular, Janssen, Kris, Lundbeck, Mitsubishi Tanabe, Neumora Therapeutics, Neurocrine, NewBridge Pharmaceuticals, Novo Nordisk, Otsuka, Pfizer, Purdue, Sage, Sanofi, Sunovion, Takeda, and Viatris and is a CEO of Braxia Scientific Corp.

MW, MST, SA, and **CC** are or were employees of Alkermes, Inc., and may own stock/options in the company.

JB has received funding from Alkermes, Intra-Cellular, Janssen, Lundbeck, Otsuka, and Pfizer and payment for medical expert services from the US Attorney's Office, Improve Health, and Maximus Federal.

ACKNOWLEDGMENTS

This study was sponsored by Alkermes, Inc. (Waltham, MA, USA). Medical writing and editorial support were provided by Peloton Advantage, LLC (Parsippany, NJ, USA), an OPEN Health company, and funded by Alkermes, Inc.

Supported by funding from Alkermes, Inc.