Association Between Depression and Disease Burden in Narcolepsy and Idiopathic Hypersomnia: Patient-Centered Results From the ASPIRE Study

Poster No. 151

Craig Chepke,[1,2] Michael J. Doane,[3] Lindsay Jesteadt,[4,5] Kristen McCausland,[6] Jane Lazar Tucker,[6] Gretchen Murphy,[3] Anne Marie Morse,[7] David T. Plante[8]

[1]Excel Psychiatric Associates, P.A., Huntersville, NC, USA; [2]Atrium Health, Charlotte, NC, USA; [3]Alkermes, Inc., Waltham, MA, USA; [4]Sleep Consortium, Inc., West Palm Beach, FL, USA; [5]Hypersomnia Foundation, Atlanta, GA, USA; [6]IQVIA Quality Metric, Johnston, RI, USA; [7]Geisinger Commonwealth School of Medicine, Geisinger Medical Center, Janet Weis Children's Hospital, Danville, PA, USA; [8]University of Wisconsin-Madison, School of Medicine and Public Health, Madison, WI, USA

Introduction

- As one of the most common psychiatric comorbidities among individuals with narcolepsy and idiopathic hypersomnia (IH), depression may contribute to overall disease burden[1–3]
- Depressive symptoms may interact with narcolepsy and IH to influence patient experience and disease burden
 - These rare sleep disorders are associated with unpredictable and disabling symptoms that may affect psychosocial functioning, independence, and engagement in daily activities, increasing the risk for depression[1–6]
 - Depressive symptoms may further influence how individuals experience and perceive the symptoms and effects of their sleep disorder, potentially amplifying clinical, functional, and psychosocial burden[1–3,7]
- Characterizing the burden associated with depression may help inform more holistic, patient-centered assessment and management of narcolepsy and IH[1,2,4]

Objective

- To evaluate associations between depression status and psychosocial outcomes, symptom severity, and treatment priorities across people living with narcolepsy and IH

Methods

- Data were analyzed from ASPIRE, a noninterventional, longitudinal, online survey of US adults diagnosed with narcolepsy type 1 (NT1), narcolepsy type 2 (NT2), or IH, as summarized in **Table 1**

Table 1. Study population

NT1 (n = 116)	NT2 (n = 127)	IH (n = 123)
Participants were ≥ 18 years of age, resided in the US, and reported a clinician's diagnosis of NT1, NT2, or IH		

IH, idiopathic hypersomnia; NT1, narcolepsy type 1; NT2, narcolepsy type 2.

- Participants were recruited through Rare Patient Voice, the Hypersomnia Foundation, and Sleep Consortium from May 9 to July 2, 2025
- Probable depression was defined as a Mental Component Summary (MCS) score of ≤ 42 on the SF-12v2® Health Survey, which has been shown to have a sensitivity of 73.7% and specificity of 80.6% in identifying individuals with clinical depression[8,9]
- Symptom severity and disease burden were assessed using patient-reported measures of excessive daytime sleepiness (EDS), fatigue, cognitive impairment, sleep inertia, daily functioning, work/activity impairment, and health-related quality of life (HRQoL), including the following:
 - Epworth Sleepiness Scale
 - Patient-Reported Outcomes Measurement Information System – Fatigue Short Form 6a
 - British Columbia Cognitive Complaints Inventory – Expanded version
 - Sleep Inertia Visual Analogue Scale
 - Functional Outcomes of Sleep Questionnaire-Short Form
 - Work Productivity and Activity Impairment Questionnaire: Specific Health Problem
 - EuroQol 5-Dimension 5-Level index score
- Validated patient-reported outcome instruments were supplemented with de novo survey items designed to quantify specific psychosocial impacts, life restrictions, and the perceived importance of treatment benefits
- Outcomes were compared by depression status (i.e., between participants with and without probable depression) using descriptive statistics, independent-samples *t* tests, chi-square tests, and logistic regression; odds ratios (ORs) were calculated for relevant outcomes
- Analyses used a two-sided significance threshold of $P < 0.05$, with no adjustment for multiple comparisons in these exploratory analyses

Results

- Among 366 participants, 59% met the MCS criterion for probable depression; rates were similar across diagnostic groups (NT1, 59%; NT2, 61%; IH, 55%) (**Figure 1A**)
- Across demographic subgroups, depression rates were generally consistent with the overall rate, with the numerically highest rates observed among participants reporting disability and those with a high school education or less (**Figure 1B**)

Figure 1. (A) Prevalence of depression by diagnosis and (B) by demographic subgroup

A

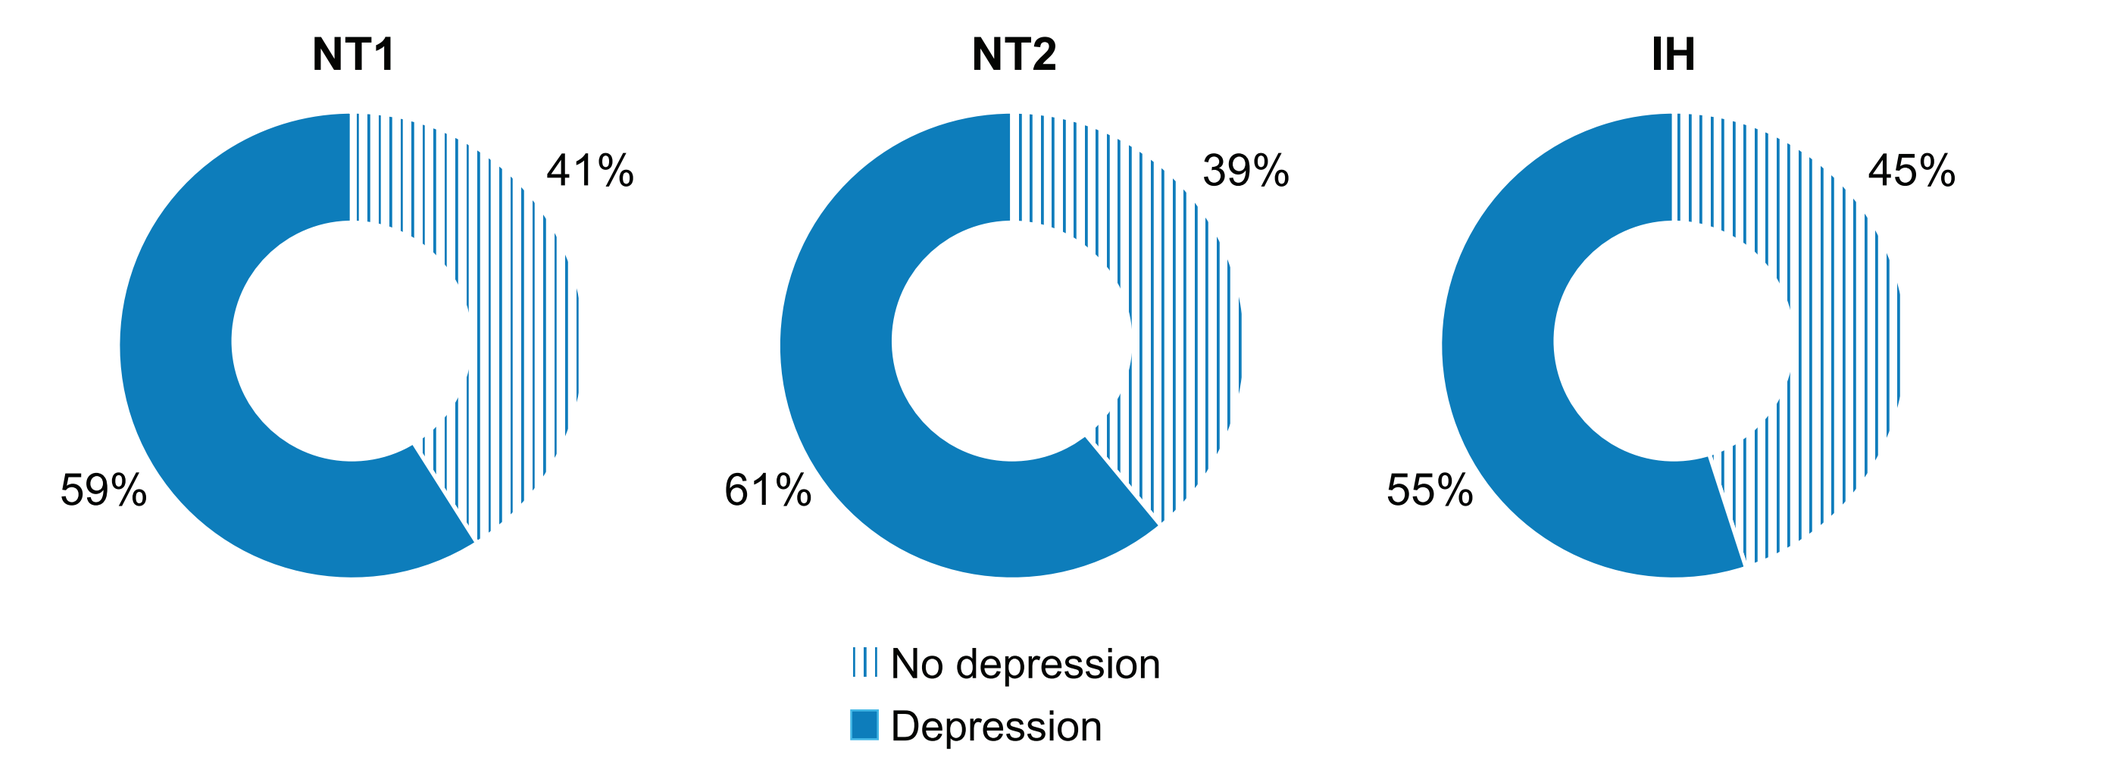
NT1
41%
59%
NT2
39%
61%
IH
45%
55%
No depression
Depression

B

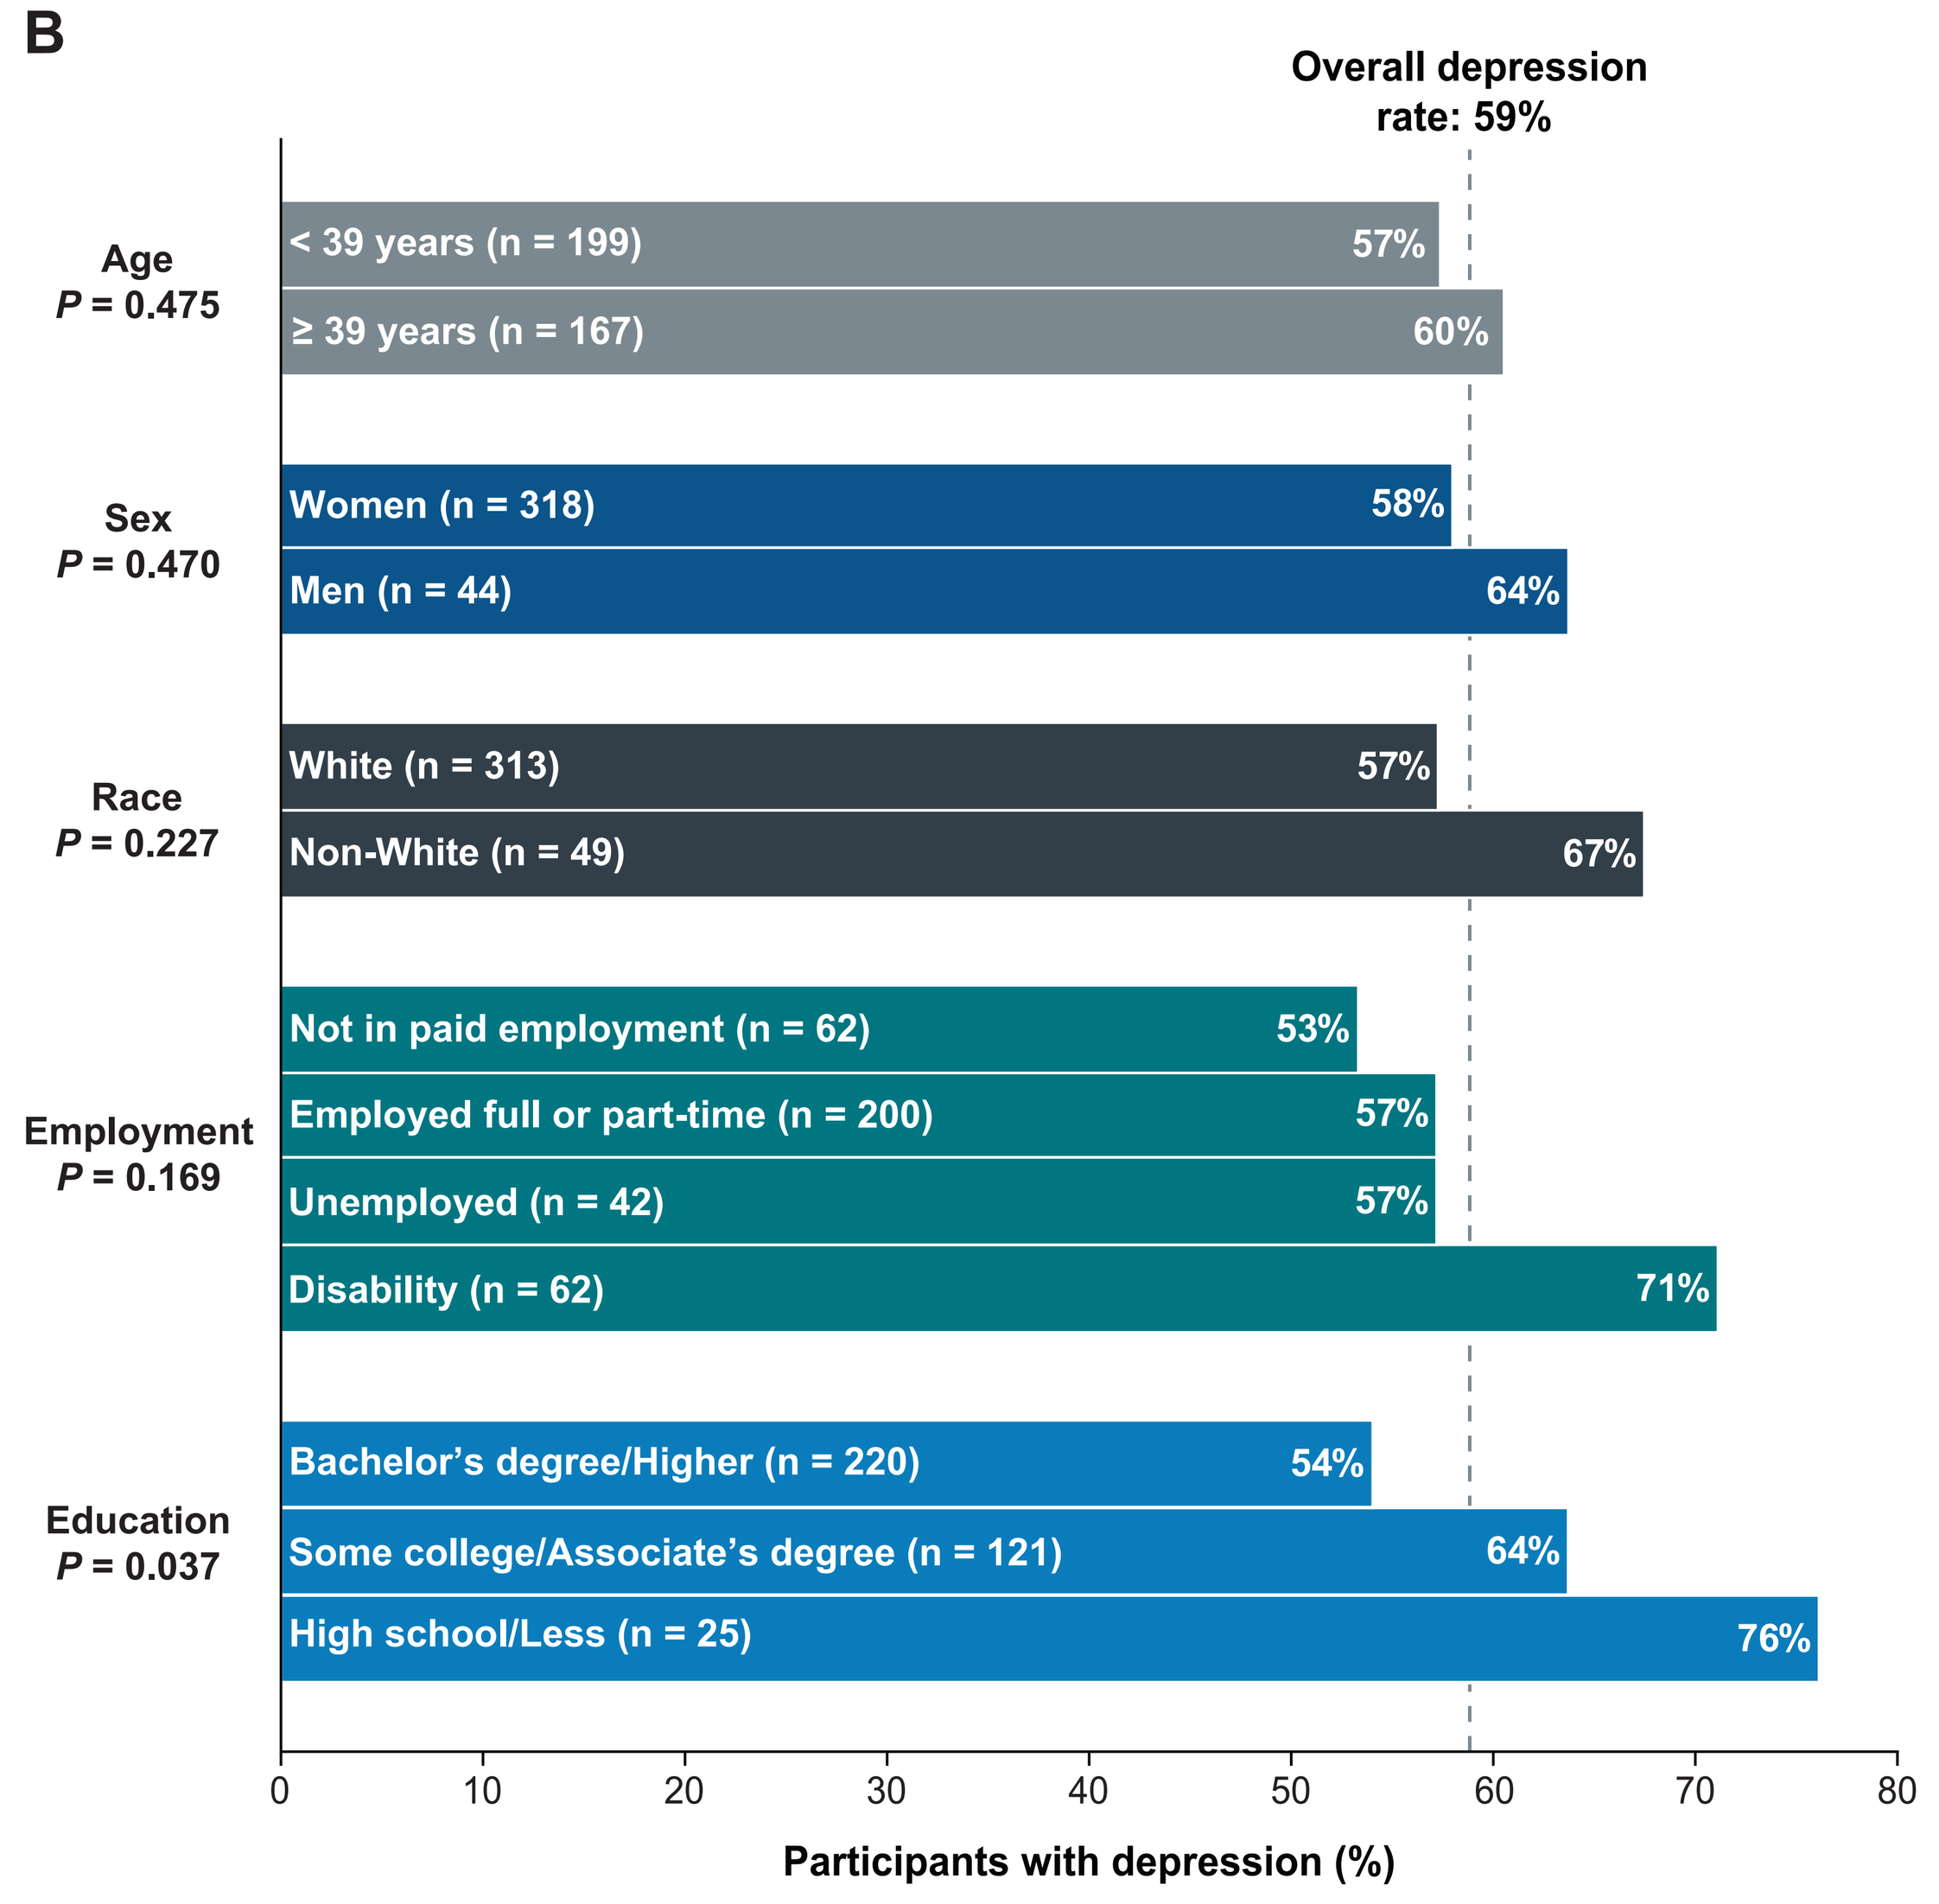
Overall depression rate: 59%
Age P = 0.475
< 39 years (n = 199) 57%
≥ 39 years (n = 167) 60%
Sex P = 0.470
Women (n = 318) 58%
Men (n = 44) 64%
Race P = 0.227
White (n = 313) 57%
Non-White (n = 49) 67%
Employment P = 0.169
Not in paid employment (n = 62) 53%
Employed full or part-time (n = 200) 57%
Unemployed (n = 42) 57%
Disability (n = 62) 71%
Education P = 0.037
Bachelor's degree/Higher (n = 220) 54%
Some college/Associate's degree (n = 121) 64%
High school/Less (n = 25) 76%
Participants with depression (%)

IH, idiopathic hypersomnia; NT1, narcolepsy type 1; NT2, narcolepsy type 2.

- Participants with depression had significantly greater odds of reporting psychosocial burden and perceived life restrictions across all assessed items (all $P < 0.05$), including feeling unable to live the life they wanted, feeling misunderstood by others, having to give up aspirations and dreams and feeling unable to be spontaneous (**Figure 2**)
 - The largest associations were observed for feeling unable to live the life they wanted and feeling misunderstood by others (OR = 3.54 and OR = 3.64, respectively)

Figure 2. Distribution of responses to selected psychosocial impact items by depression status

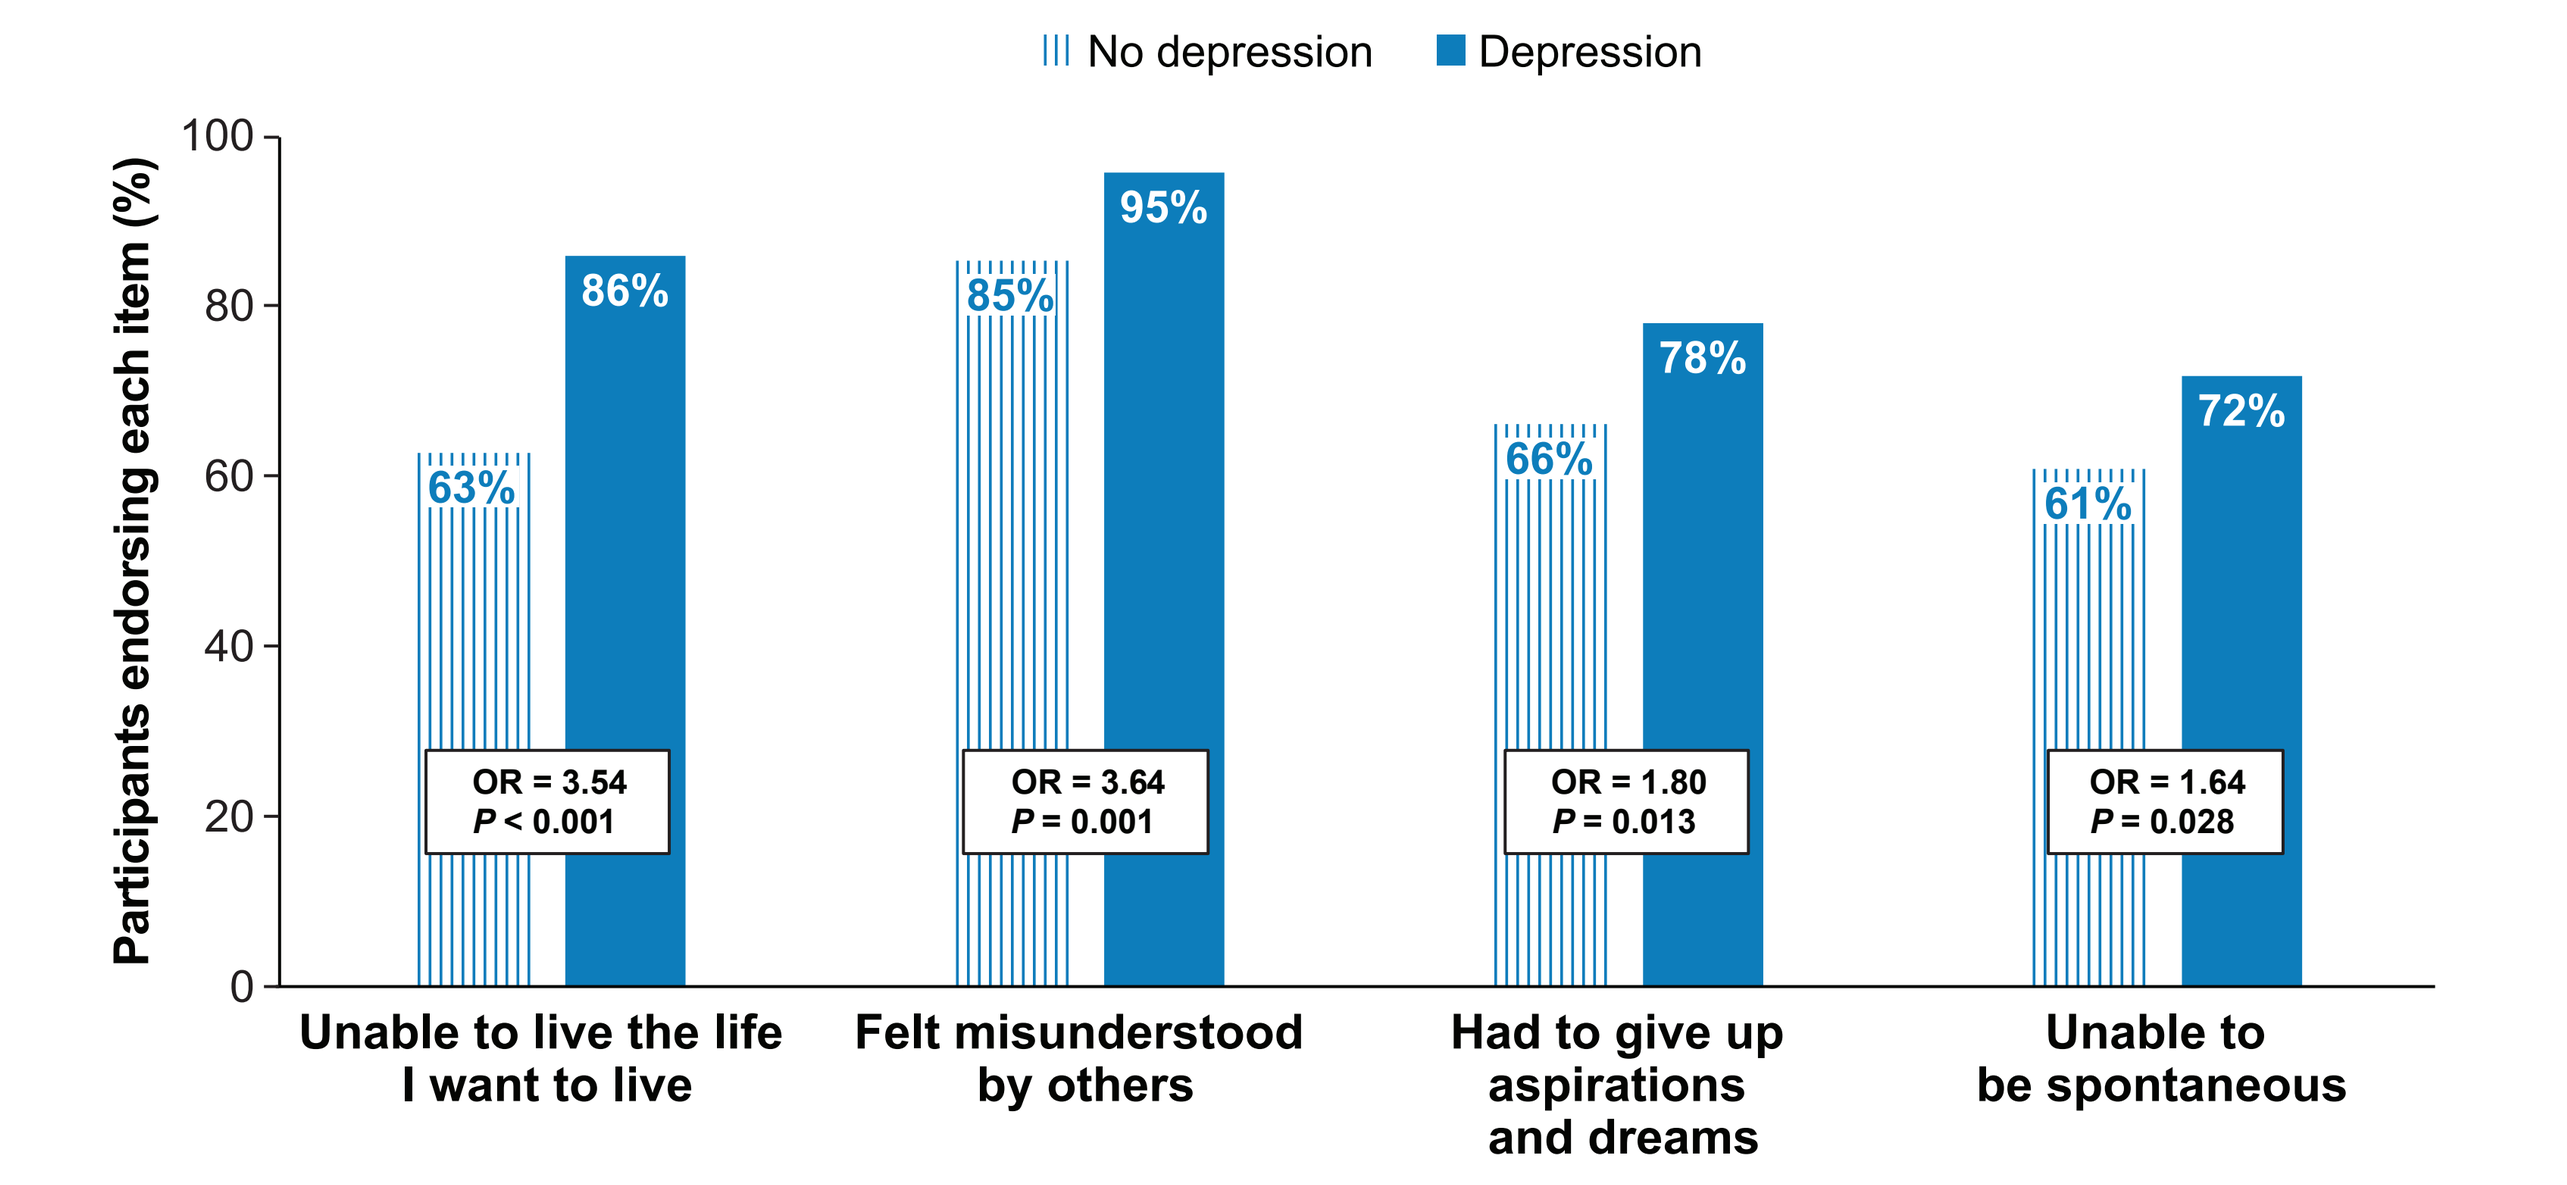
No depression
Depression
Participants endorsing each item (%)
Unable to live the life I want to live: 63% / 86%, OR = 3.54, P < 0.001
Felt misunderstood by others: 85% / 95%, OR = 3.64, P = 0.001
Had to give up aspirations and dreams: 66% / 78%, OR = 1.80, P = 0.013
Unable to be spontaneous: 61% / 72%, OR = 1.64, P = 0.028

OR, odds ratio.

- Depression was associated with significantly greater symptom severity across measures of sleepiness, fatigue, cognitive impairment, and sleep inertia (all $P < 0.001$; **Figure 3**)
 - The largest effect sizes were observed for fatigue and cognitive complaint
- Significantly greater functional, activity, and work impairment, as well as lower HRQoL, were also observed among participants with depression (all $P < 0.001$)

Figure 3. Magnitude of differences in symptom severity, functional impairment, and HRQoL by depression status

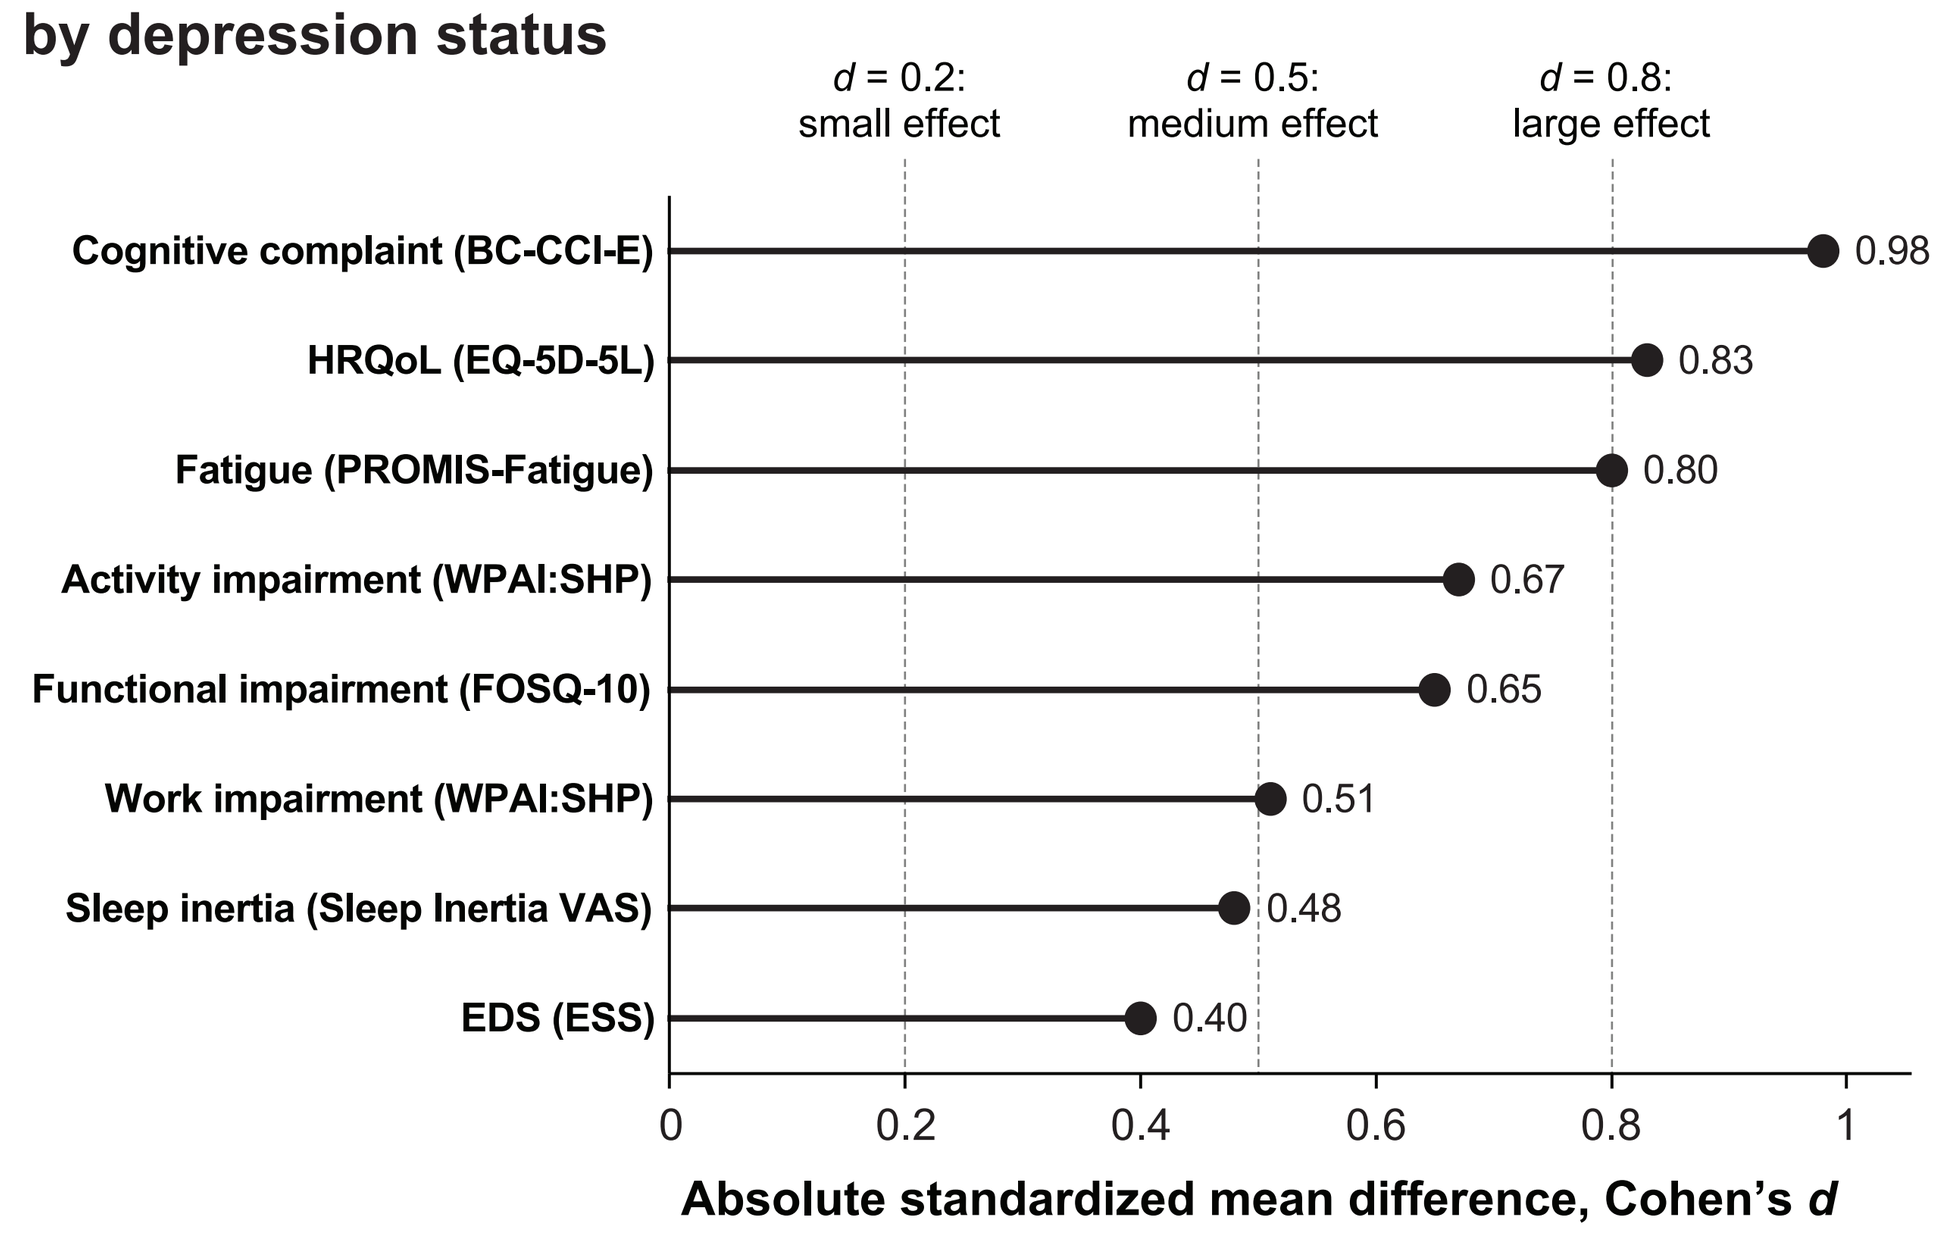
d = 0.2: small effect
d = 0.5: medium effect
d = 0.8: large effect
Cognitive complaint (BC-CCI-E) 0.98
HRQoL (EQ-5D-5L) 0.83
Fatigue (PROMIS-Fatigue) 0.80
Activity impairment (WPAI:SHP) 0.67
Functional impairment (FOSQ-10) 0.65
Work impairment (WPAI:SHP) 0.51
Sleep inertia (Sleep Inertia VAS) 0.48
EDS (ESS) 0.40
Absolute standardized mean difference, Cohen's d

	No depression	Depression
Cognitive complaint (BC-CCI-E)	7.0	10.8
HRQoL (EQ-5D-5L)	0.76	0.55
Fatigue (PROMIS-Fatigue)	62.5	68.1
Activity impairment (WPAI:SHP)	54.1%	68.0%
Functional impairment (FOSQ-10)	12.9	11.0
Work impairment (WPAI:SHP)	47.5%	59.5%
Sleep inertia (Sleep Inertia VAS)	58.9	71.4
EDS (ESS)	12.3	14.1

Note: Cohen's *d* values are presented as absolute values and ranked from largest to smallest. Higher values indicate greater standardized differences between participants with and without depression. Dotted lines indicate conventional thresholds for small (*d* = 0.2), medium (*d* = 0.5), and large (*d* = 0.8) effect sizes[10]

BC-CCI-E, British Columbia Cognitive Complaints Inventory − Expanded version; *d*, Cohen's *d* effect size; EDS, excessive daytime sleepiness; EQ-5D-5L, EuroQol 5-Dimension 5-Level index; ESS, Epworth Sleepiness Scale; FOSQ-10, Functional Outcomes of Sleep Questionnaire-Short Form; HRQoL, health-related quality of life; PROMIS-Fatigue, Patient-Reported Outcomes Measurement Information System − Fatigue Short Form 6a; VAS, visual analogue scale; WPAI:SHP, Work Productivity and Activity Impairment Questionnaire: Specific Health Problem.

- Participants with depression were significantly more likely than those without depression to report that cognitive impairment made it difficult to do their job, maintain good relationships with family and friends, and enjoy social or recreational activities or hobbies (all $P < 0.001$; **Figure 4**)

Figure 4. Impact of cognitive impairment on daily life by depression status

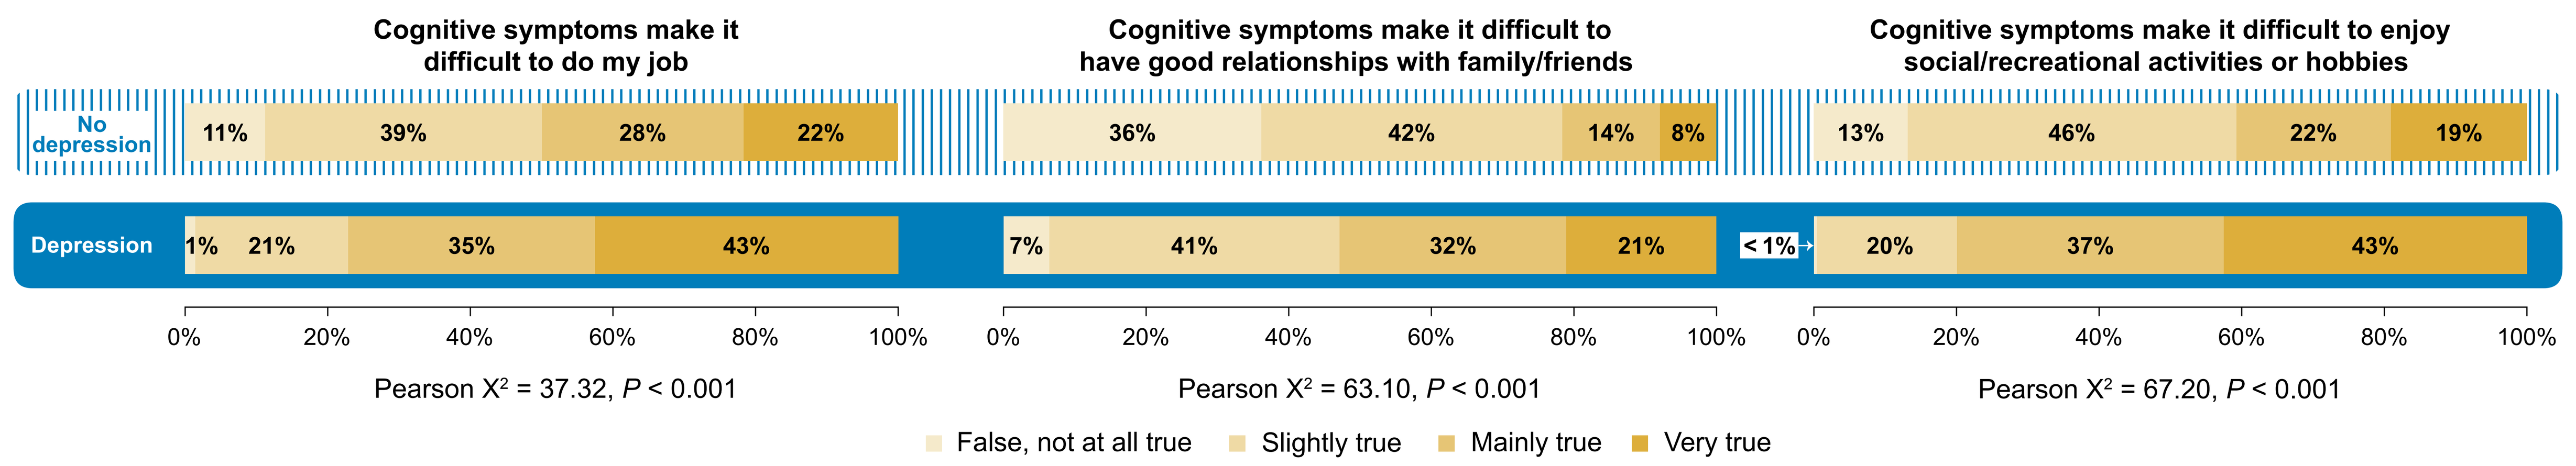
Cognitive symptoms make it difficult to do my job
Cognitive symptoms make it difficult to have good relationships with family/friends
Cognitive symptoms make it difficult to enjoy social/recreational activities or hobbies
No depression
11% 39% 28% 22%
36% 42% 14% 8%
13% 46% 22% 19%
Depression
1% 21% 35% 43%
7% 41% 32% 21%
< 1% 20% 37% 43%
Pearson X² = 37.32, P < 0.001
Pearson X² = 63.10, P < 0.001
Pearson X² = 67.20, P < 0.001
False, not at all true
Slightly true
Mainly true
Very true

Note: Percentages within each group may not sum to 100% due to rounding.

- Among participants with NT1, depression was associated with higher weekly cataplexy frequency versus no depression (7.79 vs 4.35 events/week; $P = 0.005$; **Figure 5**)
 - Most participants with NT1 reported some disruption from cataplexy; however, significant/severe disruption was more frequent among those with depression versus without depression (28% vs 4%), representing a marked shift toward a more severe cataplexy impact

Figure 5. Weekly cataplexy frequency and disruptiveness among participants with NT1 by depression status

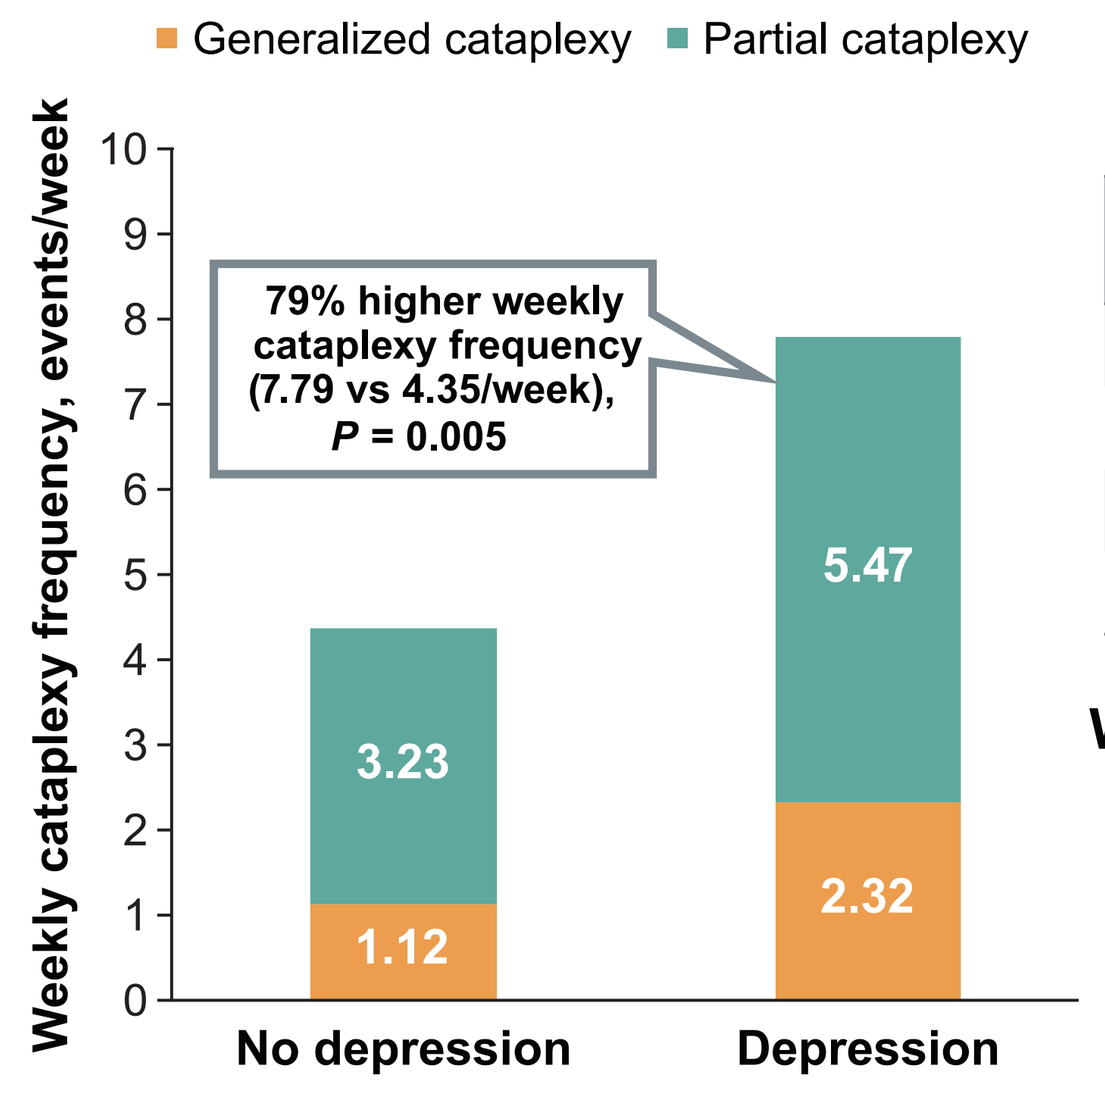
Generalized cataplexy
Partial cataplexy
Weekly cataplexy frequency, events/week
79% higher weekly cataplexy frequency (7.79 vs 4.35/week), P = 0.005
No depression: 1.12, 3.23
Depression: 2.32, 5.47

Disruptiveness of cataplexy	No depression	Depression
Not disruptive	29%	18%
Minimal/moderate disruption	67%	54%
Significant/severe disruption	4%	28%
$\chi^2(2) = 11.16$, $P = 0.0038$		

While most participants with NT1 reported some disruption from cataplexy, depression was associated with a marked shift (6.7-fold higher) toward more severe levels of impact

NT1, narcolepsy type 1.

- Depression was associated with significantly greater prioritization of psychosocial treatment benefits, including feeling less down/depressed, less isolated, less anxious, more in control, and less ashamed of their disease (all $P < 0.05$; **Figure 6**)
 - The strongest associations were observed for feeling less down/depressed, less isolated, and less anxious, with these benefits prioritized more often among participants with depression (OR = 5.95, OR = 4.07, and OR = 3.44, respectively)

Figure 6. Treatment benefits rated as important or very important by depression status

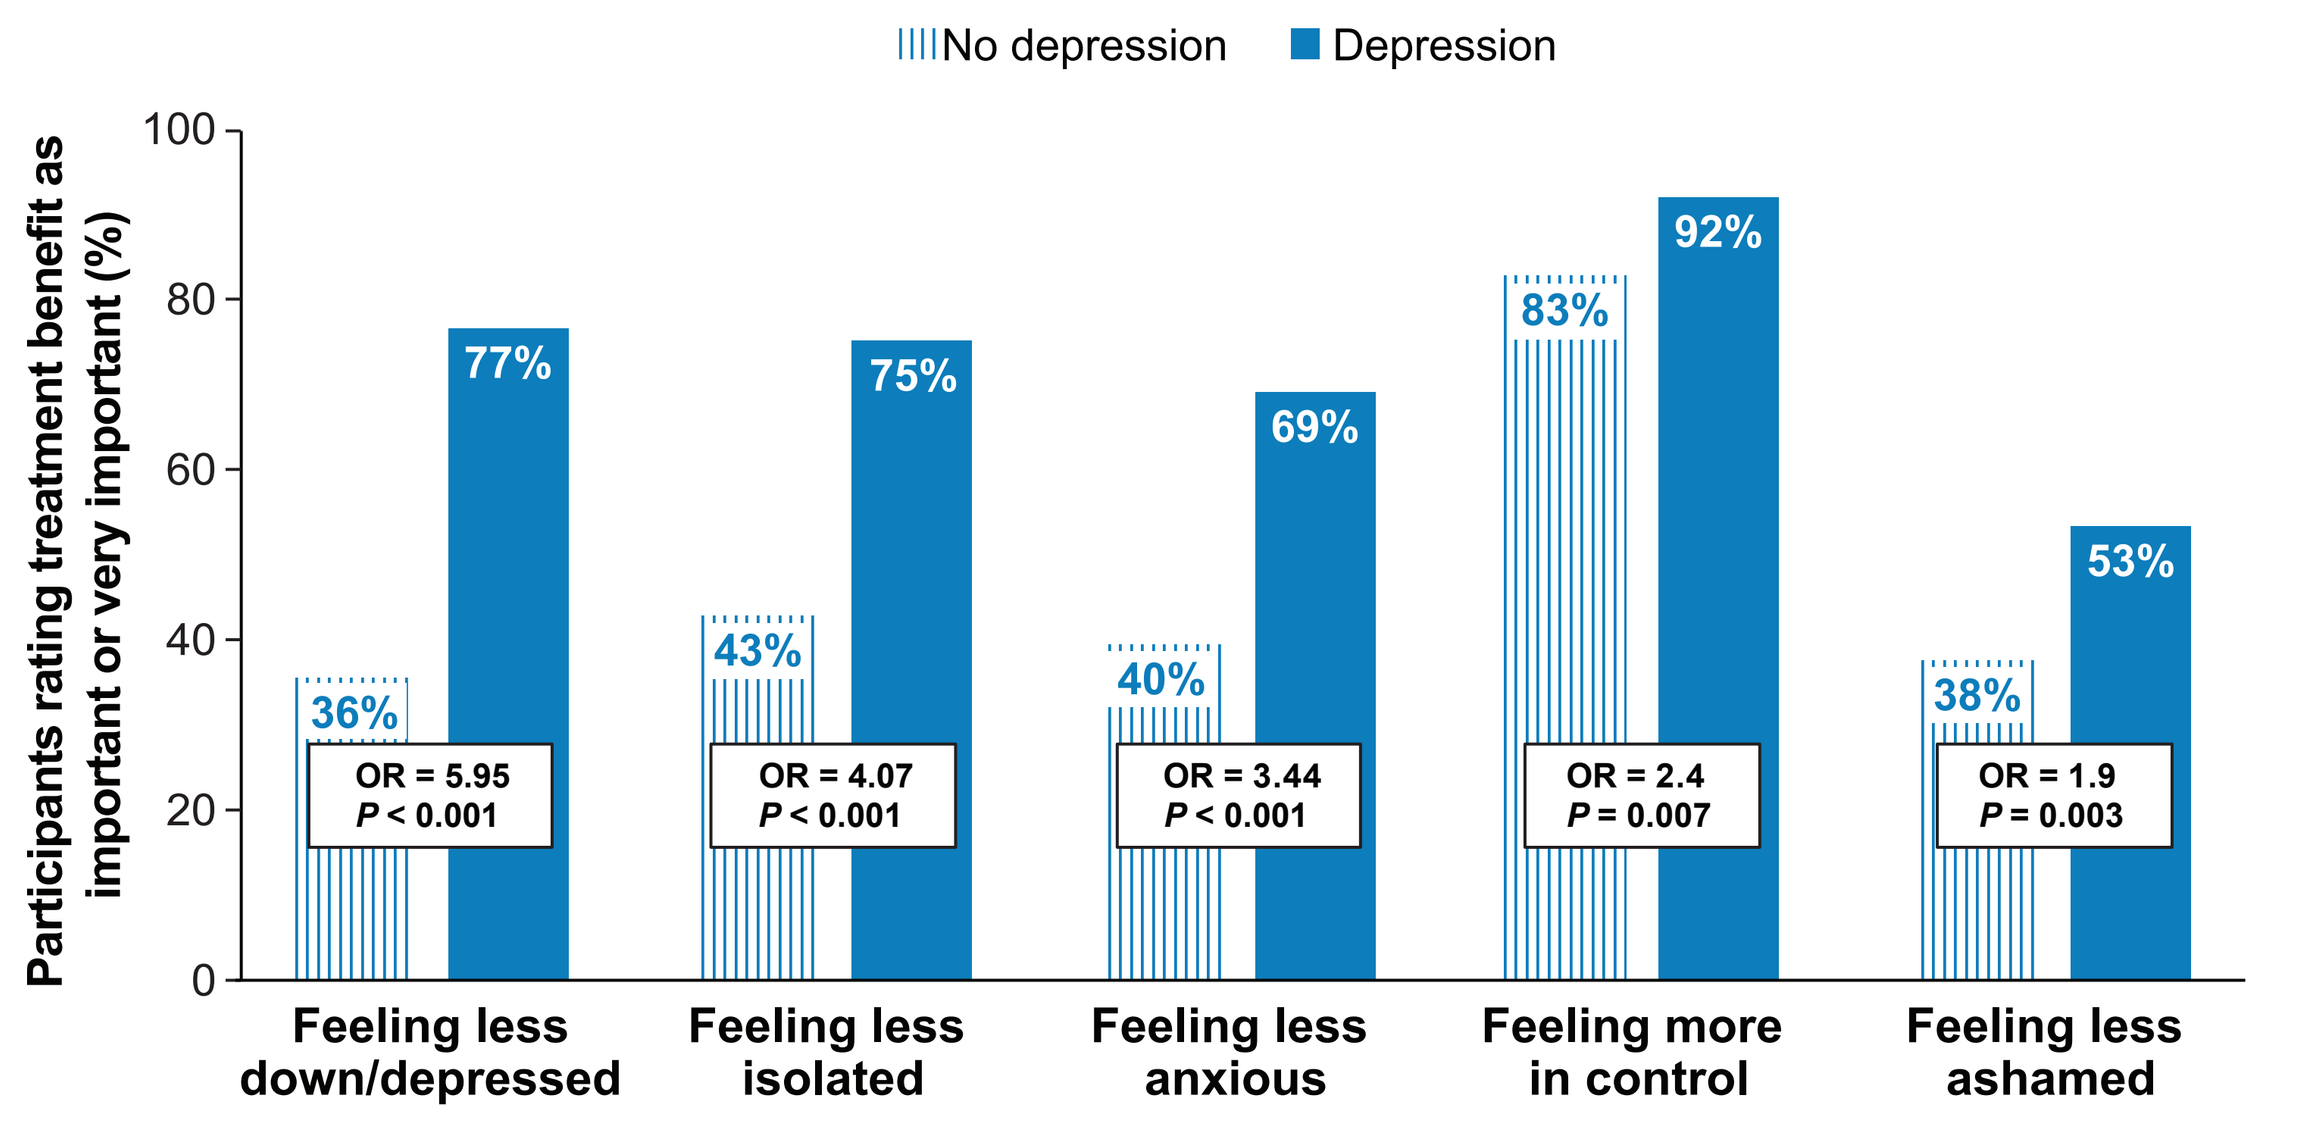
No depression
Depression
Participants rating treatment benefit as important or very important (%)
Feeling less down/depressed: 36% / 77%, OR = 5.95, P < 0.001
Feeling less isolated: 43% / 75%, OR = 4.07, P < 0.001
Feeling less anxious: 40% / 69%, OR = 3.44, P < 0.001
Feeling more in control: 83% / 92%, OR = 2.4, P = 0.007
Feeling less ashamed: 38% / 53%, OR = 1.9, P = 0.003

OR, odds ratio.

Conclusions

- Depression was common in individuals with NT1, NT2, and IH as defined by MCS scores, affecting over half of participants and representing a clinically relevant, high-burden subgroup on clinical, functional, and psychosocial outcomes
- Greater burden was observed among participants with depression, including perceived life restrictions, symptom severity, functional impairment, activity/work impairment, and lower HRQoL; these differences extended beyond EDS and were most pronounced for cognitive impairment, HRQoL, and fatigue
- Among participants with NT1, depression was associated with higher weekly cataplexy frequency and greater cataplexy-related disruption
- Participants with depression prioritized psychosocial treatment benefits, supporting routine assessment of mental health and incorporation of patient-centered outcomes when evaluating disease burden and treatment benefits in narcolepsy and IH

Acknowledgments
The authors thank all the participants in this study for their time and valuable input. This study was funded by Alkermes, Inc. Medical writing support was provided by Shawn Jaramillo, PharmD, BCACP, of Oxford PharmaGenesis Inc., Wilmington, DE, USA, and funded by Alkermes, Inc., Waltham, MA, USA.

Disclosures
CC has been a consultant or on an advisory board for or has received grant or research support from Acadia, Axsome, Harmony, Neurocrine, and Teva; has served as a consultant for AbbVie, Acadia, Alkermes, Almatica, Axsome, Biogen, Boehringer Ingelheim, Corium, Intra-Cellular Therapies, Janssen, Karuna, Lundbeck, MedinCell, Moderna, Neurocrine, Noven, Otsuka, Sage, Sumitomo, Supernus, and Teva; has received payment or honoraria for educational activities from AbbVie, Acadia, Alkermes, Axsome, Bristol Myers Squibb, Corium, Intra-Cellular Therapies, Janssen, Karuna, Lundbeck, Merck, Neurocrine, Noven, Otsuka, Sumitomo, and Teva; has received support for attending meetings/travel from AbbVie, Acadia, Alkermes, Axsome, Bristol Myers Squibb, Corium, Intra-Cellular Therapies, Janssen, Karuna, Lundbeck, Merck, Neurocrine, Noven, Otsuka, Sumitomo, Teva, and Tonix; and has served on an advisory or data safety monitoring board for AbbVie, Acadia, Alkermes, Axsome, Biogen, Bristol Myers Squibb, Corium, Idorsia, Intra-Cellular Therapies, Janssen, Karuna, Lundbeck, Moderna, Neurocrine, Noven, Otsuka, Sage, Sumitomo, and Teva. **MJD** and **GM** are employees and shareholders of Alkermes. **LJ** has received grants or contracts, disbursed to Hypersomnia Foundation and Sleep Consortium, from Alkermes, Inc., Avadel, Centessa, Harmony Biosciences, Jazz Pharmaceuticals, Takeda Pharmaceuticals, and Zevra; received consulting fees from Harmony Biosciences and Takeda Pharmaceuticals; and holds a leadership or fiduciary role with Hypersomnia Foundation. **KM** and **JLT** are employees of IQVIA Quality Metric. **AMM** has served as a consultant, speaker, and/or on advisory boards for Alkermes, Avadel Pharmaceuticals, Eisai, Harmony Biosciences, Jazz Pharmaceuticals, and Takeda Pharmaceutical Co; has received grant funding from Coverys Foundation, Geisinger Health Plan, Harmony Biosciences, Jazz Pharmaceuticals, National Institutes of Health, ResMed Foundation, and UCB Pharmaceuticals; is the CEO of DAMM Good Sleep LLC; and serves as an advisor for Floraworks and Neura Health. **DTP** has served as a consultant or advisory board participant for Aditum Bio LLC, Alkermes, Inc., Apnimed, Centessa, Eisai, Harmony Biosciences, Jazz Pharmaceuticals, Lundbeck, Takeda, and Teva.

References
1. Maski K et al. *J Clin Sleep Med*. 2017;13(3):419-425.
2. Ingravallo F et al. *Sleep Med*. 2012;13(10):1293-1300.
3. Boulanger T et al. *Sleep Adv*. 2024;5(1):zpae059.
4. Miglis MG et al. *J Clin Sleep Med*. 2020;16(5):749-756.
5. Ohayon MM. *Sleep Med*. 2013;14(6):488-492.
6. Stevens J et al. *Nat Sci Sleep*. 2023;15:593-606.
7. Trotti LM. *Sleep Med Clin*. 2017;12(3):331-344.
8. Maruish ME, ed. *User's Manual for the SF-12v2 Health Survey*. 3rd ed. QualityMetric Incorporated; 2012.
9. Ware JE Jr, Kosinski M. *SF-36 Physical and Mental Health Summary Scales: A Manual for Users of Version 1*. 2nd ed. QualityMetric Incorporated; 2001.
10. Cohen J. *Statistical Power Analysis for the Behavioral Sciences*. 2nd ed. Lawrence Erlbaum Associates; 1988.

Scan QR code to download poster